Engineering Novel AAV Capsids for Cardiac Gene Delivery

Ze Cheng, Emilee Easter, **Lindsey M. Rollosson**, Lynn Yamamoto, Karl Doerner, JianMin Lin, Timothy Hoey, Kathryn N. Ivey , Laura M. Lombardi

Tenaya Therapeutics, Inc. South San Francisco, CA Correspondence: Ze Cheng (zcheng@tenayathera.com)

Introduction

▶ We have been focusing on the genetics associated with conditions affecting the heart muscle, also known as cardiomyopathies, that can lead to heart failure. We are discovering and developing therapeutics that target the disease biology underlying cardiomyopathies using adeno-associated virus (AAV) vectors to deliver genetic medicines, including gene therapy, gene editing, and gene silencing. Our AAV-based programs require efficient cardiomyocyte targeting via systemic administration of therapeutic DNA encapsulated in AAV vectors, as superior cardiomyocyte targeting can lead to better therapeutic efficacy and/or lower dose requirement.

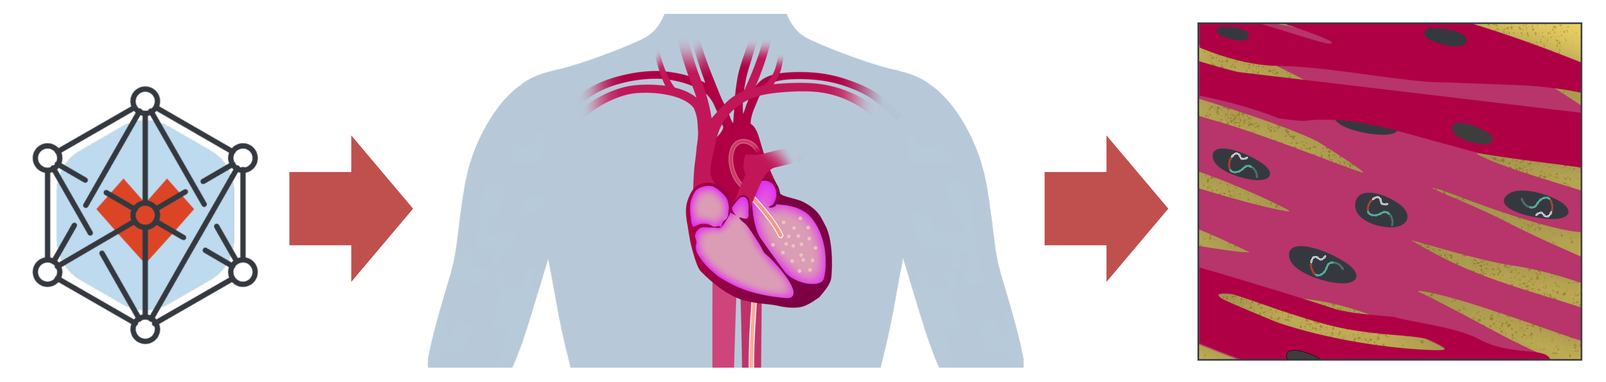

AAV-Based Cardiac Therapy:

Deliver healthy copies of genes or other therapeutic payloads to cardiomyocytes through systemic administration.

Figure 1. Tenaya AAV-based cardiac therapy platform. Our programs target heart conditions due to pathogenic mutations and seek to restore cell function by using AAV vectors to deliver healthy copies of genes or other therapeutic payloads, such as gene editors. Natural AAV serotypes often require high doses to achieve therapeutically meaningful transgene expression in cardiomyocytes following systemic administration, resulting in high manufacturing costs and increased potential for dose-related adverse events. Thus, it is critical to discover novel AAV variants with improved cardiomyocyte targeting *in vivo*.

Tenaya Selects AAV9 as the Starting Serotype of Capsid Engineering Due to its Superior *In Vivo* Cardiomyocyte Transgene Expression Relative to Other Serotypes

▶ Several natural AAV serotypes, including AAV9, AAVrh.10, and AAVrh.74, can transduce cardiomyocytes *in vivo* and are being advanced in cardiac gene therapy programs. We compared cardiomyocyte targeting efficiency of AAV9, AAVrh.10, and AAVrh.74 in murine and non-human primate (NHP) models. Our results demonstrate that AAV9 more efficiently expresses transgene in cardiomyocytes following systemic administration compared to AAVrh.10 and AAVrh.74, supporting AAV9 as the starting serotype of capsid engineering.

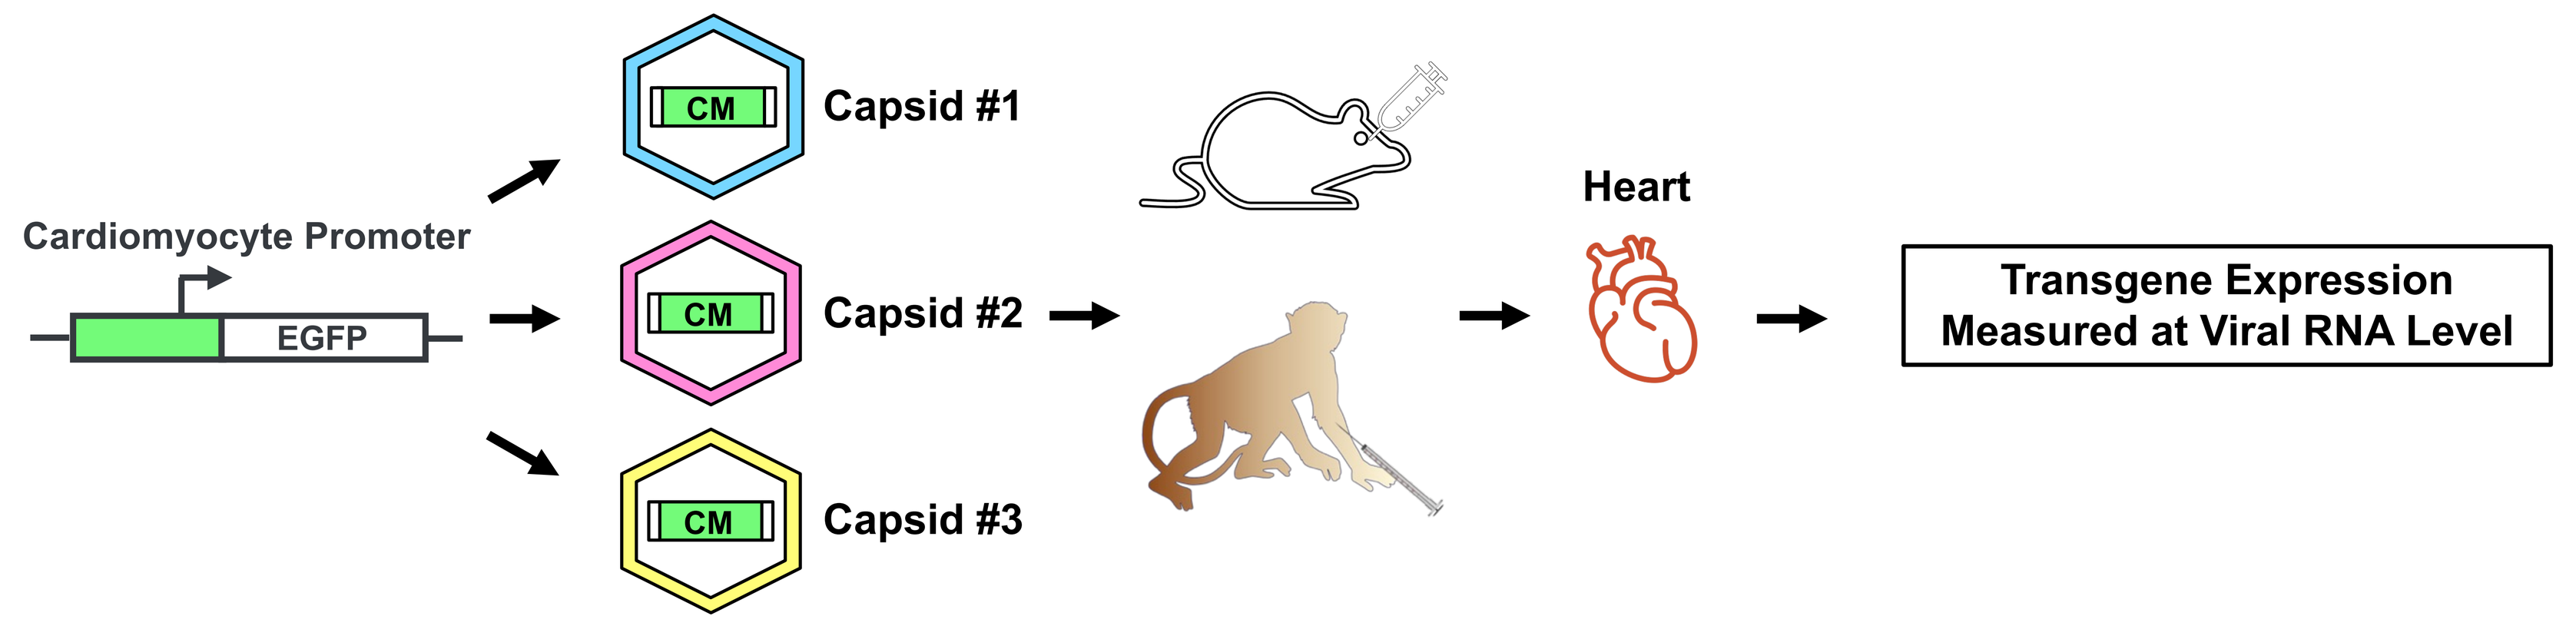
Cardiomyocyte Promoter
EGFP
CM
Capsid #1
Capsid #2
Capsid #3
Heart
Transgene Expression Measured at Viral RNA Level

Figure 2. Comparing cardiomyocyte targeting efficiency of AAV capsids *in vivo*. Multiple cell types reside in the heart and may be transduced by AAV. To specifically detect AAV-mediated expression in cardiomyocytes, we designed a cardiomyocyte transgene expression reporter (referred to as "CM" in the figure) by coupling a cardiomyocyte-specific promoter with EGFP coding sequence. This reporter was packaged in various AAV capsids and administered to mice and NHPs systemically. This approach enables *in vivo* comparison of cardiomyocyte targeting mediated by different AAV capsids, while minimizing the interference from other cell types in the heart.

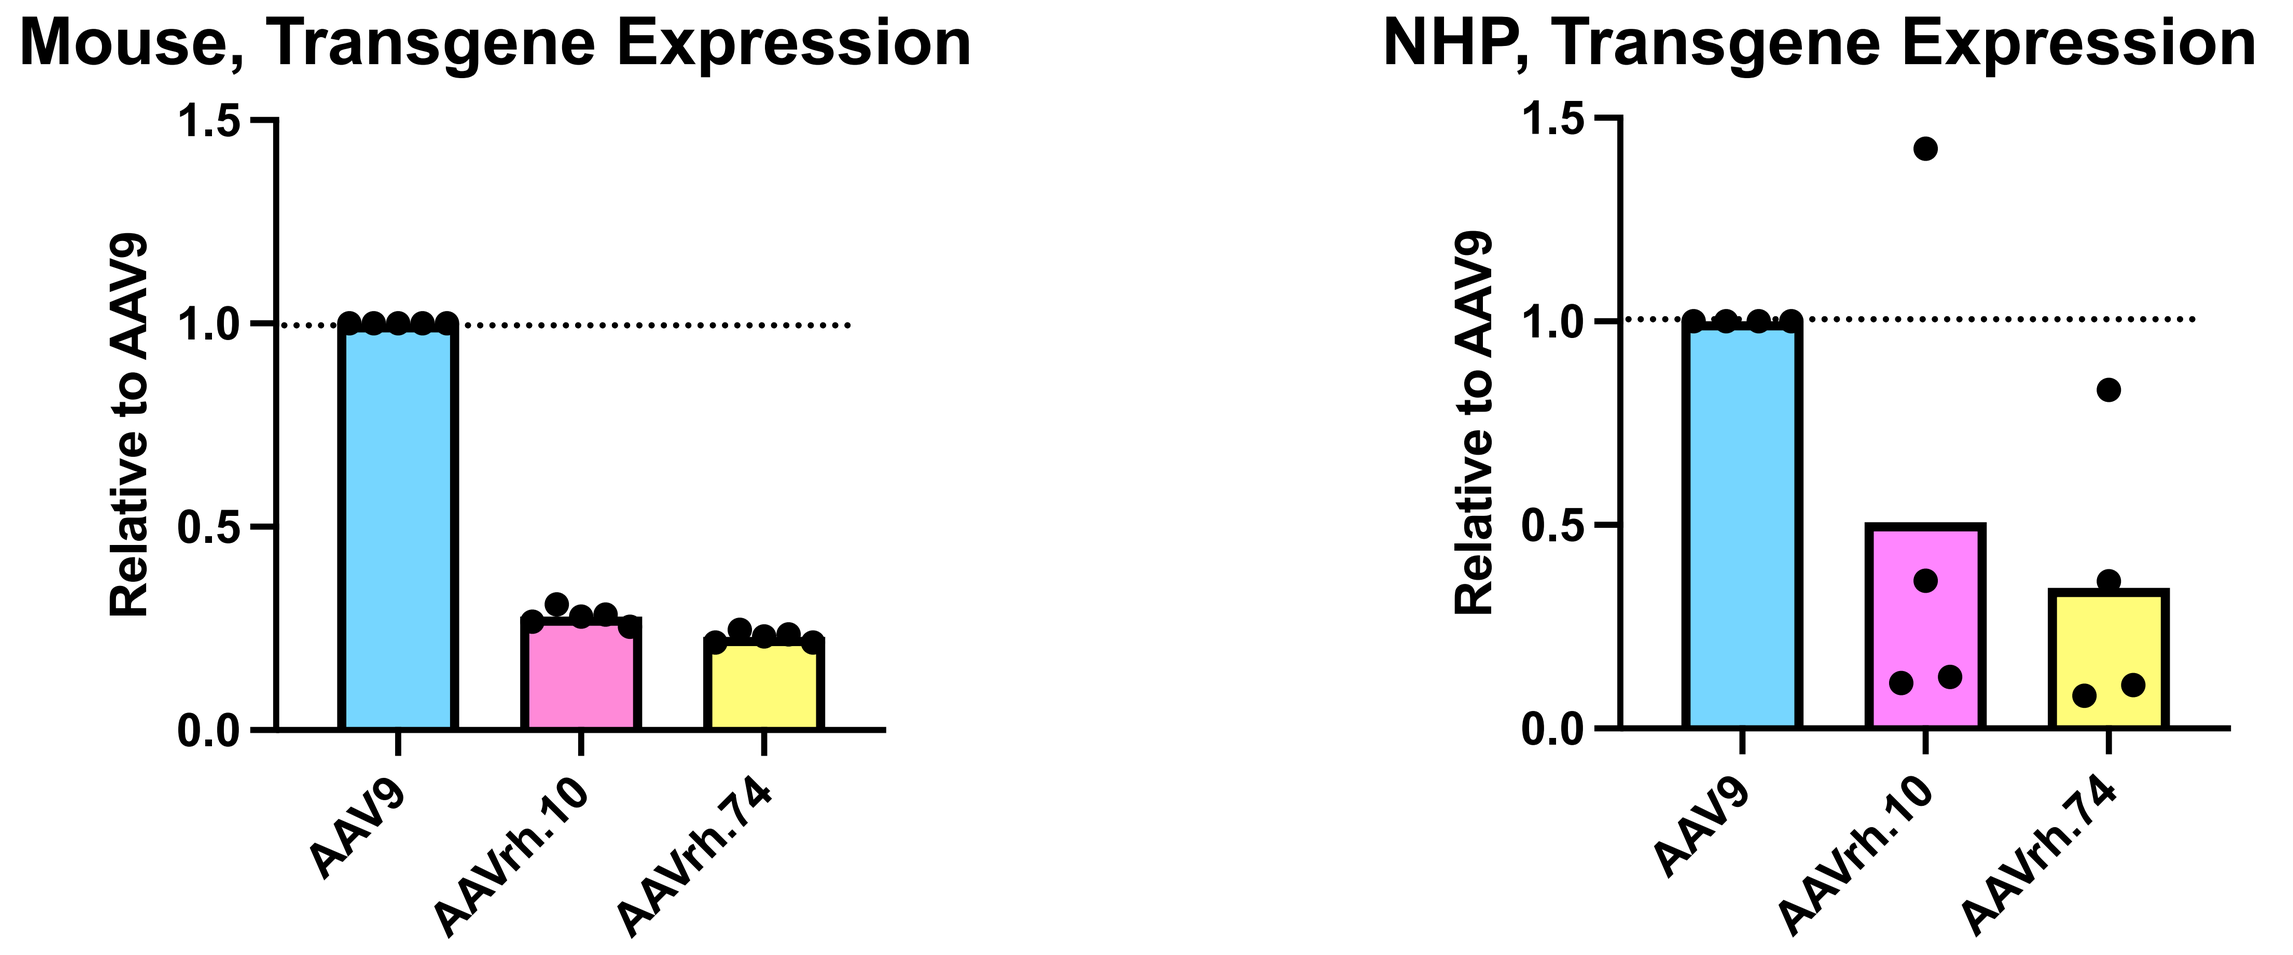
Mouse, Transgene Expression
NHP, Transgene Expression
Relative to AAV9
1.5
1.0
0.5
0.0
AAV9
AAVrh.10
AAVrh.74

Figure 3. AAV9 mediates superior cardiomyocyte transgene expression in mice and NHPs. AAV9, AAVrh.10, and AAVrh.74 capsids carrying a barcoded cardiomyocyte reporter transgene were pooled and administered to mice and Cynomolgus monkeys systemically. AAV9 generates higher expression level of cardiomyocyte-specific reporter RNA transcripts than the other two serotypes manufactured in parallel on the same platform. 5 mice and 4 NHP animals were enrolled.

Tenaya's Capsid Engineering Platform Combines High Throughput *In Vivo* Experimental Screening and Various *In Silico* Design Methods

▶ While we have previously conducted capsid engineering efforts utilizing solely wet-lab methods and have identified novel capsids with improved properties relative to wildtype AAV serotypes, we aimed at further enhancing cardiomyocyte targeting by introducing more modifications to larger and/or more regions on the capsid protein. However, traditional methods struggle with the increased number of modified positions, due to various constraints. Thus, we employed various *in silico* methods to extract information from large *in vivo* functional datasets, model the functional landscape, navigate through large sequence spaces, and design new capsid variants for subsequent functional testing and candidate selection.

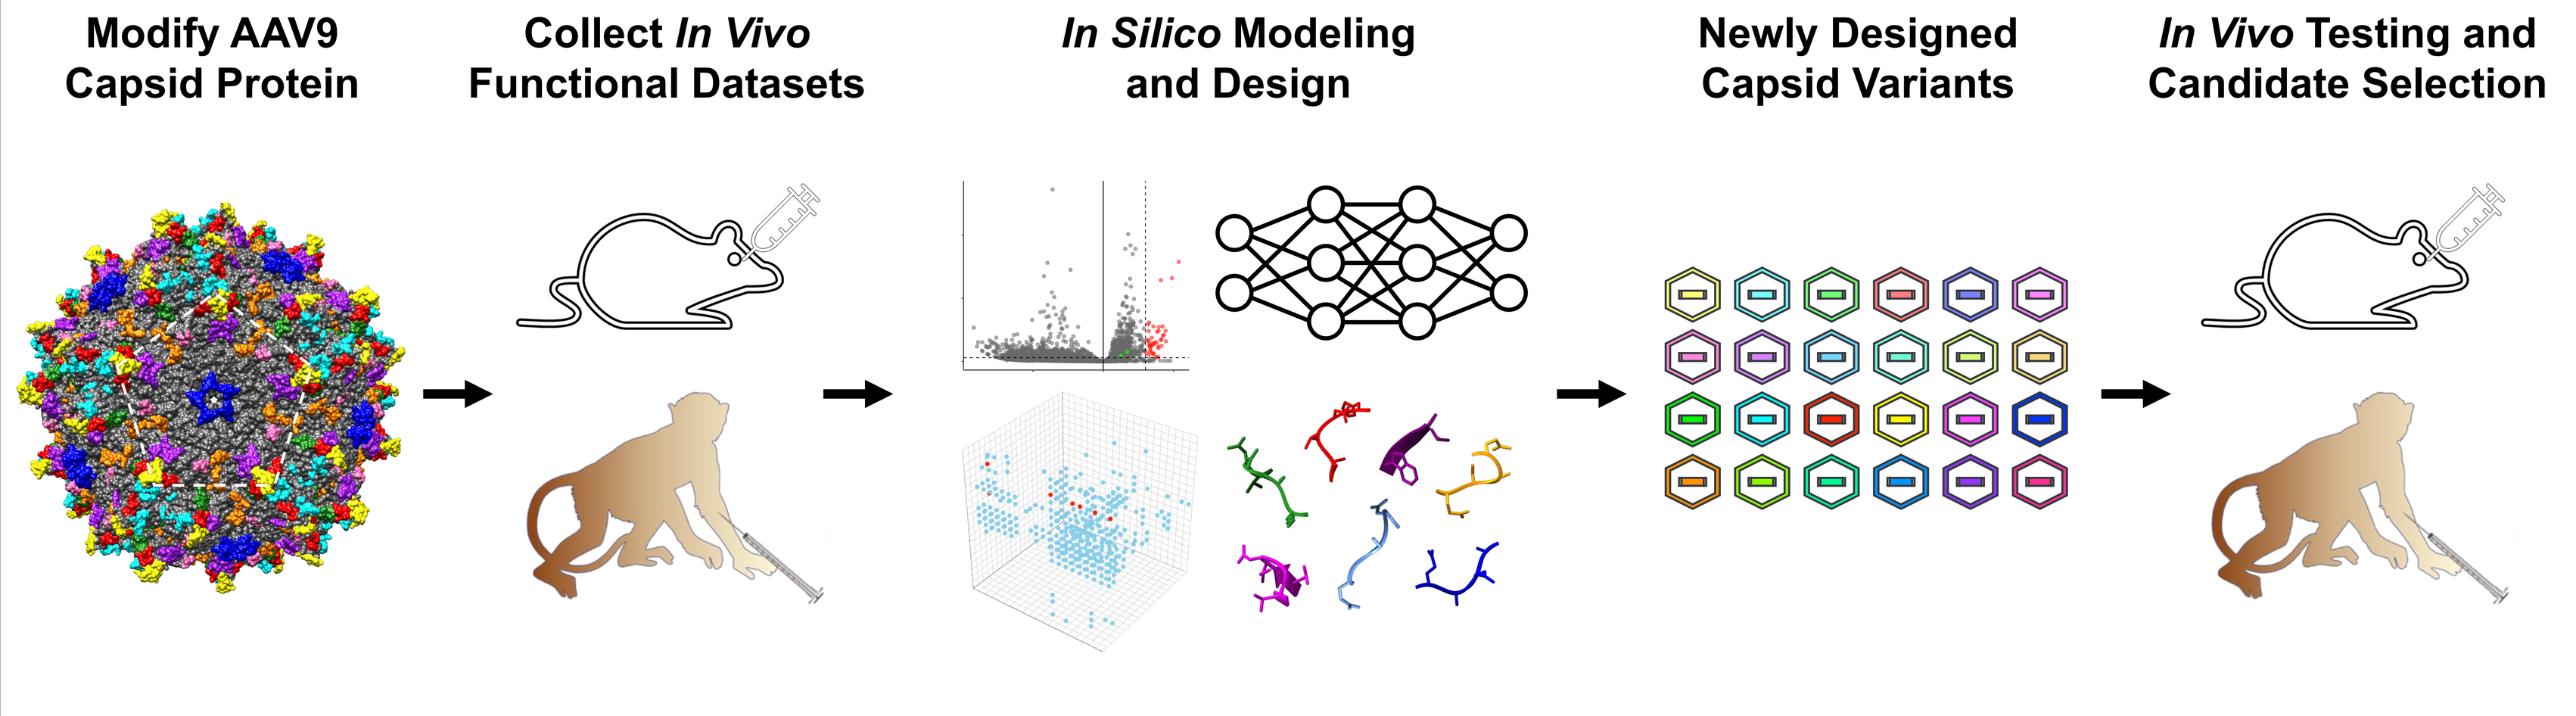
Modify AAV9 Capsid Protein
Collect In Vivo Functional Datasets
In Silico Modeling and Design
Newly Designed Capsid Variants
In Vivo Testing and Candidate Selection

Figure 4. Developing novel AAV capsids with enhanced *in vivo* cardiomyocyte targeting. Starting from wildtype AAV9, we introduced various modifications to the capsid protein and collected *in vivo* cardiomyocyte transgene expression data for thousands of capsid variants. With these datasets as input, we developed multiple *in silico* methods, including computational functional and sequence analysis, human-directed design approaches, and machine-learning based approaches, to extract sequence-to-fitness information, model the functional landscape, and design thousands of new variants that may outperform the wildtype, as well as the best-performing input variants. The newly designed variants were manufactured and tested in subsequent *in vivo* studies. Promising candidates were identified and selected for further characterization and validation.

Tenaya Novel Capsid Candidates Show Superior Performance in Both Non-Human Primates and Mice

▶ We further characterized the performance of our novel capsid candidates in an individually-packaged, pooled-injected study that enrolled both NHPs and mice. Multiple candidates show superior *in vivo* cardiomyocyte transgene expression in NHPs and cross-species consistency between NHPs and mice.

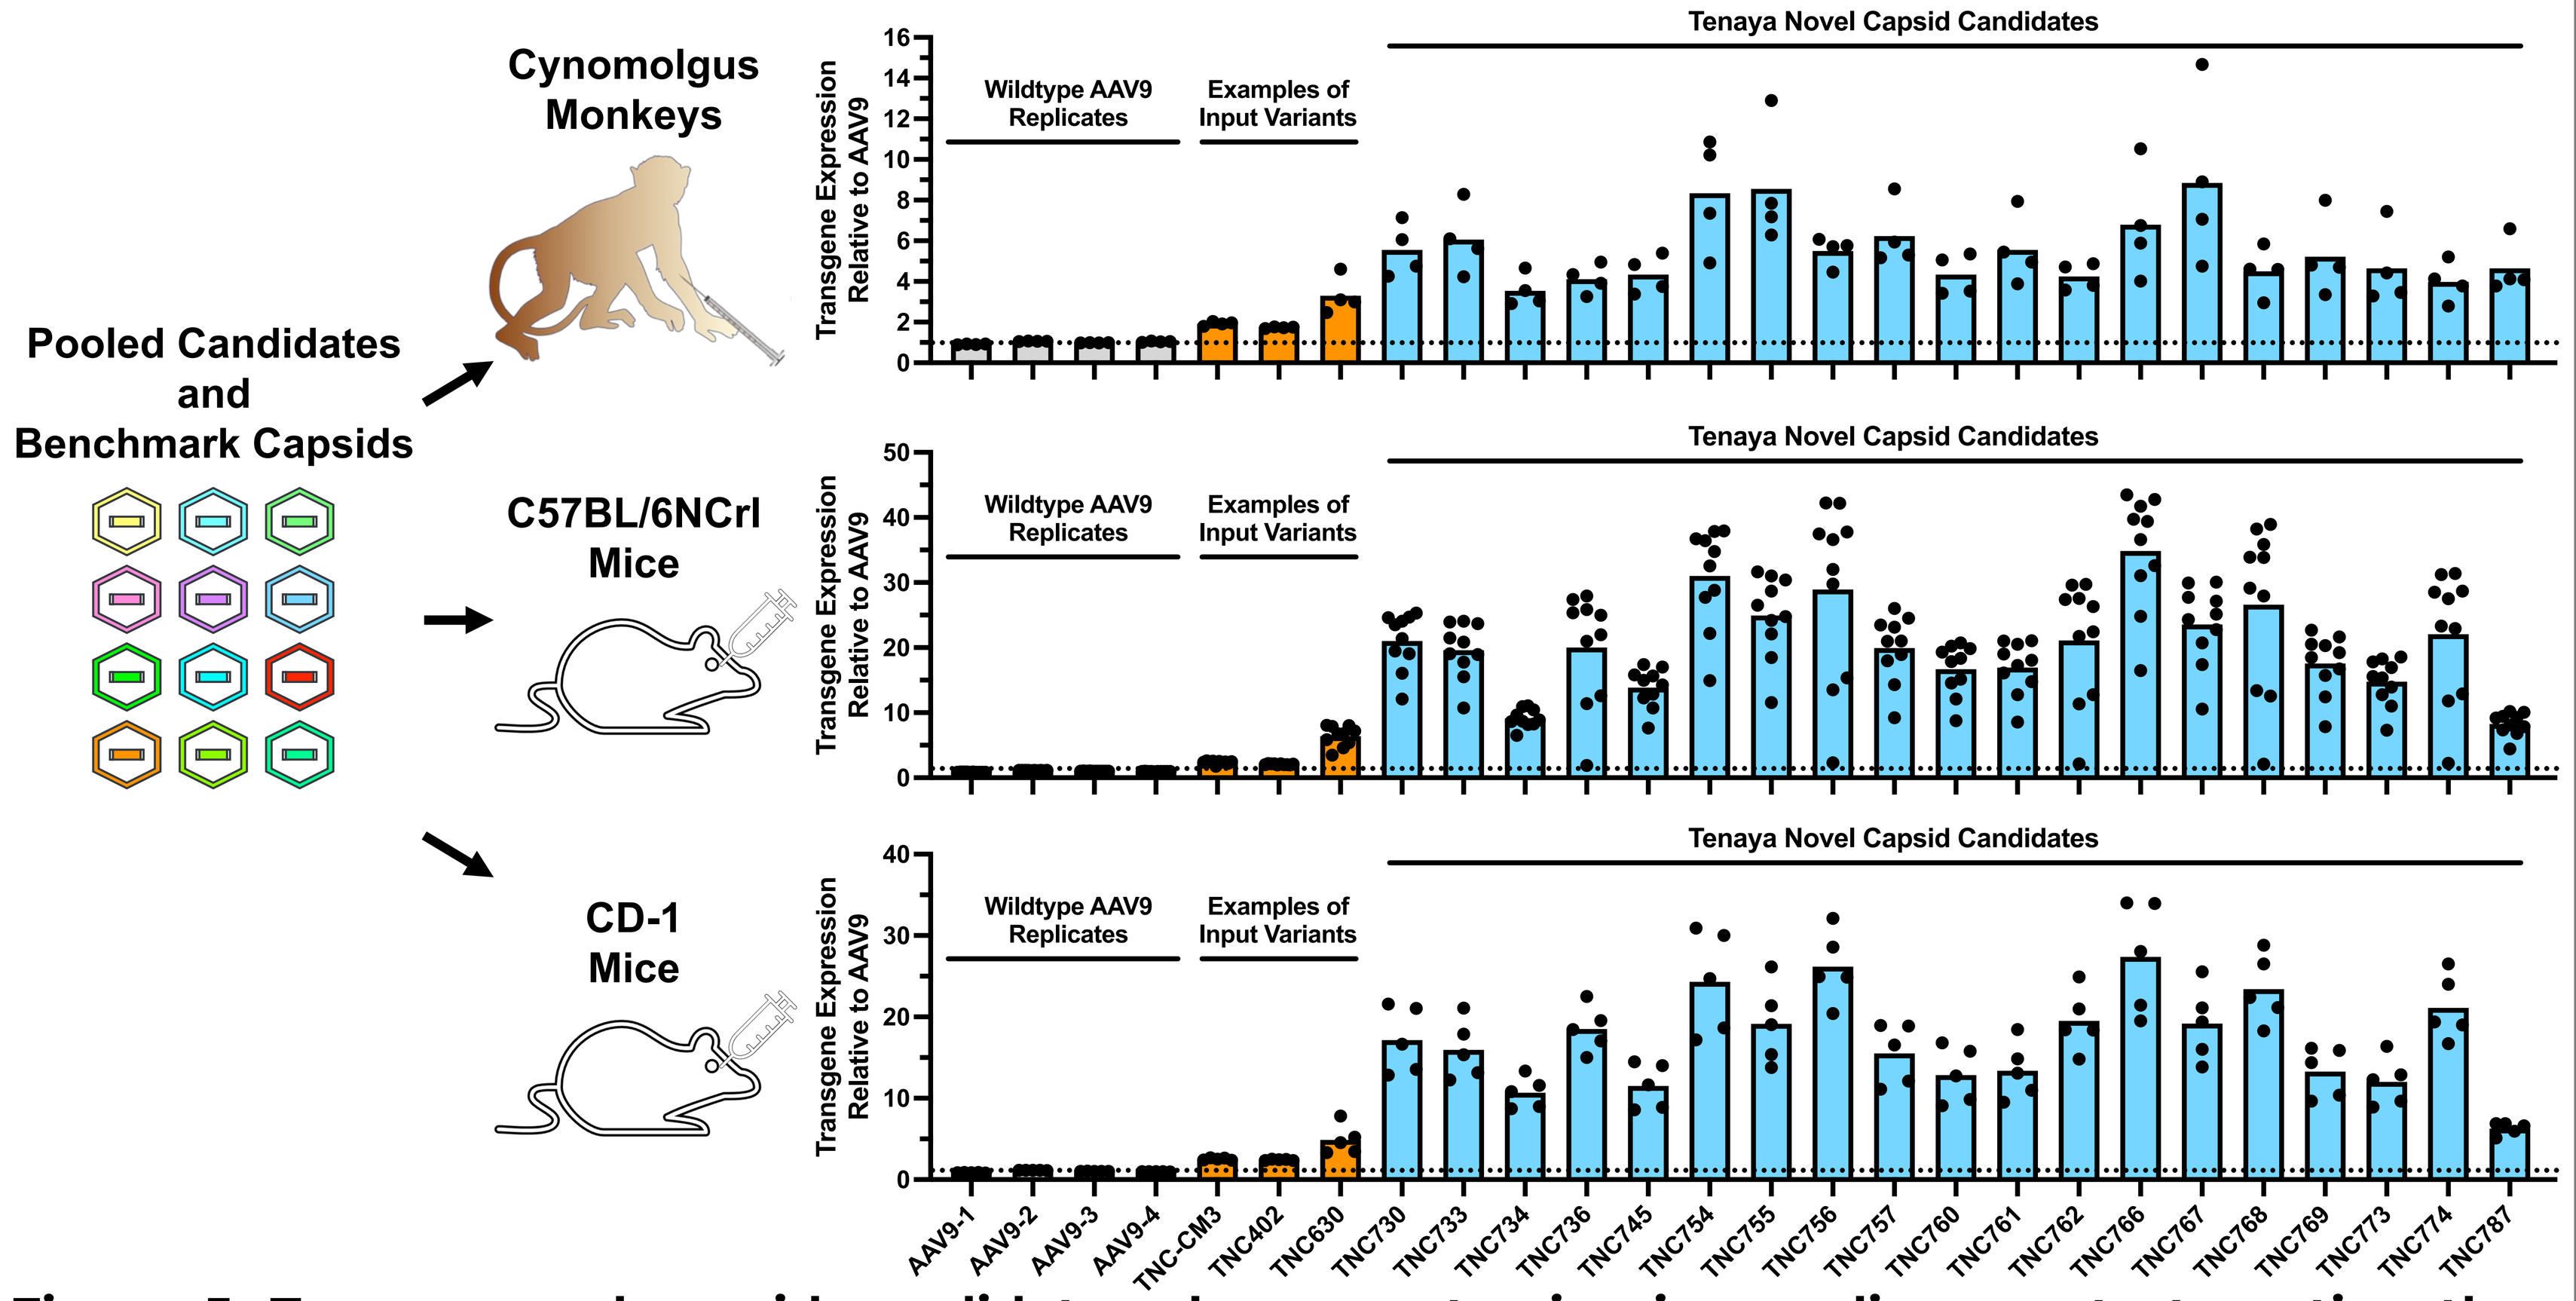
Pooled Candidates and Benchmark Capsids
Cynomolgus Monkeys
C57BL/6NCrl Mice
CD-1 Mice
Transgene Expression Relative to AAV9
Wildtype AAV9 Replicates
Examples of Input Variants
Tenaya Novel Capsid Candidates
AAV9-1
AAV9-2
AAV9-3
AAV9-4
TNC-CM3
TNC402
TNC630
TNC730
TNC733
TNC734
TNC736
TNC745
TNC754
TNC755
TNC756
TNC757
TNC760
TNC761
TNC762
TNC766
TNC767
TNC768
TNC769
TNC773
TNC774
TNC787

Figure 5. Tenaya novel capsid candidates show greater *in vivo* cardiomyocyte targeting than benchmark capsids in both NHPs and mice. Novel capsids and benchmark capsids were manufactured individually, encapsulating a barcoded cardiomyocyte expression reporter transgene. The resulting viruses were pooled and administered systemically to four Cynomolgus monkeys, ten C57BL/6NCrl mice, and five CD-1 mice. Transgene RNA expression was measured at 3 to 4 weeks post-injection by next-generation sequencing (NGS) based analysis. Each dot in this figure represents one animal.

Individually Dosed Validation in Non-Human Primates Confirms TNC755's Superior Cardiomyocyte Targeting

▶ To validate observations from the pooled capsid comparison study, we tested one of our candidates, TNC755, as a proof of principle against AAV9 in an individually dosed NHP study at a clinically relevant dosage. The TNC755-dosed animal shows stronger cardiomyocyte transgene expression and a higher percentage of transgene expression positive cardiomyocytes compared to the AAV9-dosed animal, validating TNC755's superior *in vivo* cardiomyocyte targeting.

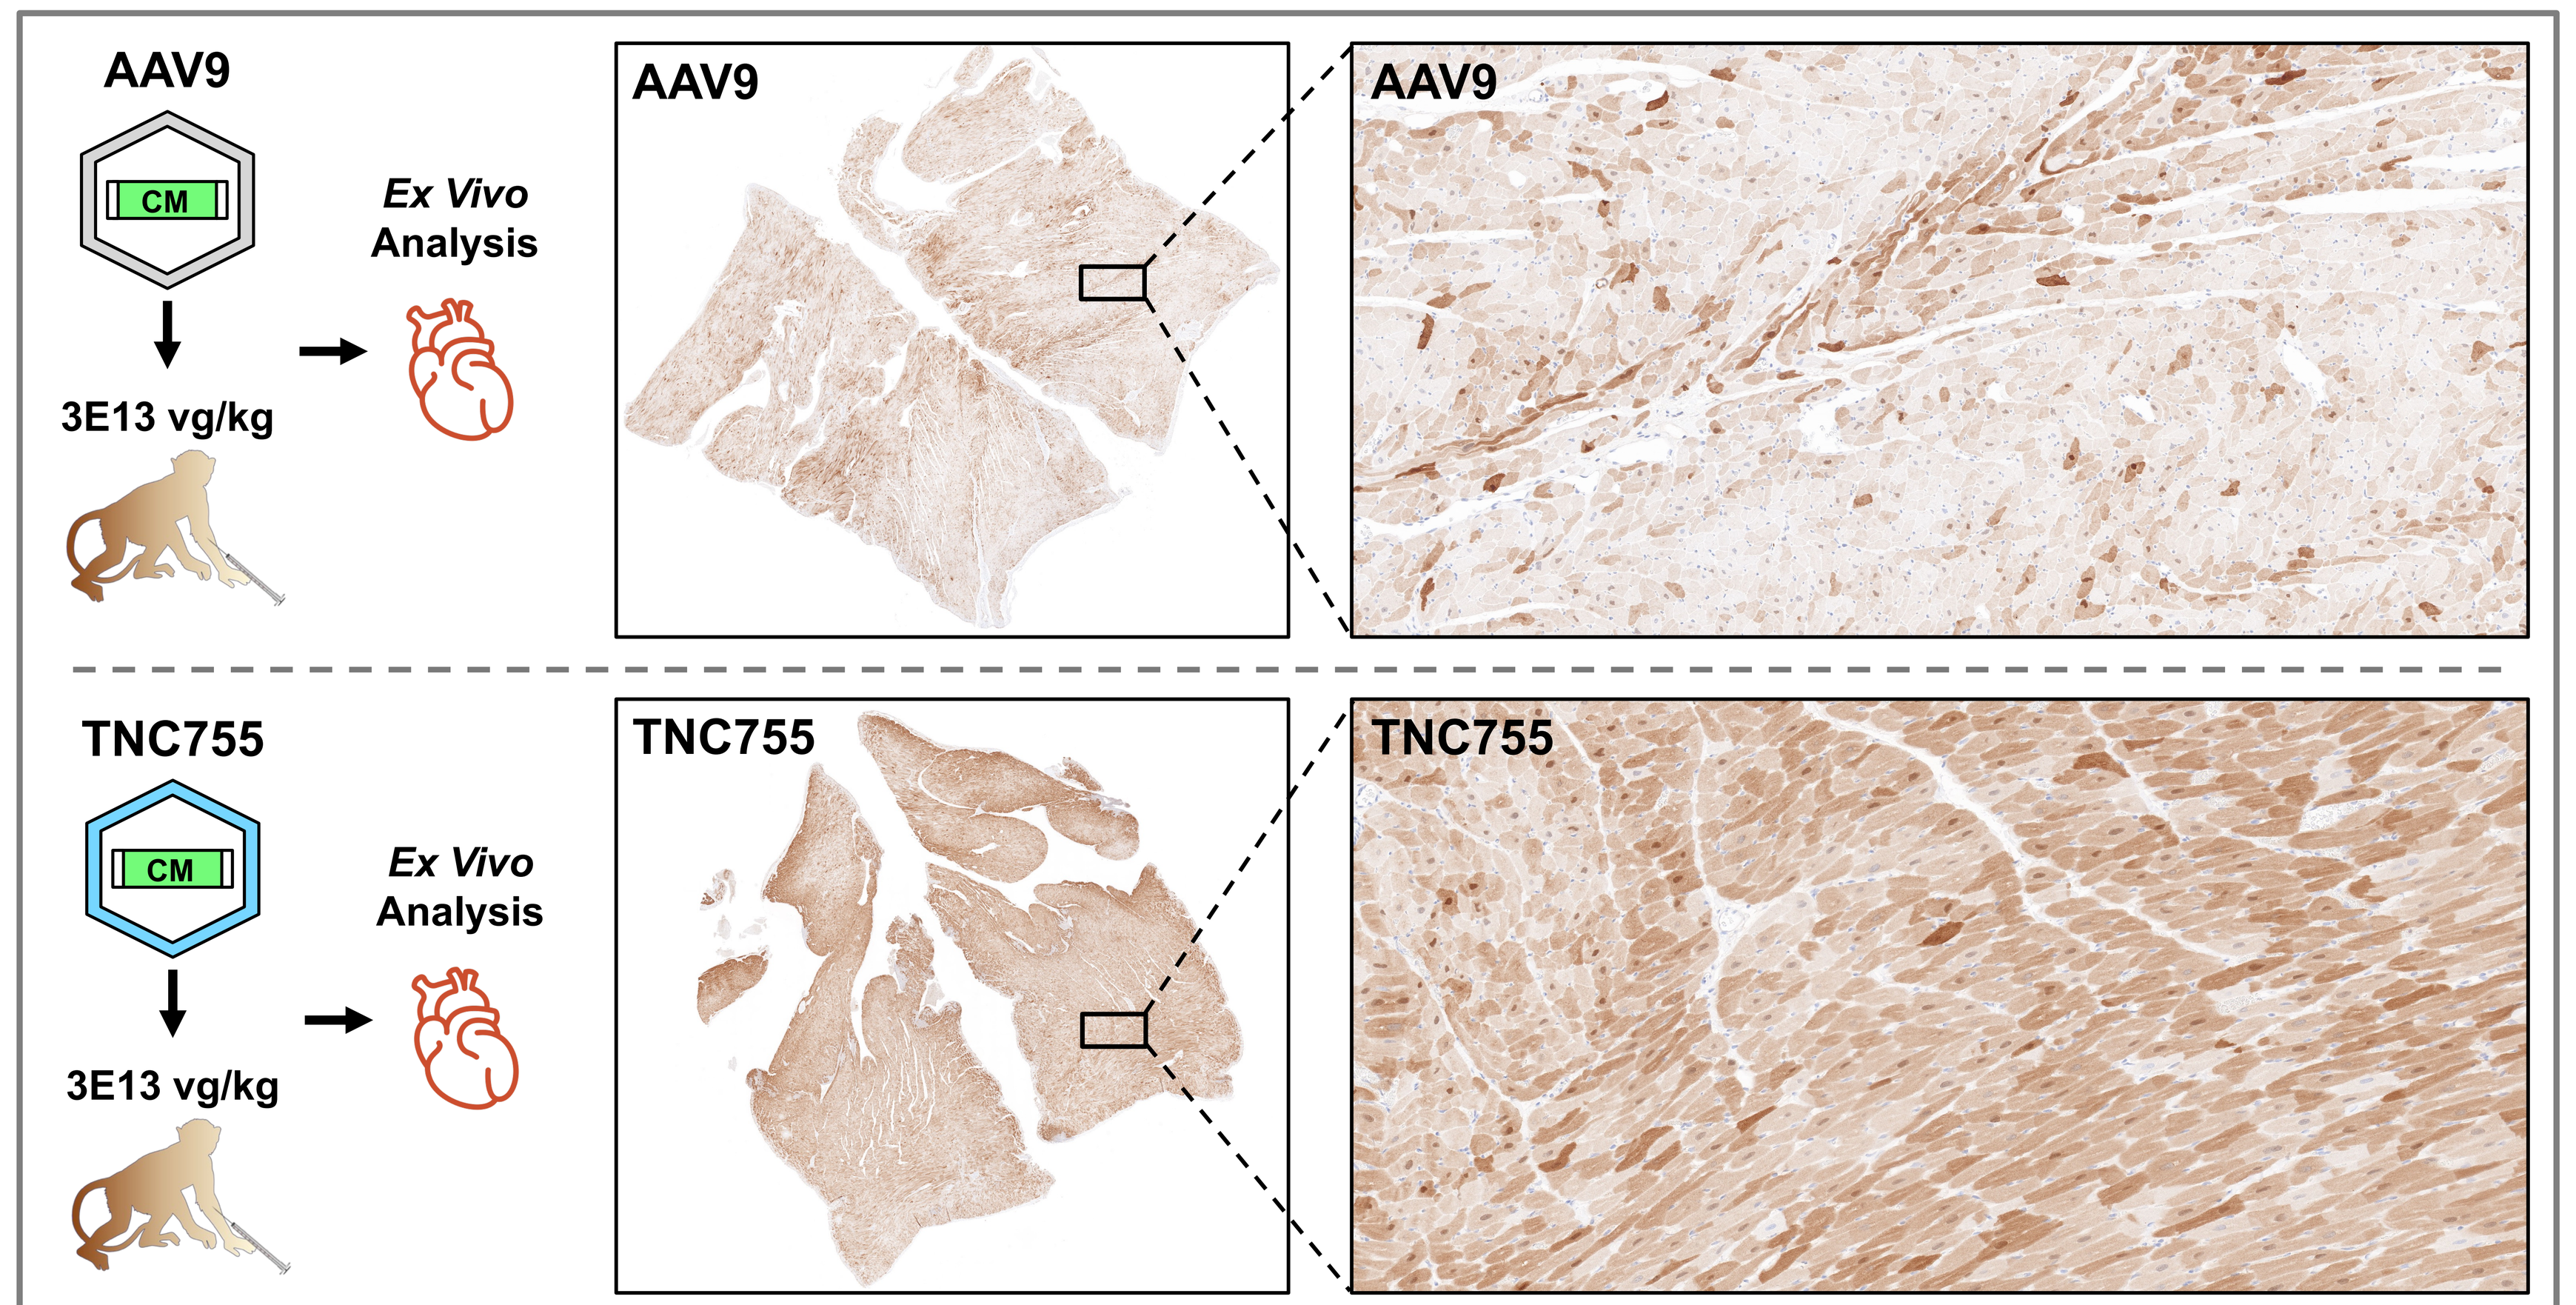
AAV9
CM
3E13 vg/kg
Ex Vivo Analysis
TNC755

Figure 6. TNC755 shows greater cardiomyocyte targeting efficiency in NHPs compared to AAV9 post individual dosing. Cynomolgus monkeys were systemically dosed with AAV9 or TNC755 encapsulating a barcoded cardiomyocyte expression reporter transgene at 3E13 vg/kg. At 4 weeks post-injection, immunohistochemistry was performed on heart tissue to detect transgene expression.

TNC755 Mediates Higher *In Vivo* Cardiac Gene Editing Efficiency Compared to AAV9

▶ Finally, we applied TNC755 to Tenaya's *in vivo* cardiac prime editing platform (Abstract #1028). TNC755 improves editing efficiency compared to AAV9, notably at mid-E12 vg/kg dosage where AAV9 is ineffective.

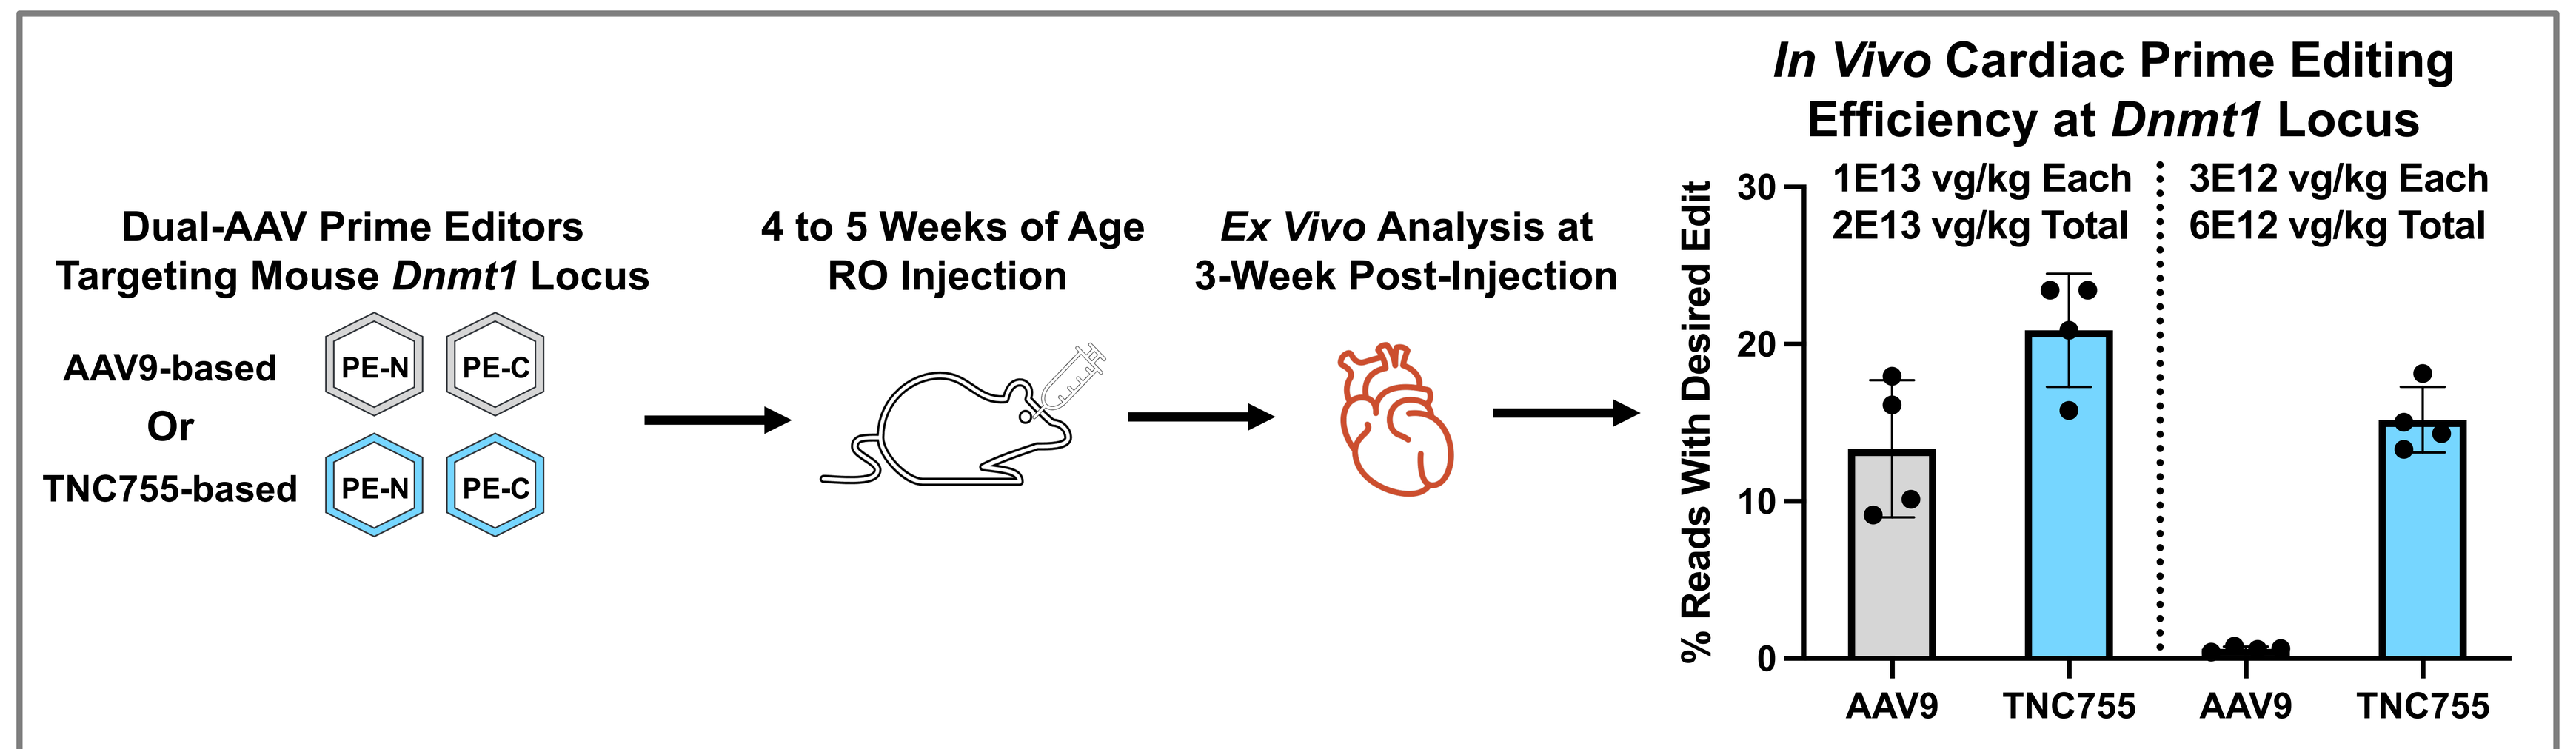
Dual-AAV Prime Editors Targeting Mouse Dnmt1 Locus
AAV9-based
Or
TNC755-based
PE-N
PE-C
4 to 5 Weeks of Age RO Injection
Ex Vivo Analysis at 3-Week Post-Injection
In Vivo Cardiac Prime Editing Efficiency at Dnmt1 Locus
1E13 vg/kg Each 2E13 vg/kg Total
3E12 vg/kg Each 6E12 vg/kg Total
% Reads With Desired Edit
30
20
10
0
AAV9
TNC755

Figure 7. TNC755 enhances *in vivo* cardiac prime editing efficiency at *Dnmt1* locus. We generated dual-AAV prime editors targeting the mouse *Dnmt1* locus encapsulated in AAV9 or TNC755, dosed mice systemically, and measured editing efficiency in the whole heart at 3 weeks post-injection. Each dot in the figure represents one animal. Our prime editor is cardiomyocyte-specific and among all genome copies in our whole heart DNA prep, around 22% are from cardiomyocytes (Abstract #1028). TNC755 edited almost all cardiomyocytes at 1E13 vg/kg each (2E13 vg/kg total) dosage. At 3E12 vg/kg each (6E12 vg/kg total), while AAV9 failed to edit, TNC755 still mediated efficient prime editing.

Conclusions

▶ We established a state-of-the-art capsid engineering platform that combines high throughput *in vivo* experimental screening and various *in silico* design methods.

▶ We identified multiple novel capsid candidates that show enhanced cardiomyocyte targeting efficiency following systemic delivery in both NHP and murine models.

▶ Our novel capsids may enable more efficacious and safer next generation gene therapies for cardiac disorders.