Efficacy of Cardiac Reprogramming via Gene Therapy in Rat with Chronic Heart Failure

Jin Yang[1], Aliya Zeng[1], Laura M. Lombardi[1], Huanyu Zhou[1], Christopher A. Reid[1], Emma Xu[1], Sam Elmojahid[1], Elena C. Leon[1], Tawny Neal Qureshi[1], Chetan Srinath[1], Zoe Duclos[1], Emily R. Nettesheim[1], Jun Liu[1], Samantha Jones[1], Charles Feathers[1], Tae Won Chung[1], Frank Jing[1], William S. Prince[1], Kathryn N. Ivey[1], Timothy Hoey[1], JianMin Lin[1]

[1] Tenaya Therapeutics, 171 Oyster Point Blvd, 500, South San Francisco, CA Email: jyang@tenayathera.com

TENAYA THERAPEUTICS

Background & Purpose

Myocardial infarction (MI) results in ischemic cell death of heart muscle cells caused by prolonged inadequate supply of oxygen-rich blood to the heart. Current therapies for heart failure after MI are limited and non-curative. In comparison to treatment immediately post-MI, there is a greater unmet medical need for patients with severe reduced ejection fraction (HFrEF) at the chronic stage post-MI. We have developed a reprogramming therapeutic in a single AAV expressing cardiac reprogramming factors that enables conversion of cardiac fibroblasts into functioning myocardial cells. In murine acute MI models, delivery of viral vectors at the injury site at the time of MI has shown improved cardiac function. It is of interest to determine whether the reprogramming approach can be efficacious in the setting of chronic MI where viral vector delivery occurs later, after the period of fibroblast proliferation and fibrotic scar formation.

Methods

- **Induction of myocardial infarct (MI)**: Sprague Dawley rats (Charles River Laboratories) at 8 to 12 weeks of age, were subjected to left anterior descending (LAD) coronary artery ligation via open heart surgery to induce myocardial infarction.
- **Vector delivery:** 2 weeks after MI, via a second open-chest surgery, the vectors were injected into three sites around the edge of the infarct region.
- **Identification of gene expression:** In distribution experiment, heart tissues were harvested 1 week after AAV5:GFP delivery at 2 weeks post-MI and subjected to IHC using a rabbit anti-GFP antibody.
- **Cardiac function measurement:** In efficacy study, Echocardiography and ECG were conducted at pre-MI, 2, 4, 6, 8, 10, 18, and 23 weeks post MI.

Results

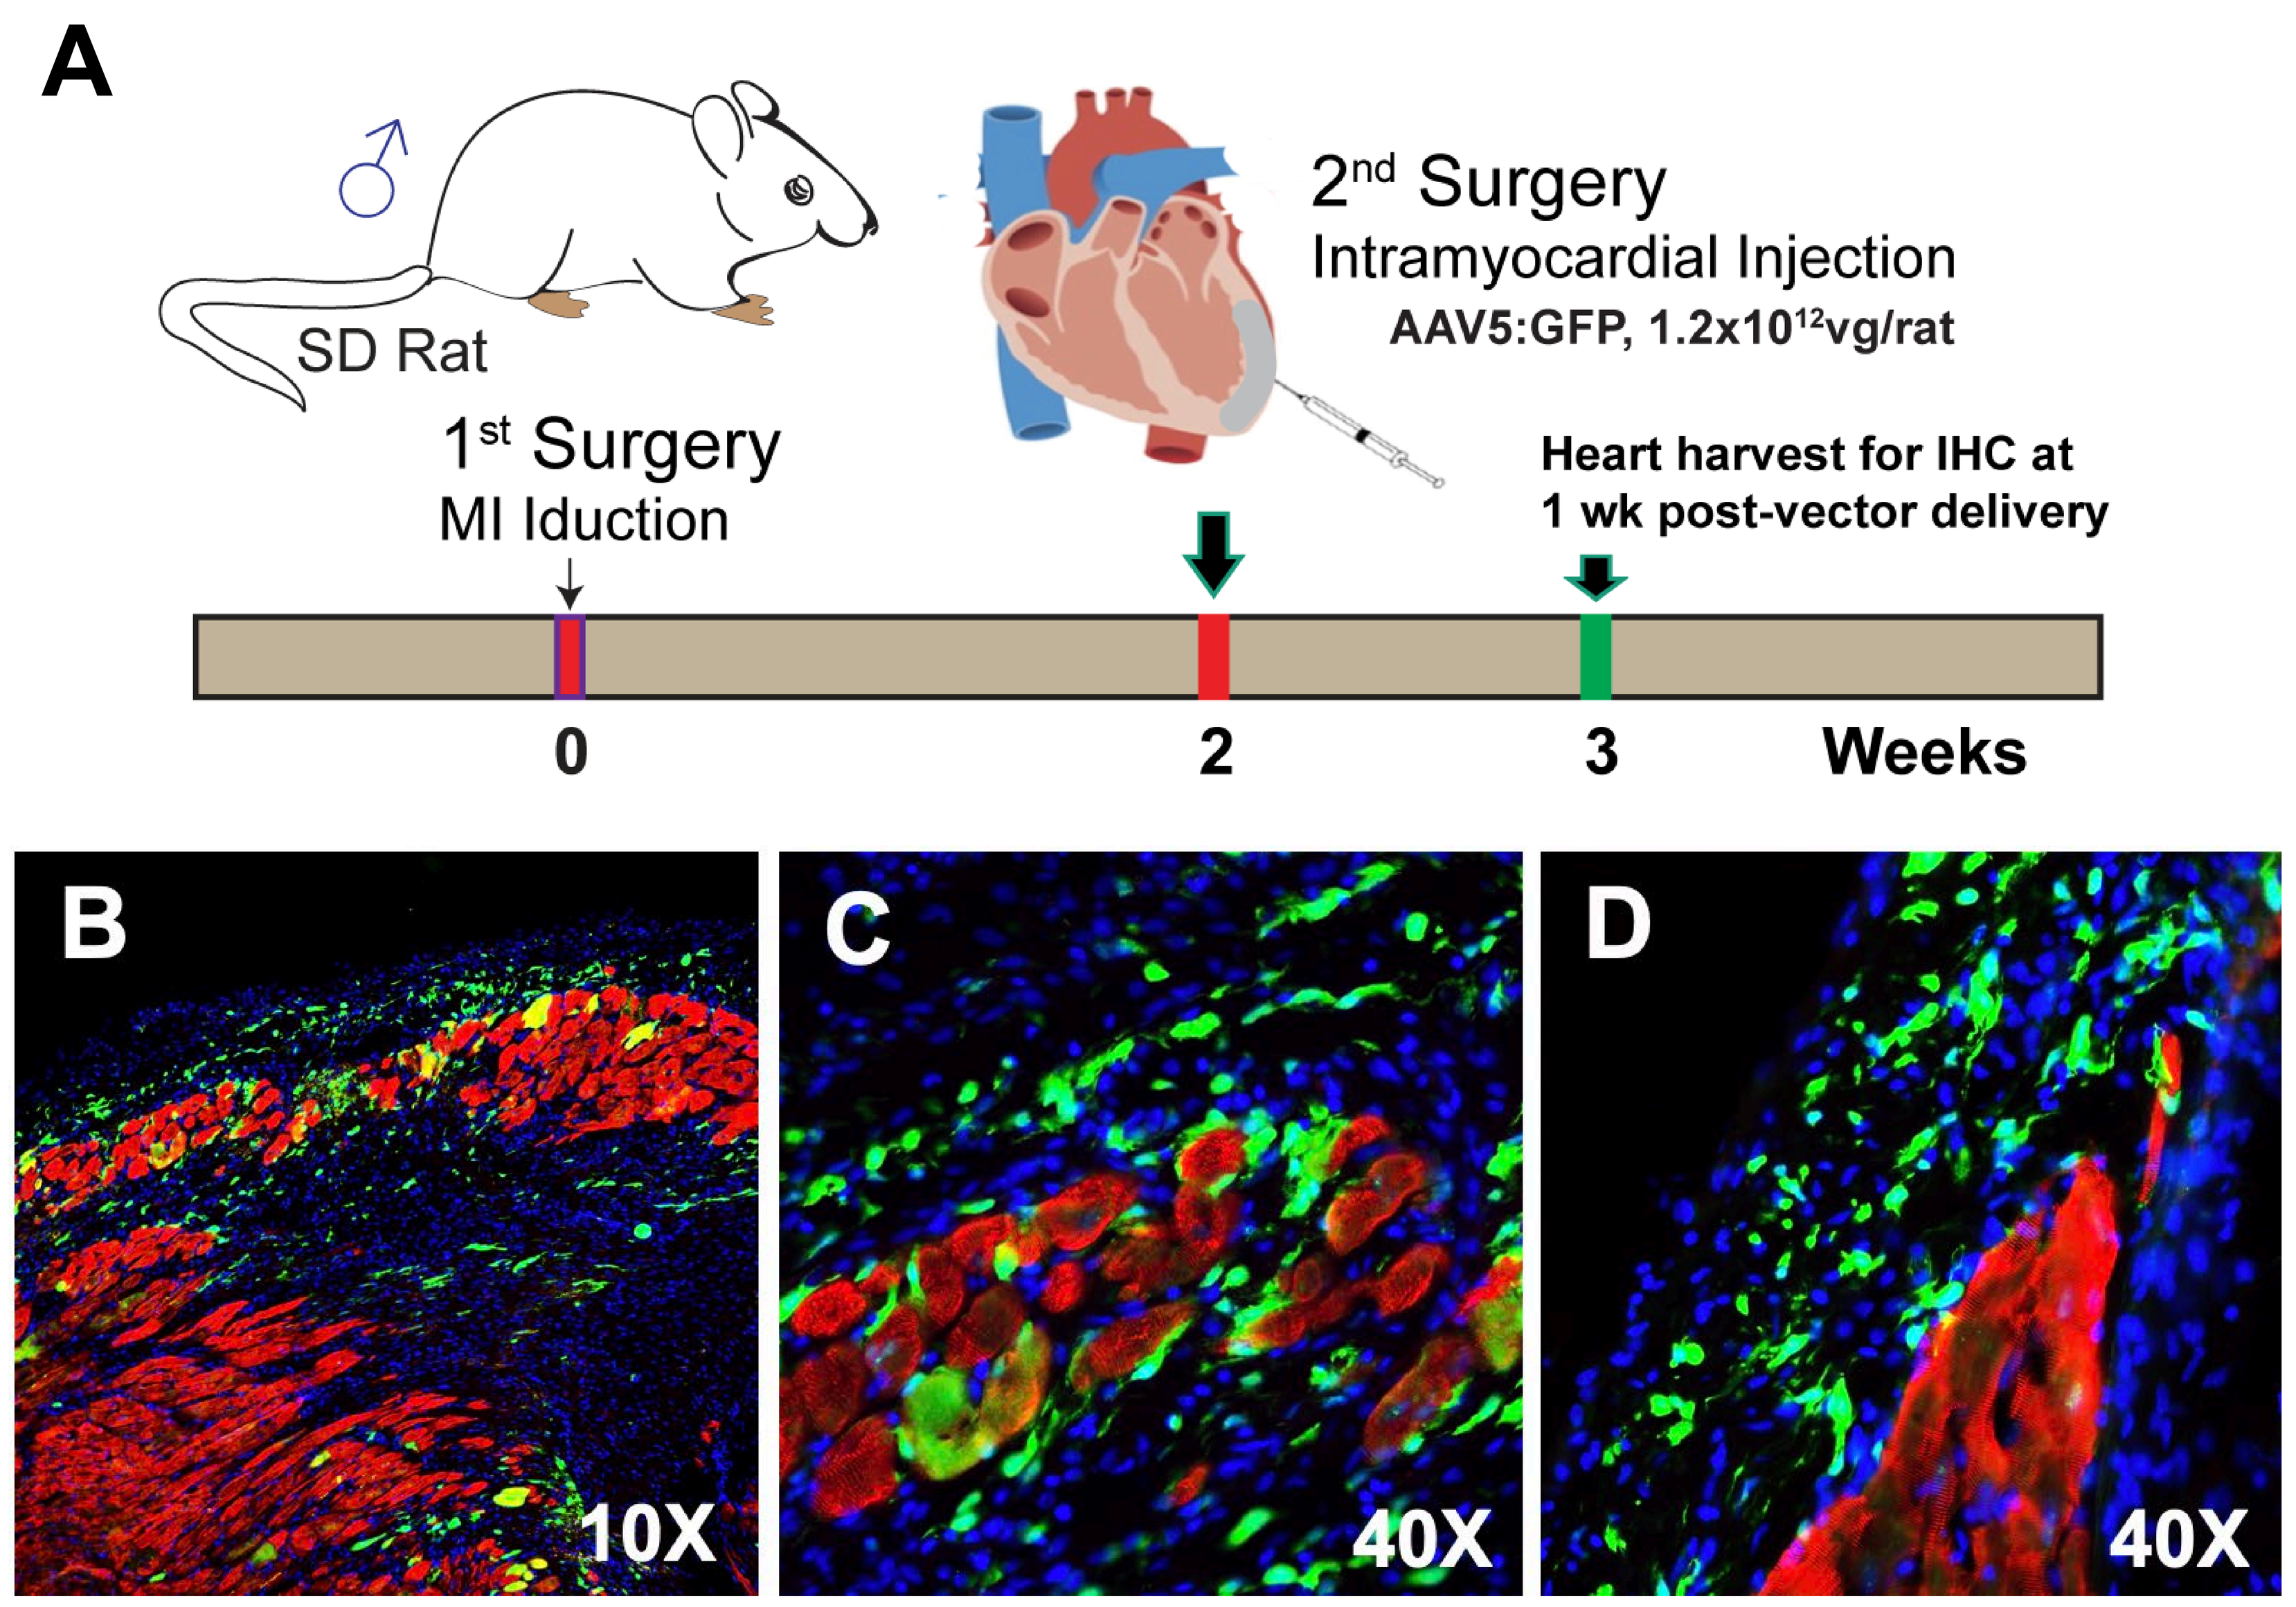
A
SD Rat
1st Surgery
MI Iduction
2nd Surgery
Intramyocardial Injection
AAV5:GFP, 1.2x10^12vg/rat
Heart harvest for IHC at
1 wk post-vector delivery
0
2
3
Weeks
B
10X
C
40X
D
40X

Injection at 2wks post-MI

Red - Cardiomyocyte (alpha-Actinin); Green – GFP; Blue – Nuclei

Fig1. Injection of AAV5:GFP at 2wks post-MI achieved high transduction of non-CMs in ischemia zone
(A) Schematic diagram of assessing AAV vector delivery at various times after MI. (**B-D**) IHC analysis of heart tissue injected with AAV:GFP at 2 weeks post-MI.

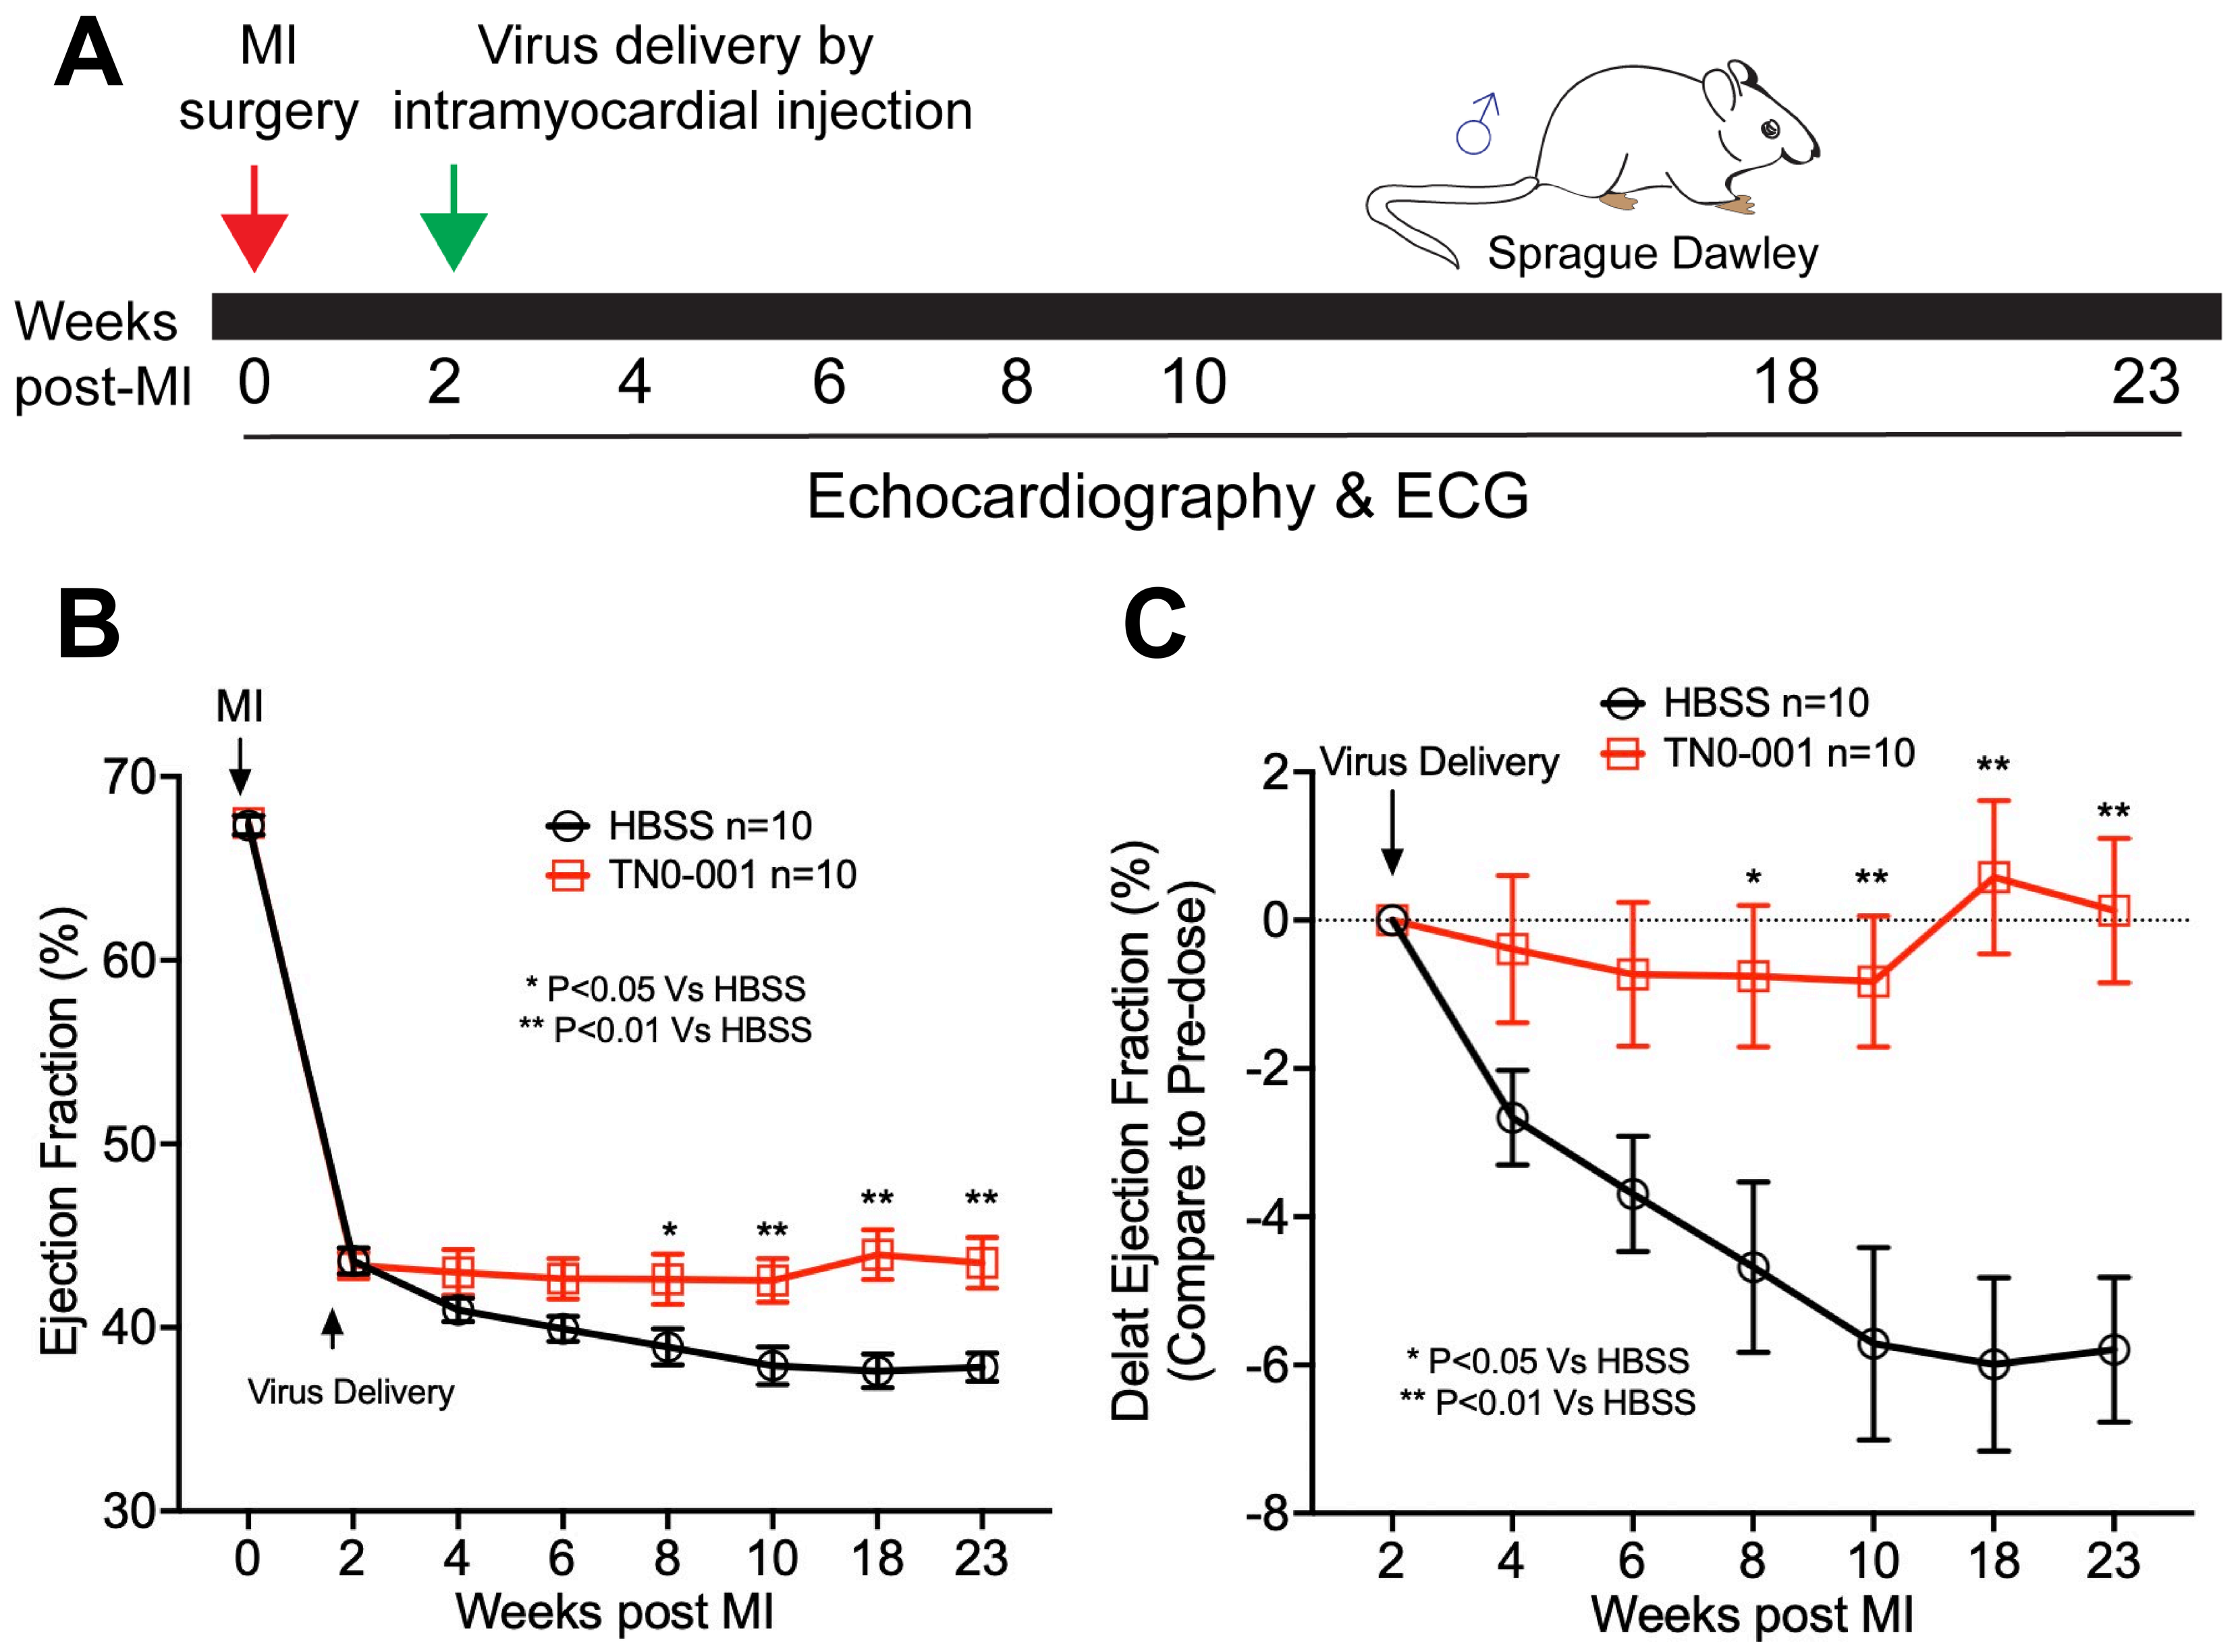
A
MI surgery
Virus delivery by intramyocardial injection
Sprague Dawley
Weeks post-MI
0 2 4 6 8 10 18 23
Echocardiography & ECG
B
MI
Virus Delivery
HBSS n=10
TN0-001 n=10
* P<0.05 Vs HBSS
** P<0.01 Vs HBSS
Ejection Fraction (%)
Weeks post MI
C
Virus Delivery
Delat Ejection Fraction (%)
(Compare to Pre-dose)
Weeks post MI

Results

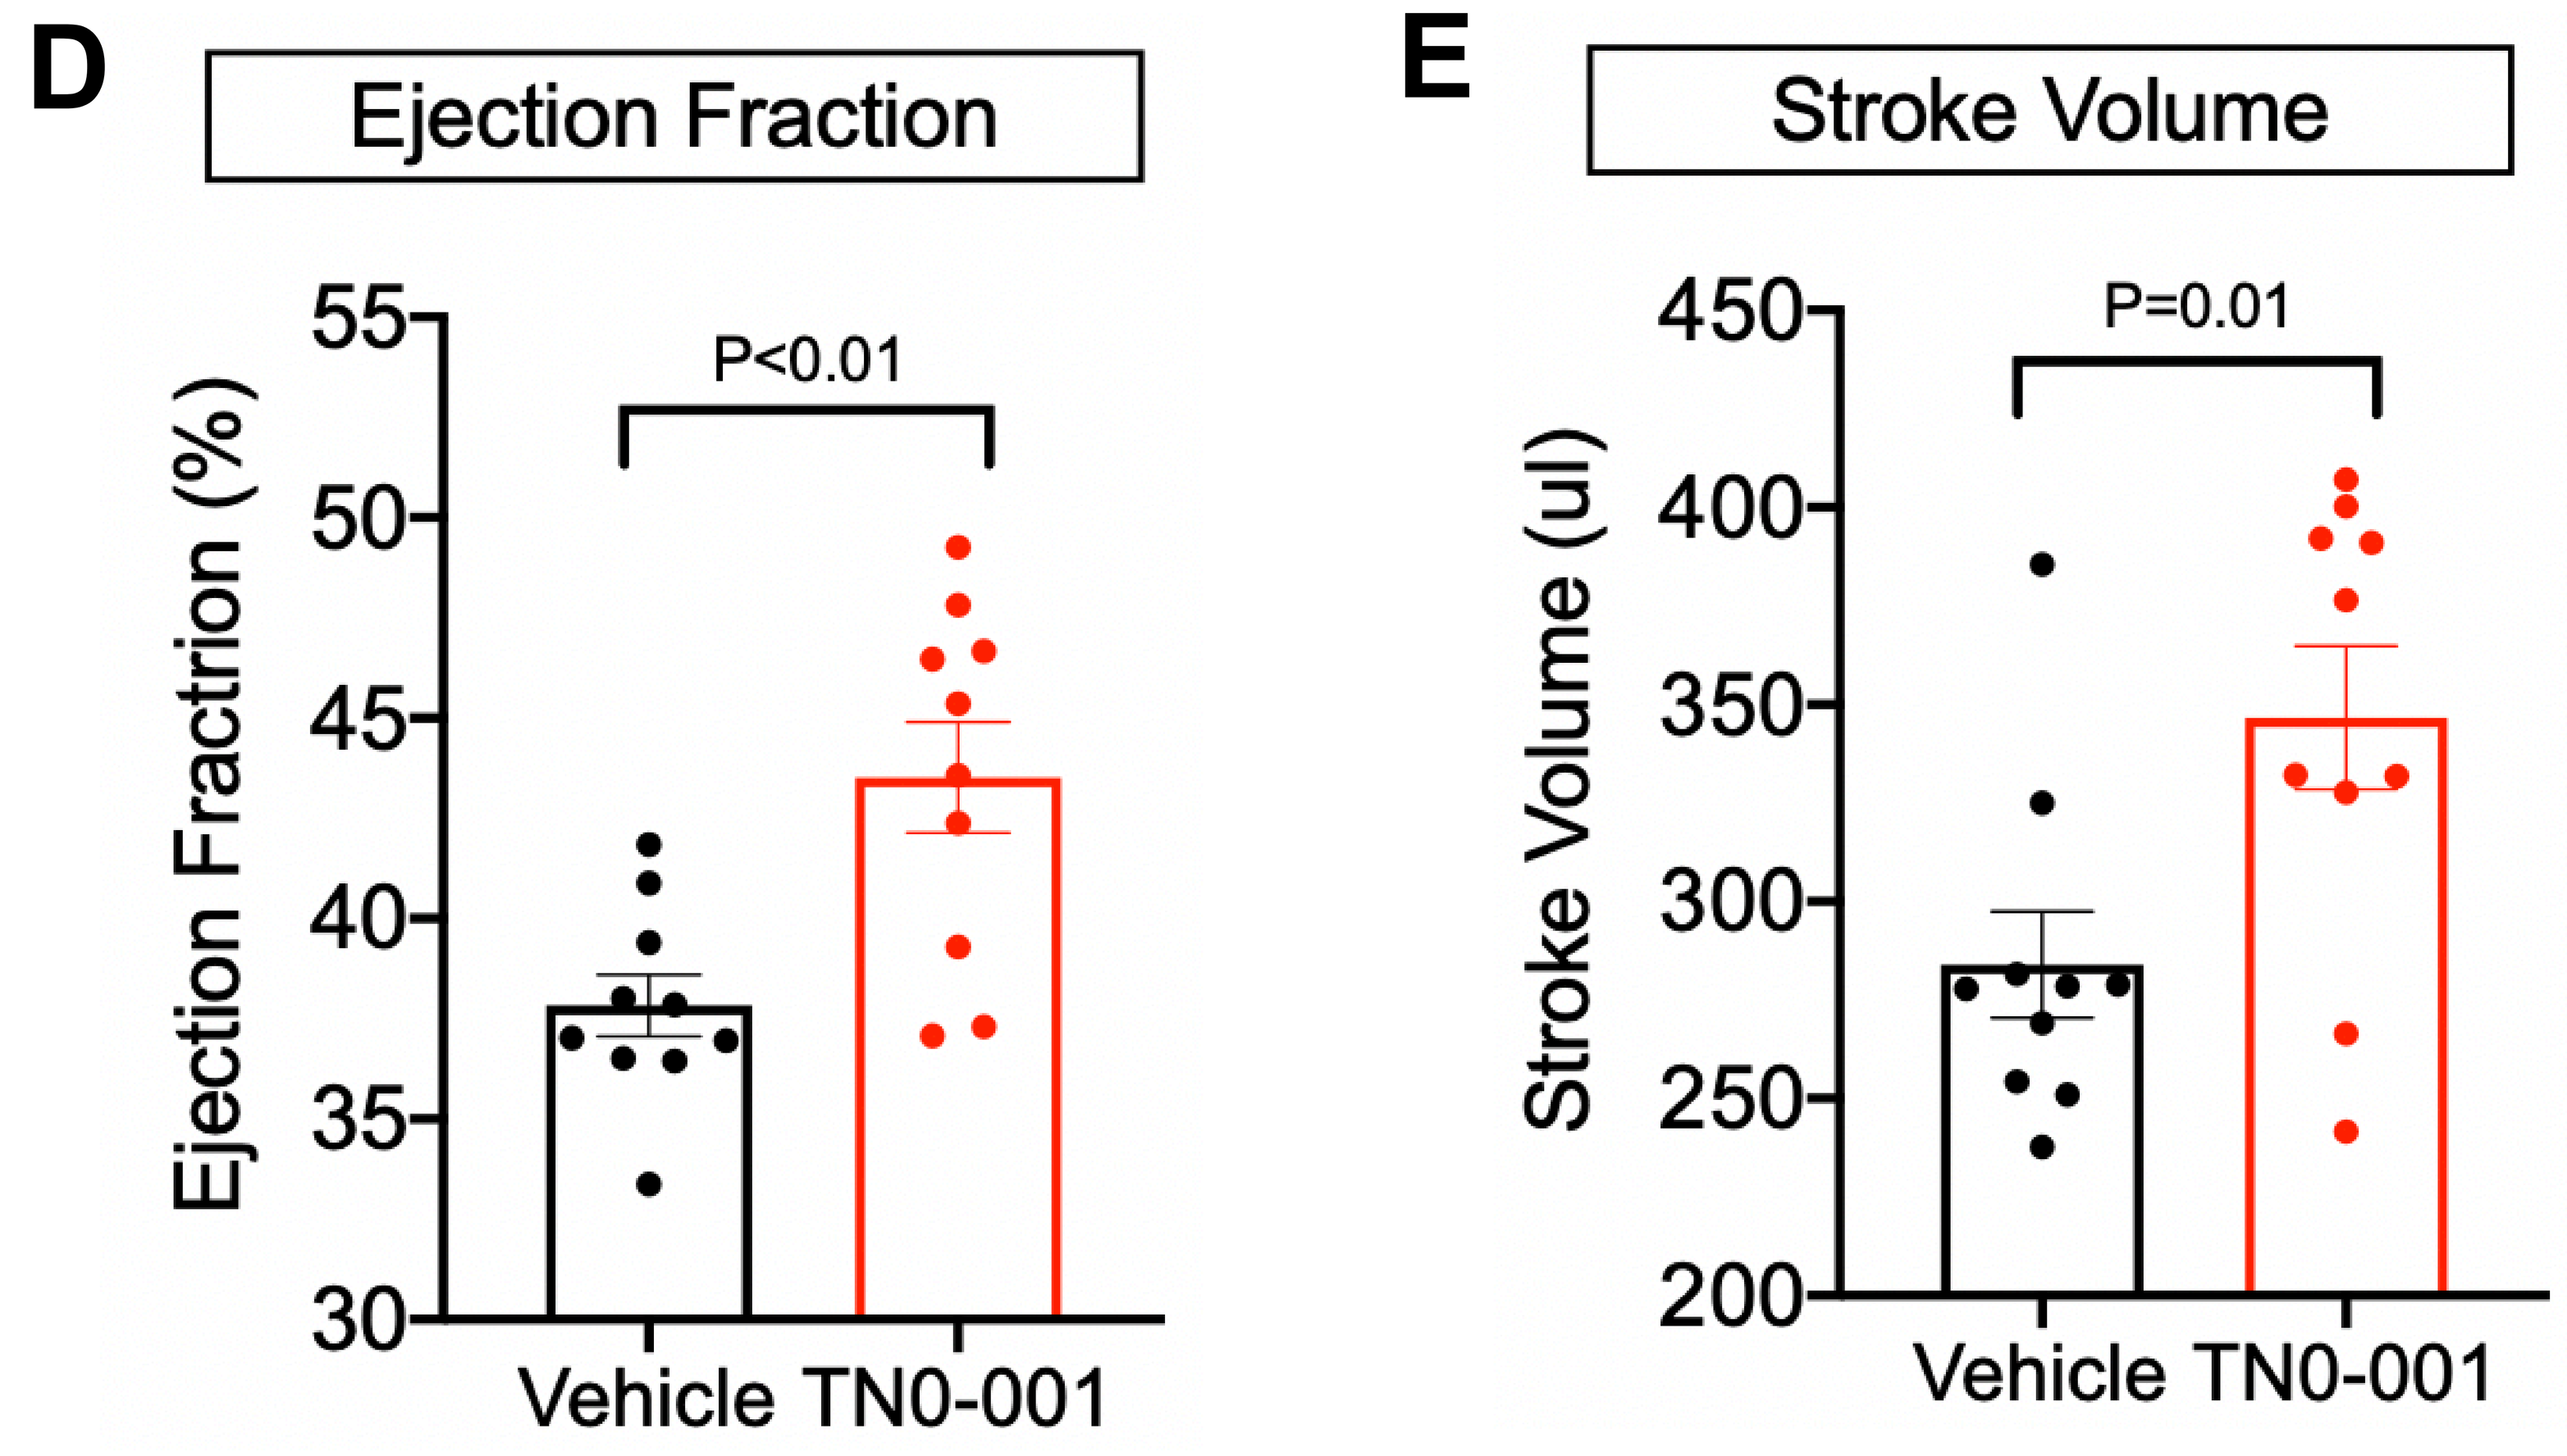
D
Ejection Fraction
P<0.01
Ejection Fractrion (%)
Vehicle TN0-001
E
Stroke Volume
P=0.01
Stroke Volume (ul)
Vehicle TN0-001

Fig2. Efficacy of reprogramming in rat chronic MI model
(**A**) Schematic diagram of rat chronic MI efficacy study. Dose: $5x10^{11}$ vector genome (vg)/rat. (**B-C**) TN0-001 improves cardiac function and halts progression of heart failure in rat chronic MI model. (**D**) 23wks after MI, TN0-001 improved ejection fraction (EF) by ~6% compared to vehicle, similar to pre-Tx baseline. (E) TN0-001 improved overall heart performance by stroke volume (SV) compared to vehicle.
No test article-related arrhythmias were observed by ECG.

Conclusions

We established an MI model in rats and assessed AAV vector delivery at various times after MI. The optimized model allows direct intramyocardial injections at the infarct region at later time points after the acute stage. In this model, the injected AAV vector mediates expression in cardiac fibroblasts in the ischemic zone. AAV-mediated gene delivery of the reprograming cocktail significantly improved LV function when delivered fourteen days after cardiac infarct. There were no test-article related arrhythmias or other safety findings observed in the efficacious dose range. Currently, we are evaluating our lead reprogramming therapeutics in the rat chronic MI setting and aiming to advance this approach to clinical studies.