A Humanized *RBM20* Mouse Model Exhibits Dilated Cardiomyopathy Phenotypes and Enables Development of *In Vivo* Prime Editing for Treating Human *RBM20* Cardiomyopathy Patients

Wenjing Liang, Emilee Easter, **Lindsey M. Rollosson**, Amara Greer-Short, Laura M. Lombardi, Timothy Hoey, Kathryn N. Ivey, **Ze Cheng**

Tenaya Therapeutics, Inc. South San Francisco, CA Correspondence: Wenjing Liang (wliang@tenayathera.com), Ze Cheng (zcheng@tenayathera.com)

INTRODUCTION

► RNA-binding Motif Protein 20 (RBM20) plays a critical role in regulating RNA splicing in cardiomyocytes, and mutations in the *RBM20* gene are a significant cause of dilated cardiomyopathy (DCM). This genetic form of DCM is characterized by high penetrance and an aggressive clinical course, frequently leading to severe outcomes such as heart failure, arrhythmias, and sudden cardiac death. The majority of *RBM20* disease-causing mutations cluster in a 5-amino-acid arginine/serine-rich (R/S rich) region, which mediates nuclear localization of RBM20 protein in cardiomyocytes. Currently, no disease-specific therapy targeting the underlying genetic cause of *RBM20* cardiomyopathy is available, we have developed an *in vivo* cardiomyocyte prime editing (PE) strategy (Abstract #1028) which may be suitable for correcting the mutations in many RBM20 patients, as PE can mediate all 12 types of base-to-base conversions and various mutations in the *RBM20* hotspot may be addressed by the same PE therapeutic.

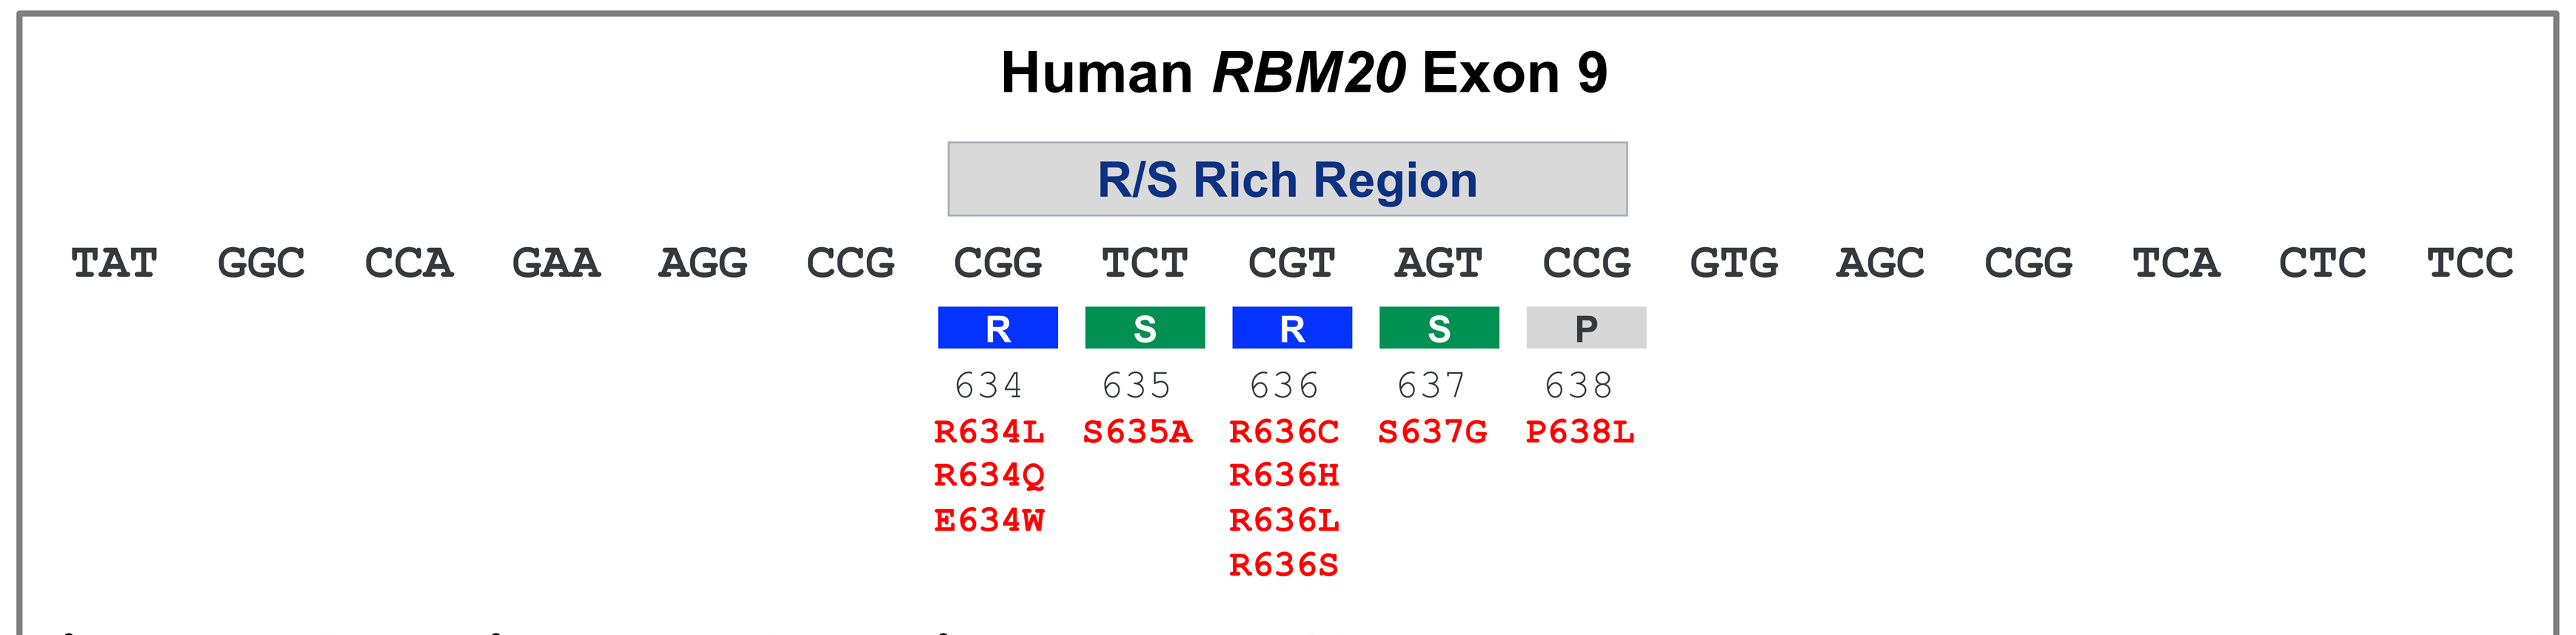
Human *RBM20* Exon 9
R/S Rich Region
TAT GGC CCA GAA AGG CCG CGG TCT CGT AGT CCG GTG AGC CGG TCA CTC TCC
R S R S P
634 635 636 637 638
R634L R634Q E634W
S635A
R636C R636H R636L R636S
S637G
P638L

Figure 1. Schematic DNA and protein sequence of human *RBM20*. The encoding DNA sequence of the 5-amino-acid R/S rich region resides in the exon 9 of human *RBM20* gene. Pathogenic variants reported in literature are highlighted in this figure.

Development of Humanized *RBM20* Mouse Models

► To model the disease and enable *in vivo* testing of human *RBM20* targeting PE therapeutics, we engineered a humanized *RBM20* mouse model with one allele carrying the human wildtype (WT) DNA sequence and one allele carrying the human R634Q (equivalent to R636Q with mouse numbering) mutant DNA sequence in the hotspot and flanking regions (*hRBM20^{R634Q}/hRBM20^{+}*). The total humanized region was designed to be large enough to accommodate binding of guide RNAs (prime editing guide RNA and nicking guide RNA) in potential human RBM20 PE therapeutics, while being restricted to avoid deviating from the native mouse protein coding outcome.

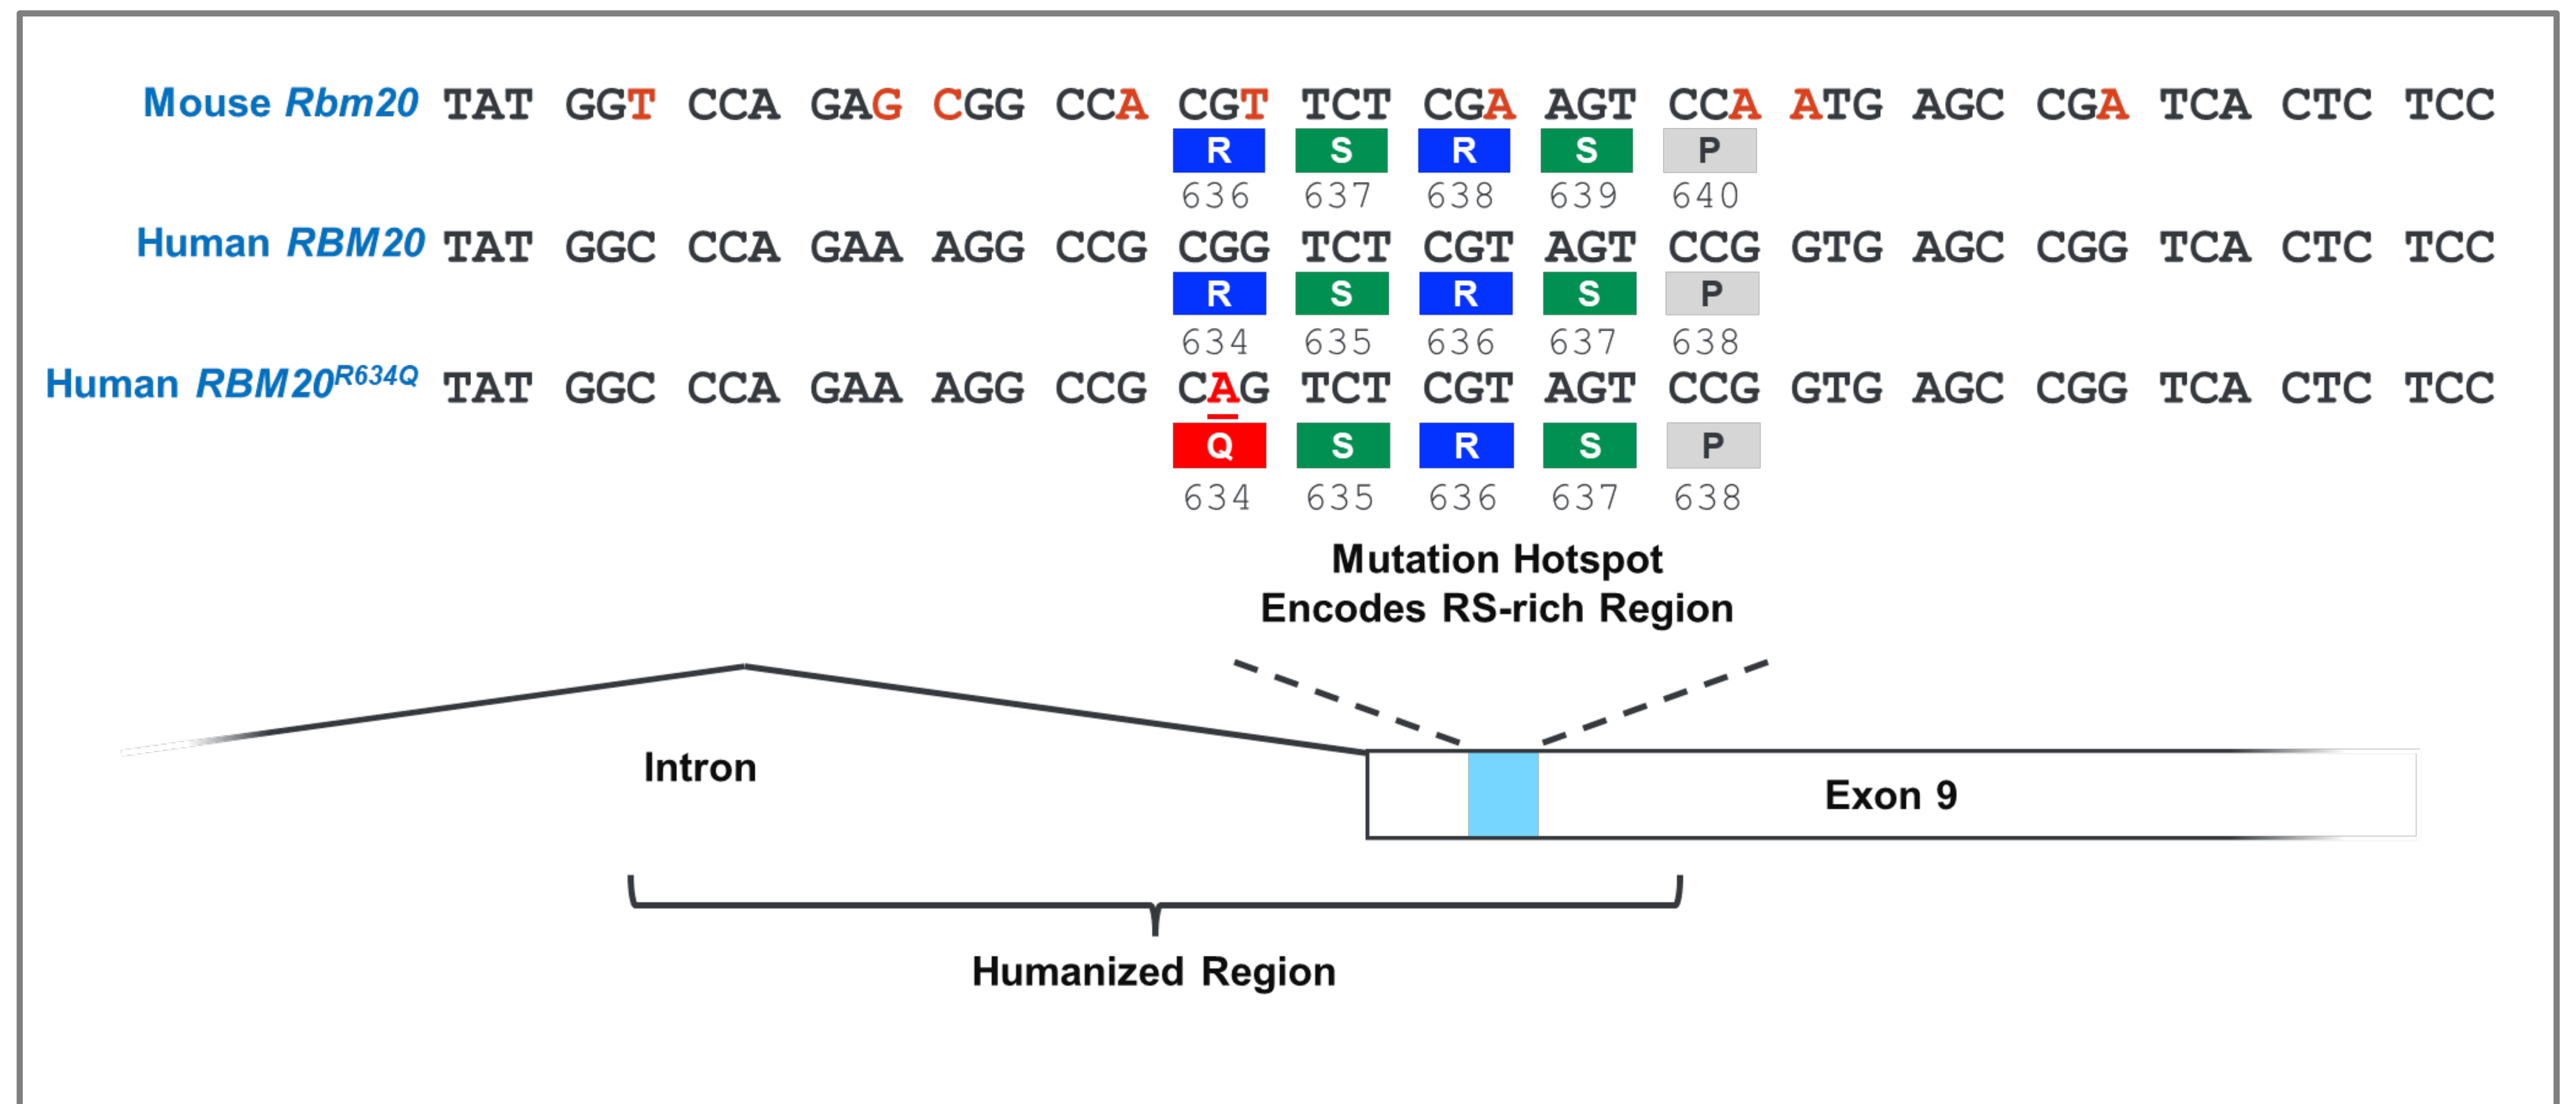
Mouse *Rbm20* TAT GGT CCA GAG CGG CCA CGT TCT CGA AGT CCA ATG AGC CGA TCA CTC TCC
R S R S P
636 637 638 639 640
Human *RBM20* TAT GGC CCA GAA AGG CCG CGG TCT CGT AGT CCG GTG AGC CGG TCA CTC TCC
R S R S P
634 635 636 637 638
Human *RBM20^{R634Q}* TAT GGC CCA GAA AGG CCG CAG TCT CGT AGT CCG GTG AGC CGG TCA CTC TCC
Q S R S P
634 635 636 637 638
Mutation Hotspot
Encodes RS-rich Region
Intron
Exon 9
Humanized Region

Figure 2. Generation of humanized *RBM20* mice. DNA sequences, protein sequences, and amino acid numbering alignment of mouse *Rbm20*, human *RBM20*, and human *RBM20^{R634Q}* at the mutation hotspot region are shown. The human *RBM20* DNA sequences, including those for WT *RBM20* and the R634Q (G>A) mutation, were knocked into exon 9 and the upstream intron to replace the mouse sequence. This strategy ensures a large enough humanized region for guide RNA binding.

Humanized *RBM20 R634Q* Mice Develop DCM that Mirrors the Pathophysiology of Human *RBM20* Cardiomyopathy

► We initiated model development in 3-4 weeks old mice and tracked their heart function using echocardiography. Humanized WT control mice (*hRBM20^{+}/hRBM20^{+}*) displayed no abnormalities in heart function. In contrast, heterozygous mutant mice (*hRBM20^{R634Q}/hRBM20^{+}*) developed a DCM phenotype, characterized by a moderate decline in ejection fraction (EF) and left ventricular chamber enlargement. Homozygous mutant mice (*hRBM20^{R634Q}/hRBM20^{R634Q}*) exhibited a severe DCM phenotype, featuring a dramatic EF drop, significant left ventricular enlargement, and lifespan of less than 3 months. These phenotypes recapitulate the cardiac dysfunction seen in the mouse *Rbm20 R636Q* model[1] and human *RBM20* Cardiomyopathy.

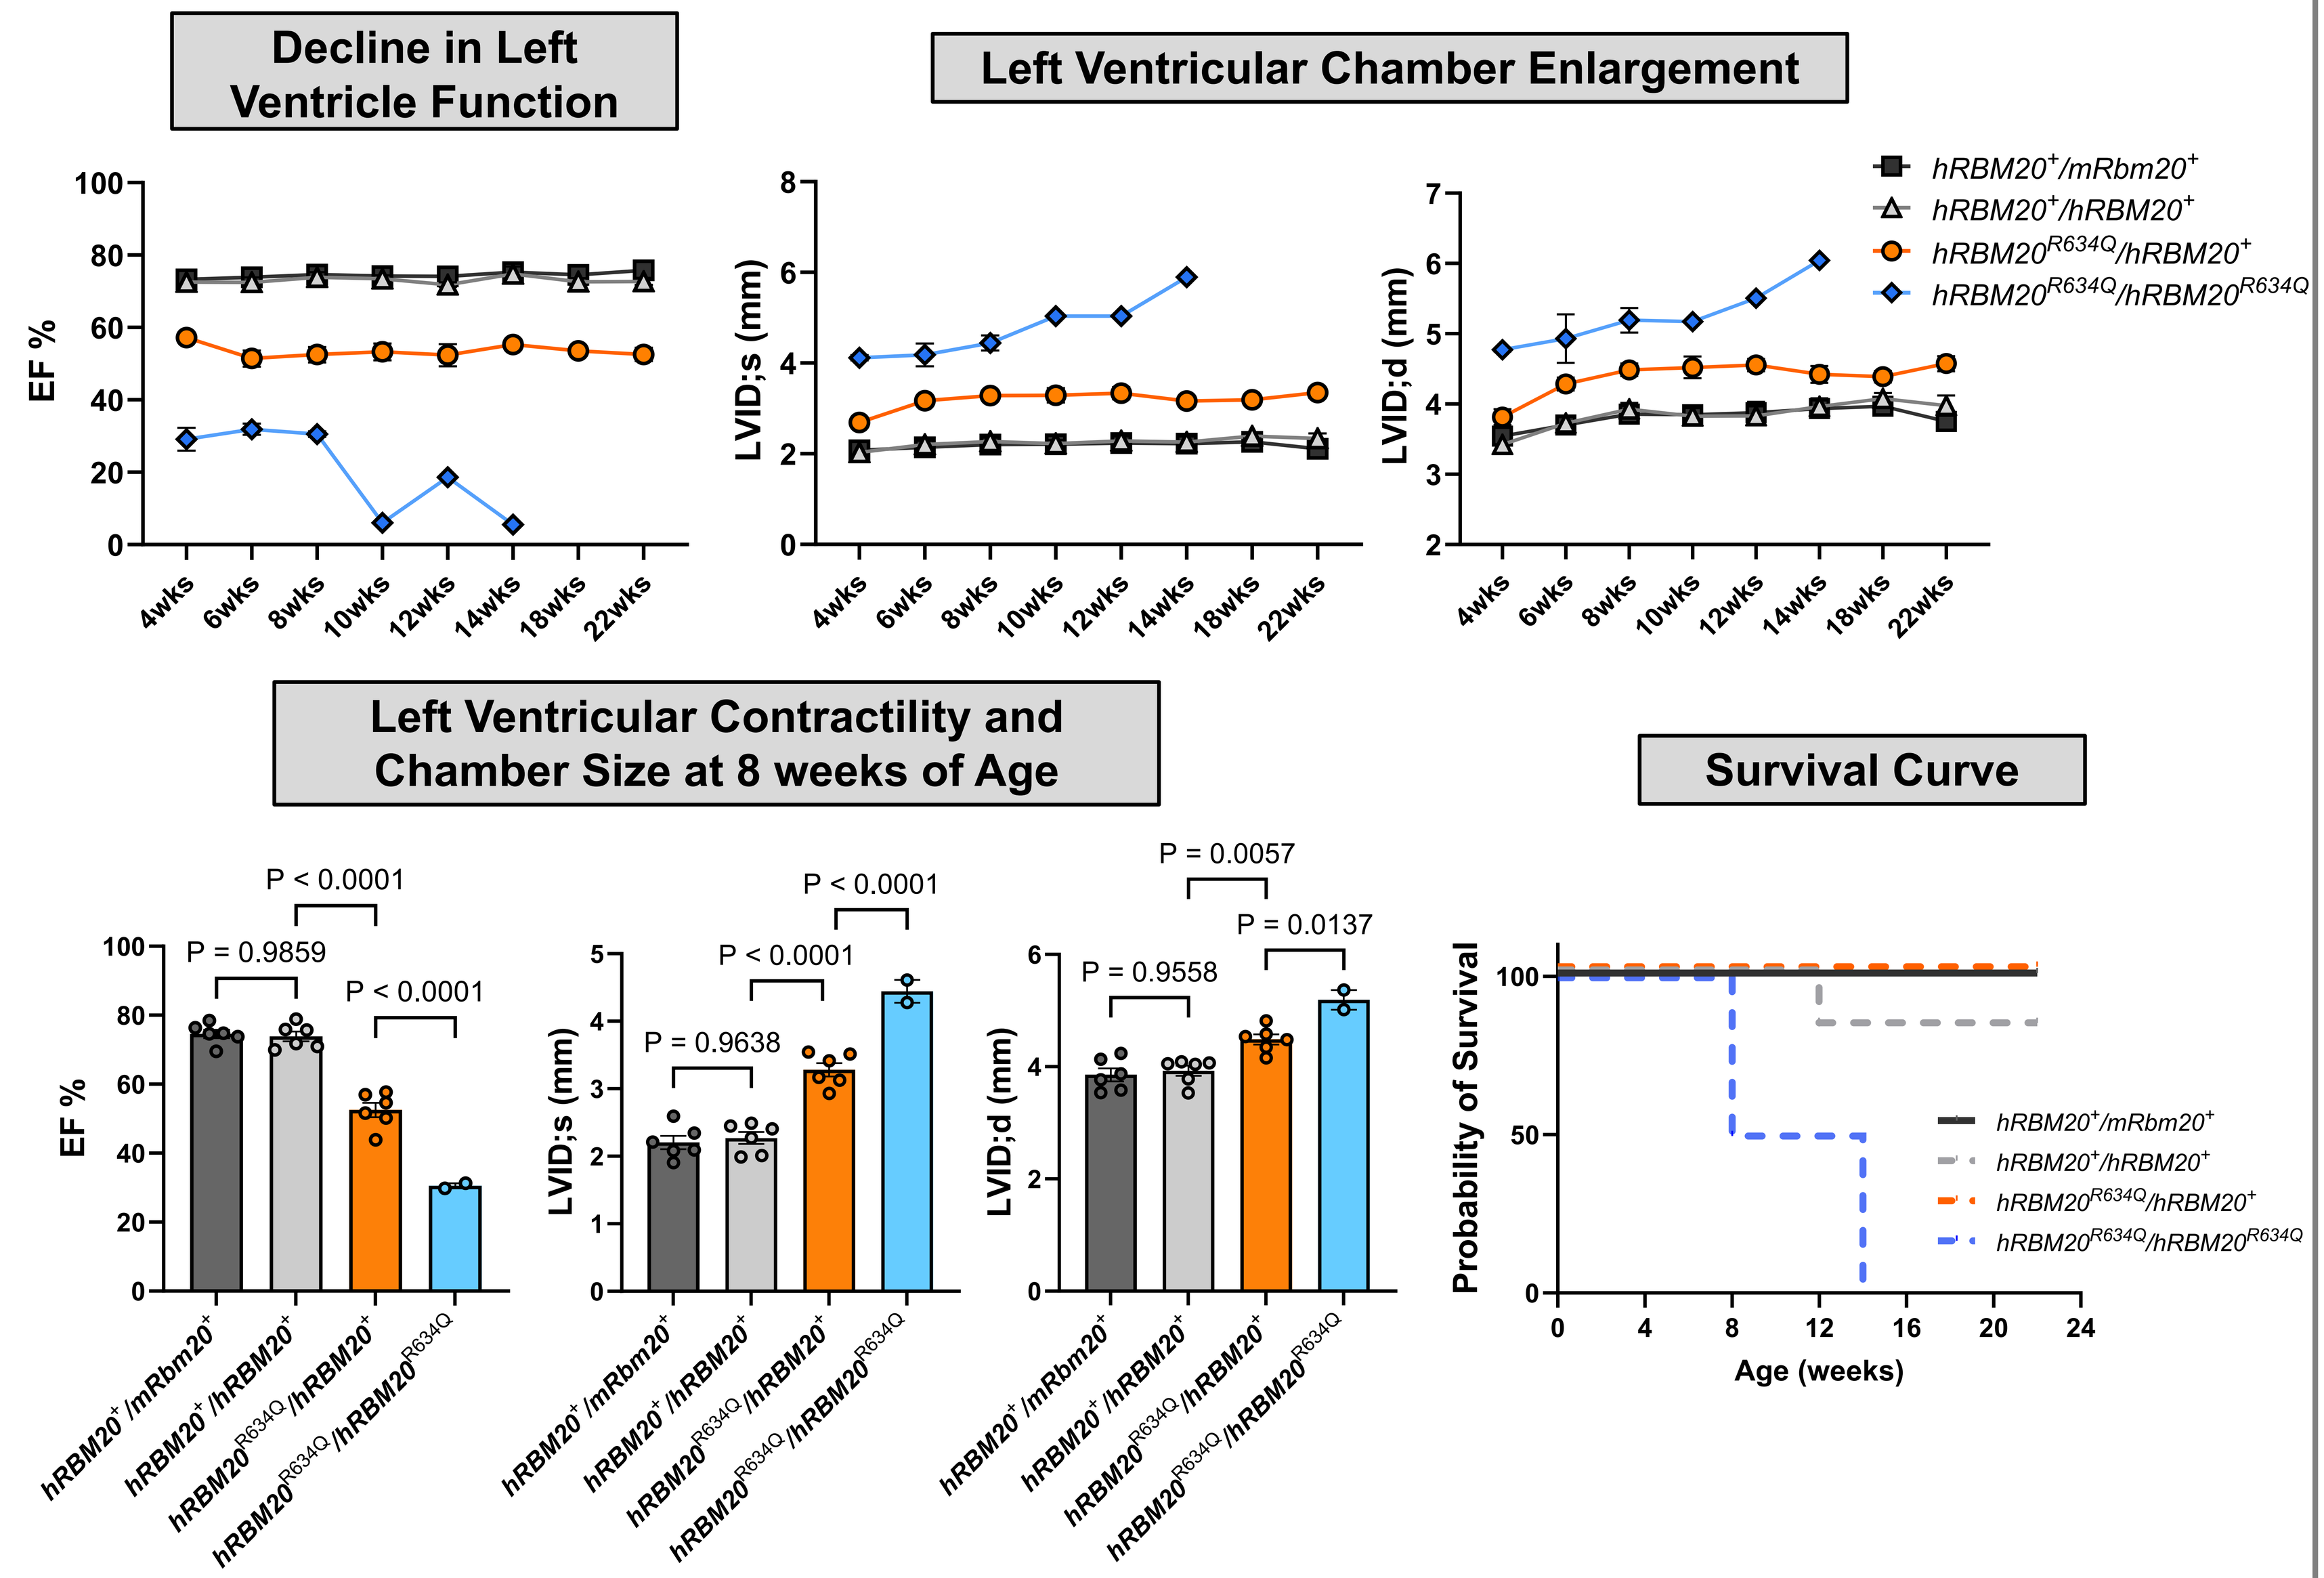
Decline in Left Ventricle Function
Left Ventricular Chamber Enlargement
EF %
LVID;s (mm)
LVID;d (mm)
4wks 6wks 8wks 10wks 12wks 14wks 18wks 22wks
hRBM20^{+}/mRbm20^{+}
hRBM20^{+}/hRBM20^{+}
hRBM20^{R634Q}/hRBM20^{+}
hRBM20^{R634Q}/hRBM20^{R634Q}
Left Ventricular Contractility and Chamber Size at 8 weeks of Age
P < 0.0001
P = 0.9859
P < 0.0001
P = 0.9638
P < 0.0001
P < 0.0001
P = 0.0057
P = 0.9558
P = 0.0137
Survival Curve
Probability of Survival
Age (weeks)

Figure 3. Humanized *RBM20 R634Q* mice show DCM phenotype. Cardiac function in mice was evaluated via echocardiography from 3-4 weeks of age up to 22 weeks. During this period, cardiac-related mortality was exclusively observed within the homozygous mice group.

Biomarker Assessment in Humanized *RBM20 R634Q* Mice

► To further characterize this new humanized mouse model, we performed gene expression profiling studies and observed upregulation of genes indicative of heart failure, including natriuretic peptide A (*Nppa*) and natriuretic peptide B (*Nppb*) in the heart tissue of heterozygous mutant mice (*hRBM20^{R634Q}/hRBM20^{+}*) and homozygous mutant mice (*hRBM20^{R634Q}/hRBM20^{R634Q}*).

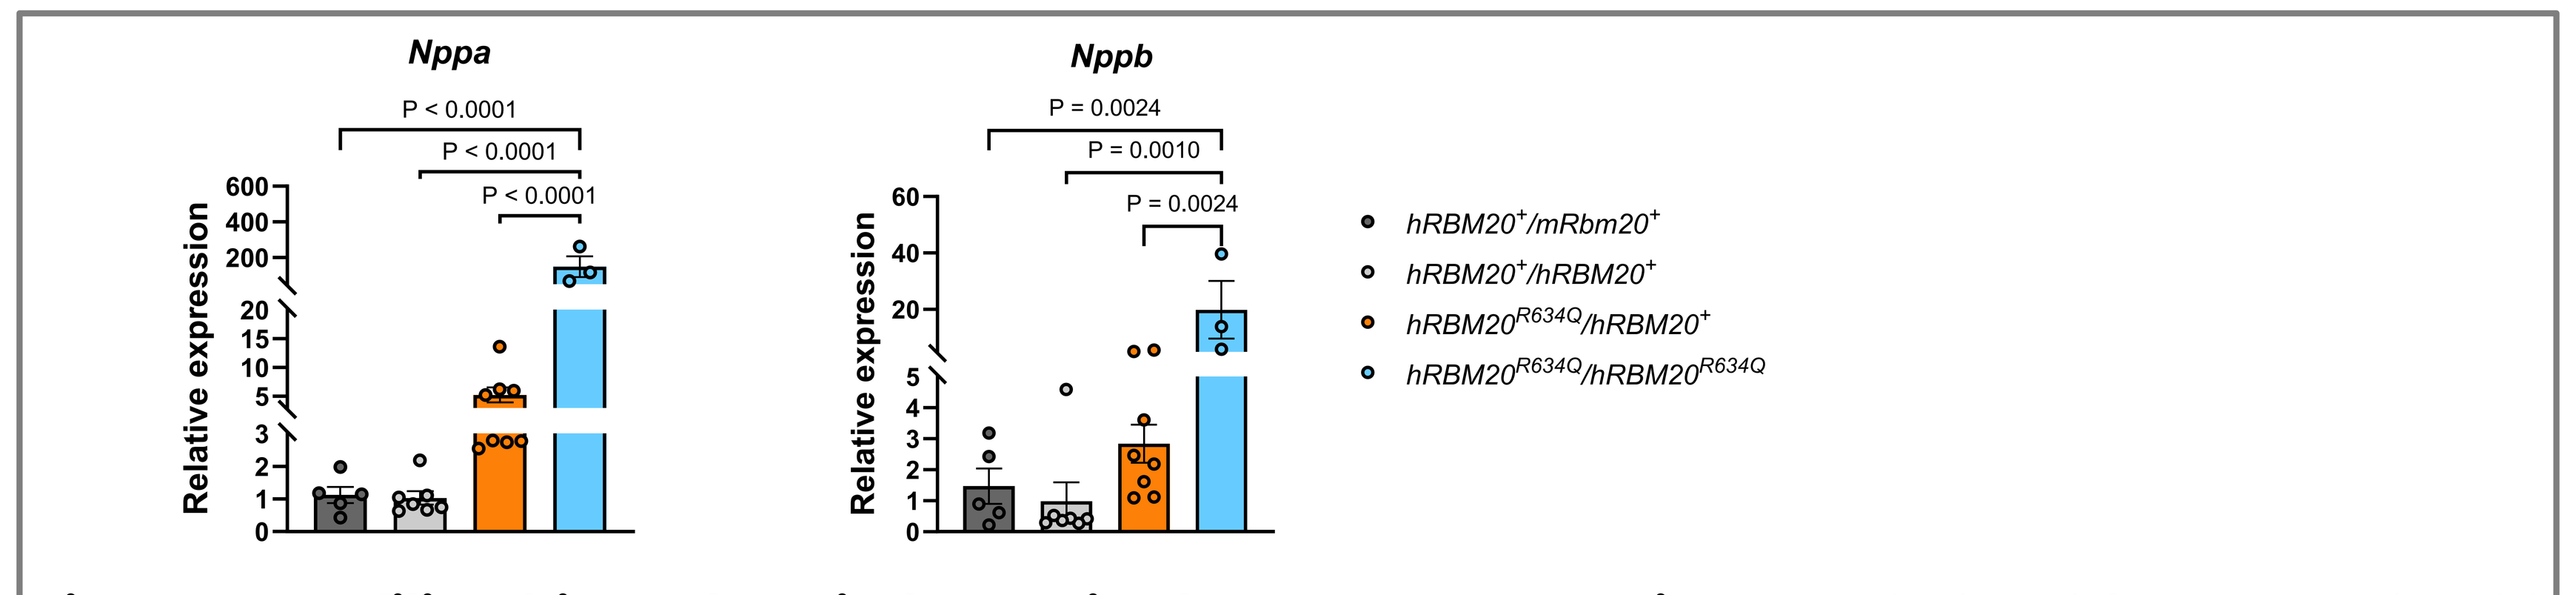
Nppa
P < 0.0001
P < 0.0001
P < 0.0001
Nppb
P = 0.0024
P = 0.0010
P = 0.0024
Relative expression
hRBM20^{+}/mRbm20^{+}
hRBM20^{+}/hRBM20^{+}
hRBM20^{R634Q}/hRBM20^{+}
hRBM20^{R634Q}/hRBM20^{R634Q}

Figure 4. Profiling biomarkers in humanized *RBM20 R634Q* mice. RNA isolated from 8 weeks old mice was used to generate cDNA for RT-qPCR analysis of heart failure markers.

Humanized *RBM20 R634Q* Mice Display Abnormal Splicing of RBM20 Target Genes

► Next, we assessed the splicing pattern of RBM20 targets, *Ttn* and *Camk2d*, in humanized *RBM20 R634Q* mice. Both heterozygous (*hRBM20^{R634Q}/hRBM20^{+}*) and homozygous mutant mice (*hRBM20^{R634Q}/hRBM20^{R634Q}*) show abnormal splicing of *Ttn* and *Camk2d* genes. This observation suggests that the humanized mouse model described herein recapitulates gene splicing phenotypes of *RBM20* cardiomyopathy.

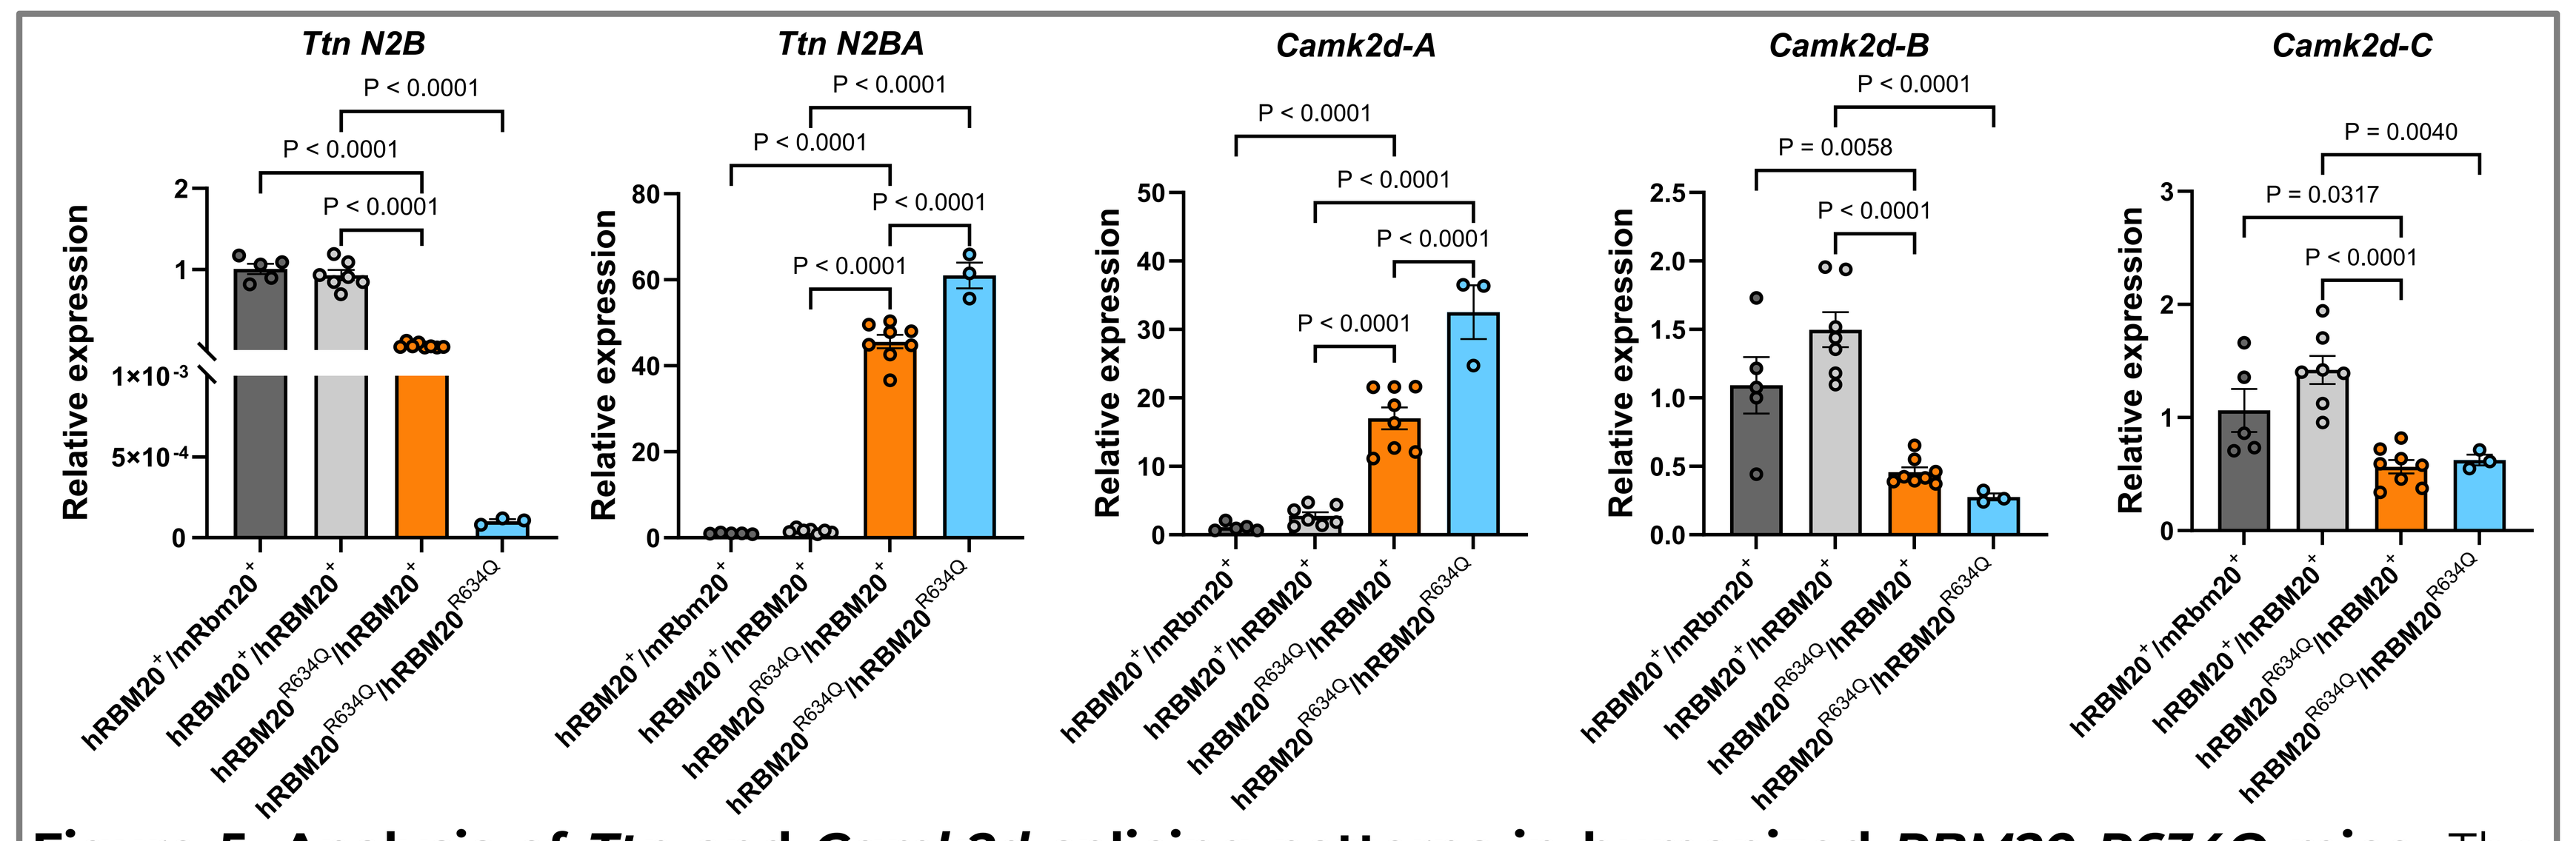
Ttn N2B
Ttn N2BA
Camk2d-A
Camk2d-B
Camk2d-C
Relative expression
P < 0.0001
P = 0.0058
P = 0.0317
P = 0.0040
hRBM20^{+}/mRbm20^{+}
hRBM20^{+}/hRBM20^{+}
hRBM20^{R634Q}/hRBM20^{+}
hRBM20^{R634Q}/hRBM20^{R634Q}

Figure 5. Analysis of *Ttn* and *Camk2d* splicing patterns in humanized *RBM20 R634Q* mice. The splicing isoforms of *Ttn* and *Camk2d* were evaluated in 8 weeks old humanized mice by RT-qPCR.

Humanized *RBM20 R634Q* Mice Enable *In Vivo* Testing of Prime Editing Therapeutics Targeting Human *RBM20*

► Finally, we employed humanized *RBM20 R634Q* mice to test Tenaya *in vivo* cardiac prime editing therapeutics (Abstract 1028) targeting human *RBM20* mutations. This platform allows us to identify efficient versions of prime editors and/or measure the dose response, potentially enabling future efficacy studies.

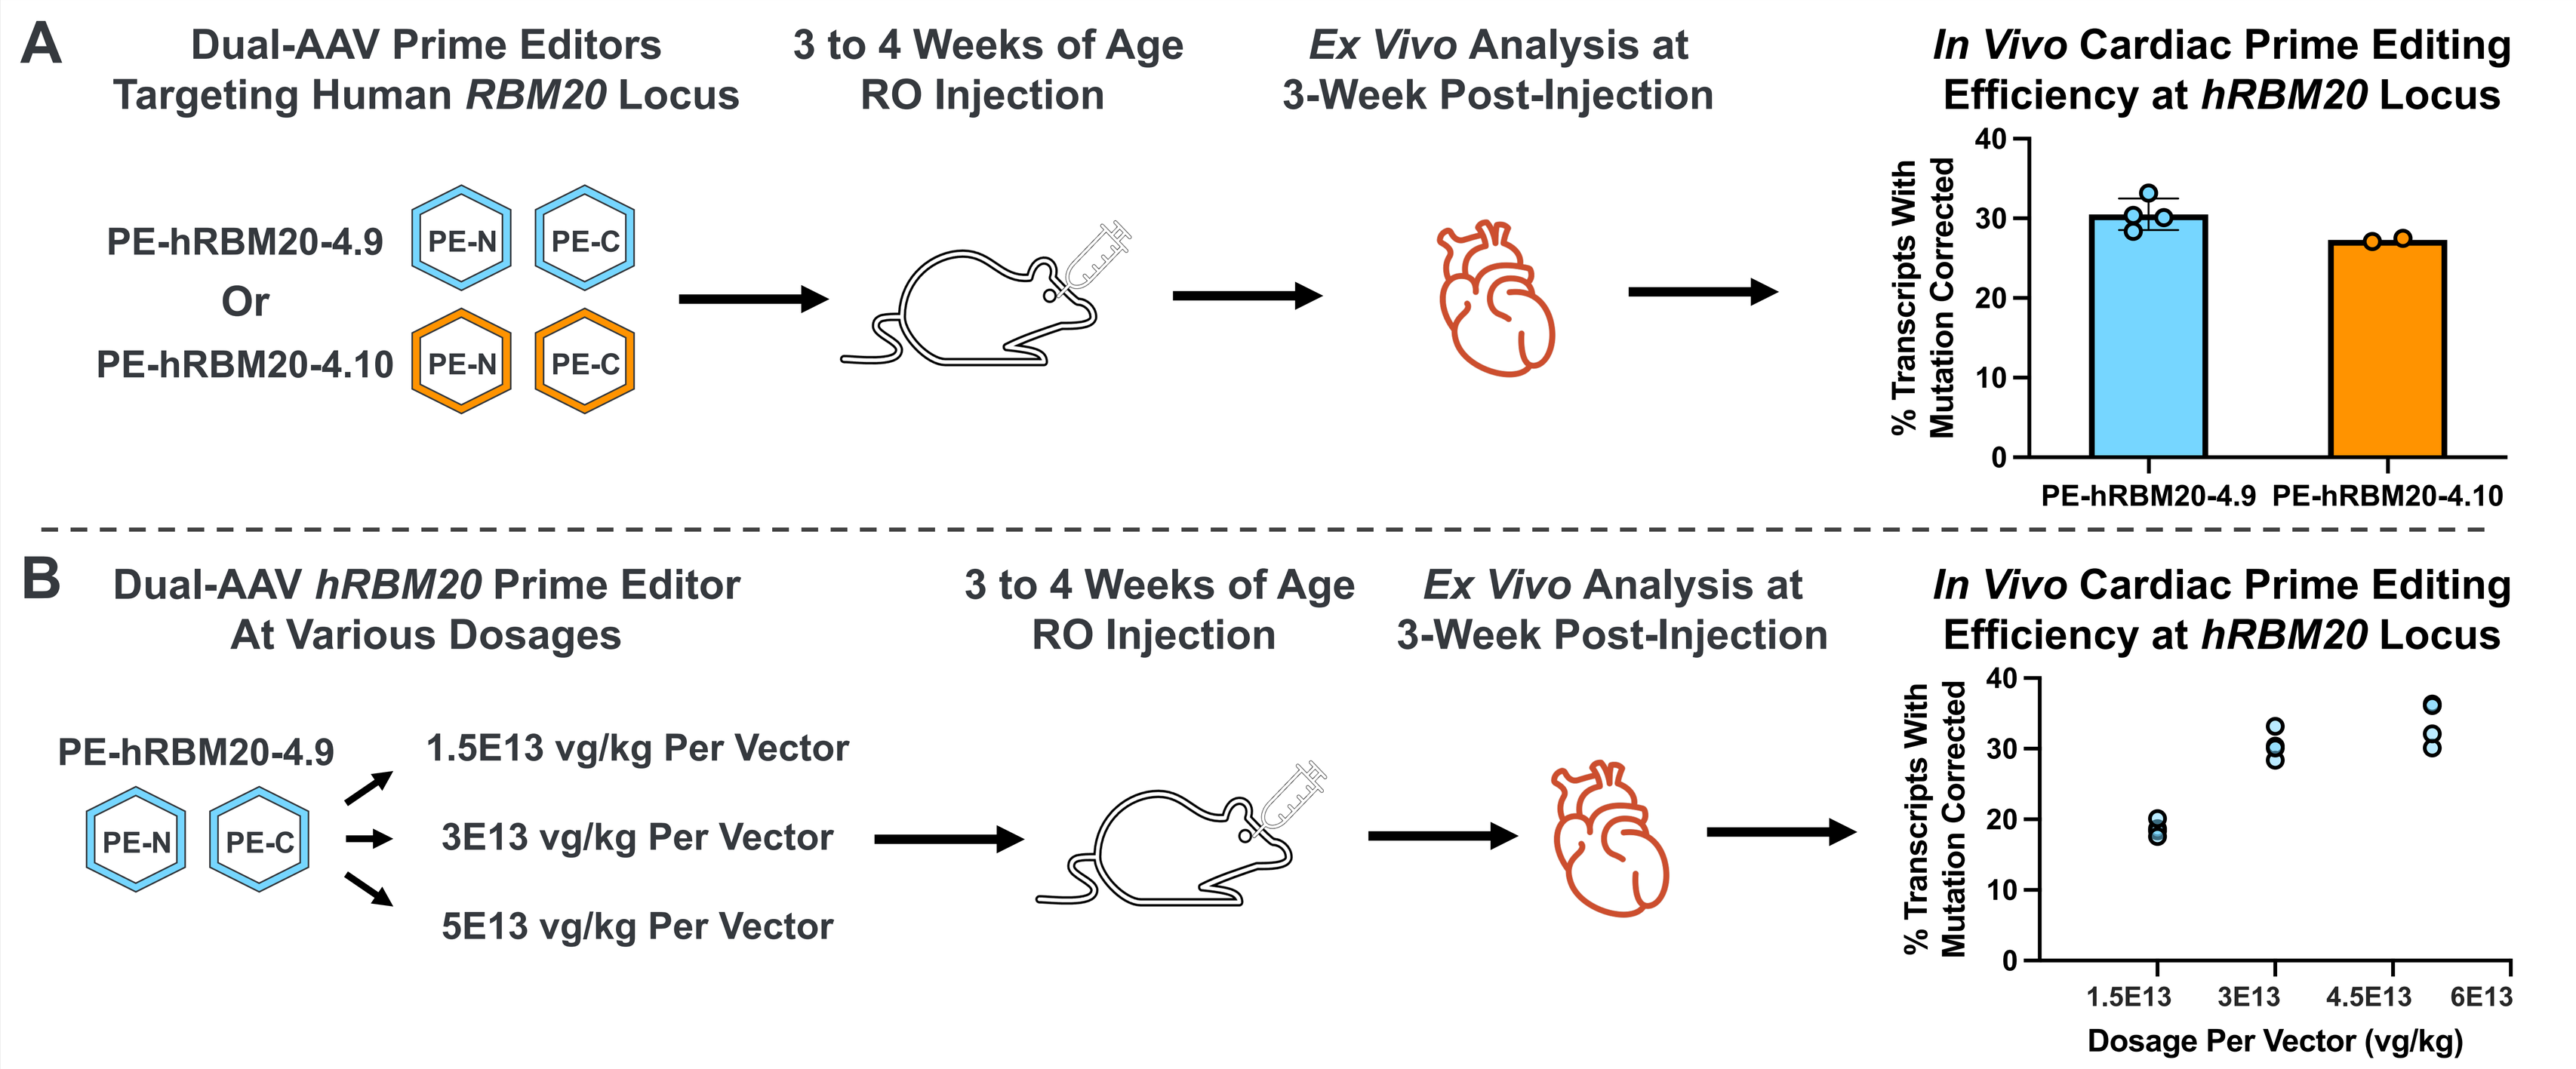
A
Dual-AAV Prime Editors Targeting Human *RBM20* Locus
3 to 4 Weeks of Age RO Injection
Ex Vivo Analysis at 3-Week Post-Injection
In Vivo Cardiac Prime Editing Efficiency at *hRBM20* Locus
PE-hRBM20-4.9
Or
PE-hRBM20-4.10
PE-N PE-C
% Transcripts With Mutation Corrected
B
Dual-AAV *hRBM20* Prime Editor At Various Dosages
1.5E13 vg/kg Per Vector
3E13 vg/kg Per Vector
5E13 vg/kg Per Vector
1.5E13 3E13 4.5E13 6E13
Dosage Per Vector (vg/kg)

Figure 6. Characterization of human *RBM20* targeting prime editing therapeutics in humanized *RBM20 R634Q* mice. (A) Different versions of prime editors or (B) different dosages of prime editor were administered systemically to humanized *RBM20 R634Q* mice. At 3-weeks post-injection, editing of the humanized allele was assessed at RNA transcript level.

CONCLUSIONS

► We constructed a novel mouse DCM model with humanized *RBM20* sequences, exhibiting expected heart function and gene-splicing phenotypes of *RBM20* cardiomyopathy.

► Our humanized *RBM20* mouse model provides a platform for rigorous *in vivo* validation of prime editing, facilitating the translation of this precise gene correction technique into a clinically viable therapeutic for RBM20-related dilated cardiomyopathy

Reference: *1. Nishiyama, Takahiko, et al. "Precise genomic editing of pathogenic mutations in RBM20 rescues dilated cardiomyopathy." Science Translational Medicine 14.672 (2022): eade1633.*