Histone Deacetylase 6 Inhibition Demonstrates Comparable Efficacy as Empagliflozin in a Mouse Model of Heart Failure with Preserved Ejection Fraction

Sara Ranjbarvaziri[1], Aliya Zeng[1], Iris Wu[1], Amara Greer-Short[1], Farshad Farshidfar[1], Charles E MacKay[1], Reva Shenwai[1], Xiaomei Song[1], Gretchen Argast[1], Jay Vora[1], Matt Holdren[1], Tim Hoey[1*], Jin Yang[1]

1. Tenaya Therapeutics, South San Francisco, CA

** Correspondence: thoey@tenayathera.com*

Background

Heart failure with preserved ejection fraction (HFpEF) is a major health problem associated with high morbidity and mortality, yet there are few effective therapies. Previously, we demonstrated that TN-301 (TYA-631), a highly selective small molecule inhibitor of histone deacetylase 6 (HDAC6), has a multi-modal mechanism of action that works directly on the heart and systematically on metabolism and inflammation. In mouse models of HFpEF, TN-301 improved cardiac structure and diastolic function. Additionally, in diet-induced obesity mouse models, HDAC6 inhibition resulted in improvements in glucose tolerance, insulin resistance, and inflammation. Empagliflozin, a selective inhibitor of sodium-glucose cotransporter protein-2 (SGLT2), was recently approved in the U.S. as a therapy for patients with HFpEF, based upon clinical trials showing it reduces the risk of cardiovascular death and hospitalization in this population. In this study, we aimed to compare the efficacy of HDAC6 and SGLT2 inhibitors in an established mouse model of diastolic dysfunction with preserved ejection fraction.

Methods

To recapitulate systemic and cardiovascular features of HFpEF in humans, we induced diastolic dysfunction in mice using a combination of high fat diet (60%) and N$^{\omega}$-nitrol-arginine methyl ester (L-NAME, 0.5 g/L)[(1)]. After the HFpEF phenotype was established, mice were orally dosed daily for 9 weeks with either 15 mg/kg TYA-018 (TN-301-like HDAC6-specific inhibitor),10 mg/kg empagliflozin (SGLT2 selective inhibitor), or vehicle. In previous studies, these doses were determined to be maximally efficacious doses. Echocardiography, blood glucose measurements, and end point invasive hemodynamic analyses were performed. To better understand TYA-018 and empagliflozin underlying mechanisms of action, transcriptional changes in heart tissue from mice with HFpEF were analyzed using bulk tissue RNA sequencing.

Results

TYA-018 Reverses Pre-existing LV Hypertrophy and Diastolic Dysfunction Similar to SGLT2i

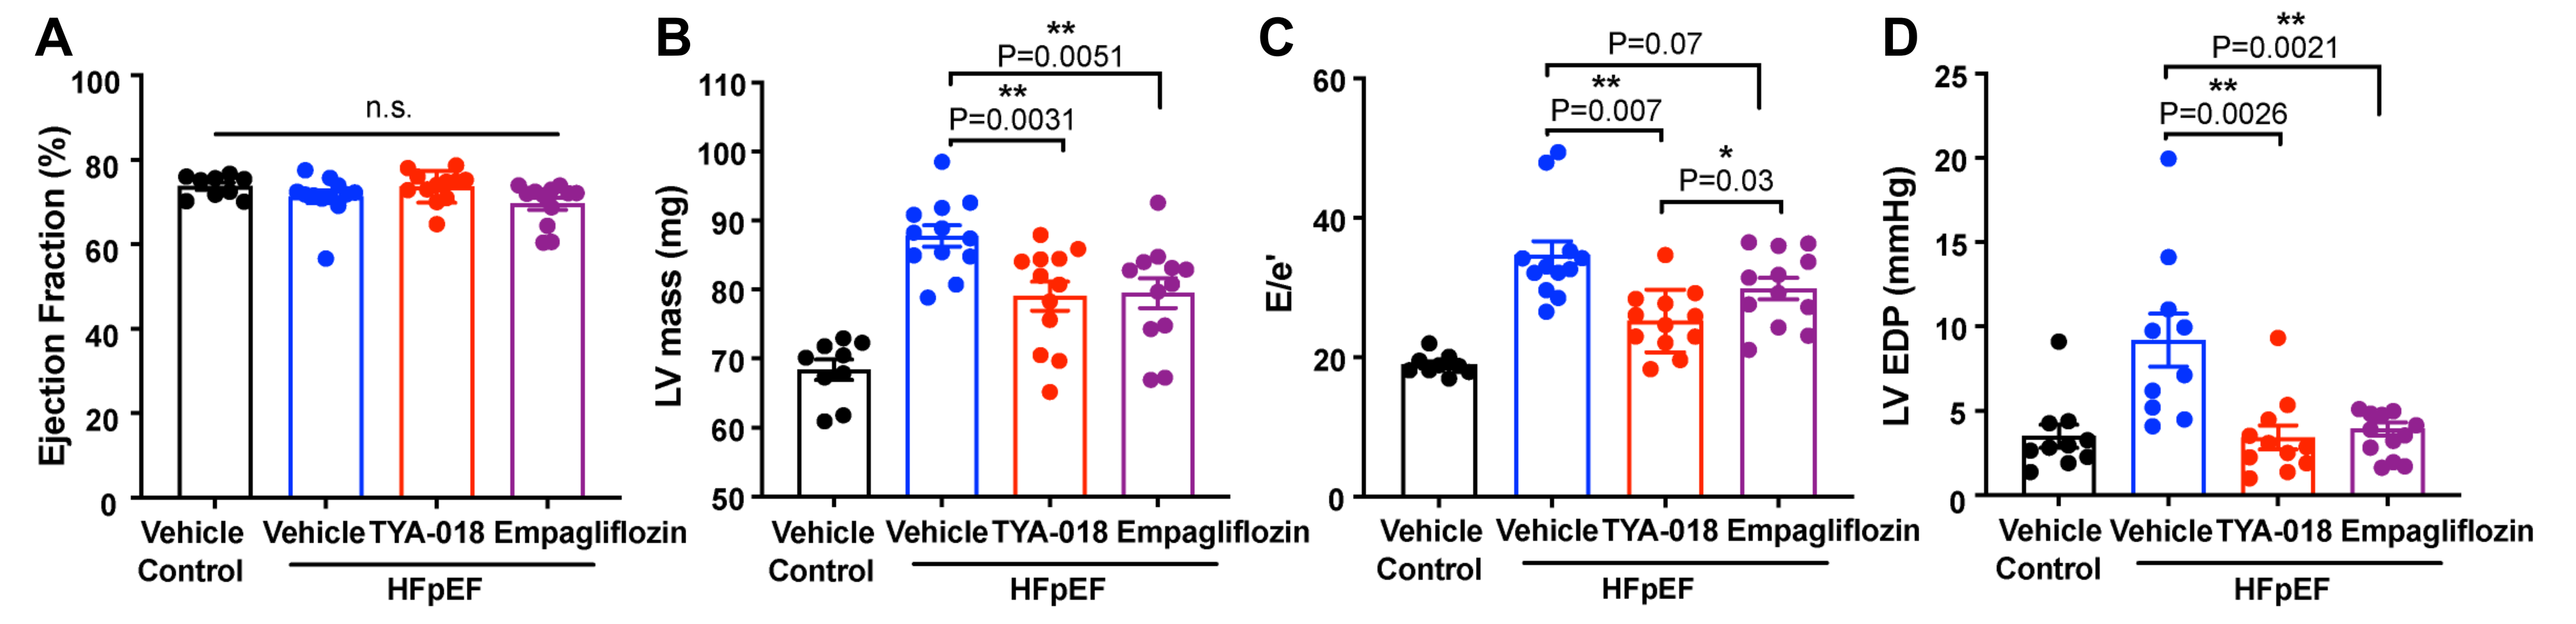
A
Ejection Fraction (%)
n.s.
B
LV mass (mg)
P=0.0051
P=0.0031
C
E/e'
P=0.07
P=0.007
P=0.03
D
LV EDP (mmHg)
P=0.0021
P=0.0026
Vehicle Control
Vehicle TYA-018 Empagliflozin
HFpEF

(A) Ejection fraction was preserved in treated and untreated HFpEF mice. (B-D) Daily oral dosing of TYA-018 for 9 weeks, normalized cardiac structure and function as measured by left ventricular (LV) mass (B), E/e' ratio (C), and end diastolic pressure (D) in HFpEF mice. These effects were comparable to empagliflozin.

All data are presented as the mean ± SEM. N=8-12 mice per group. n.s. not significant, *P < 0.05, and **P < 0.01 by unpaired t-test.

TYA-018 Significantly Reduced BNP *G*ene Expression Greater than Empagliflozin

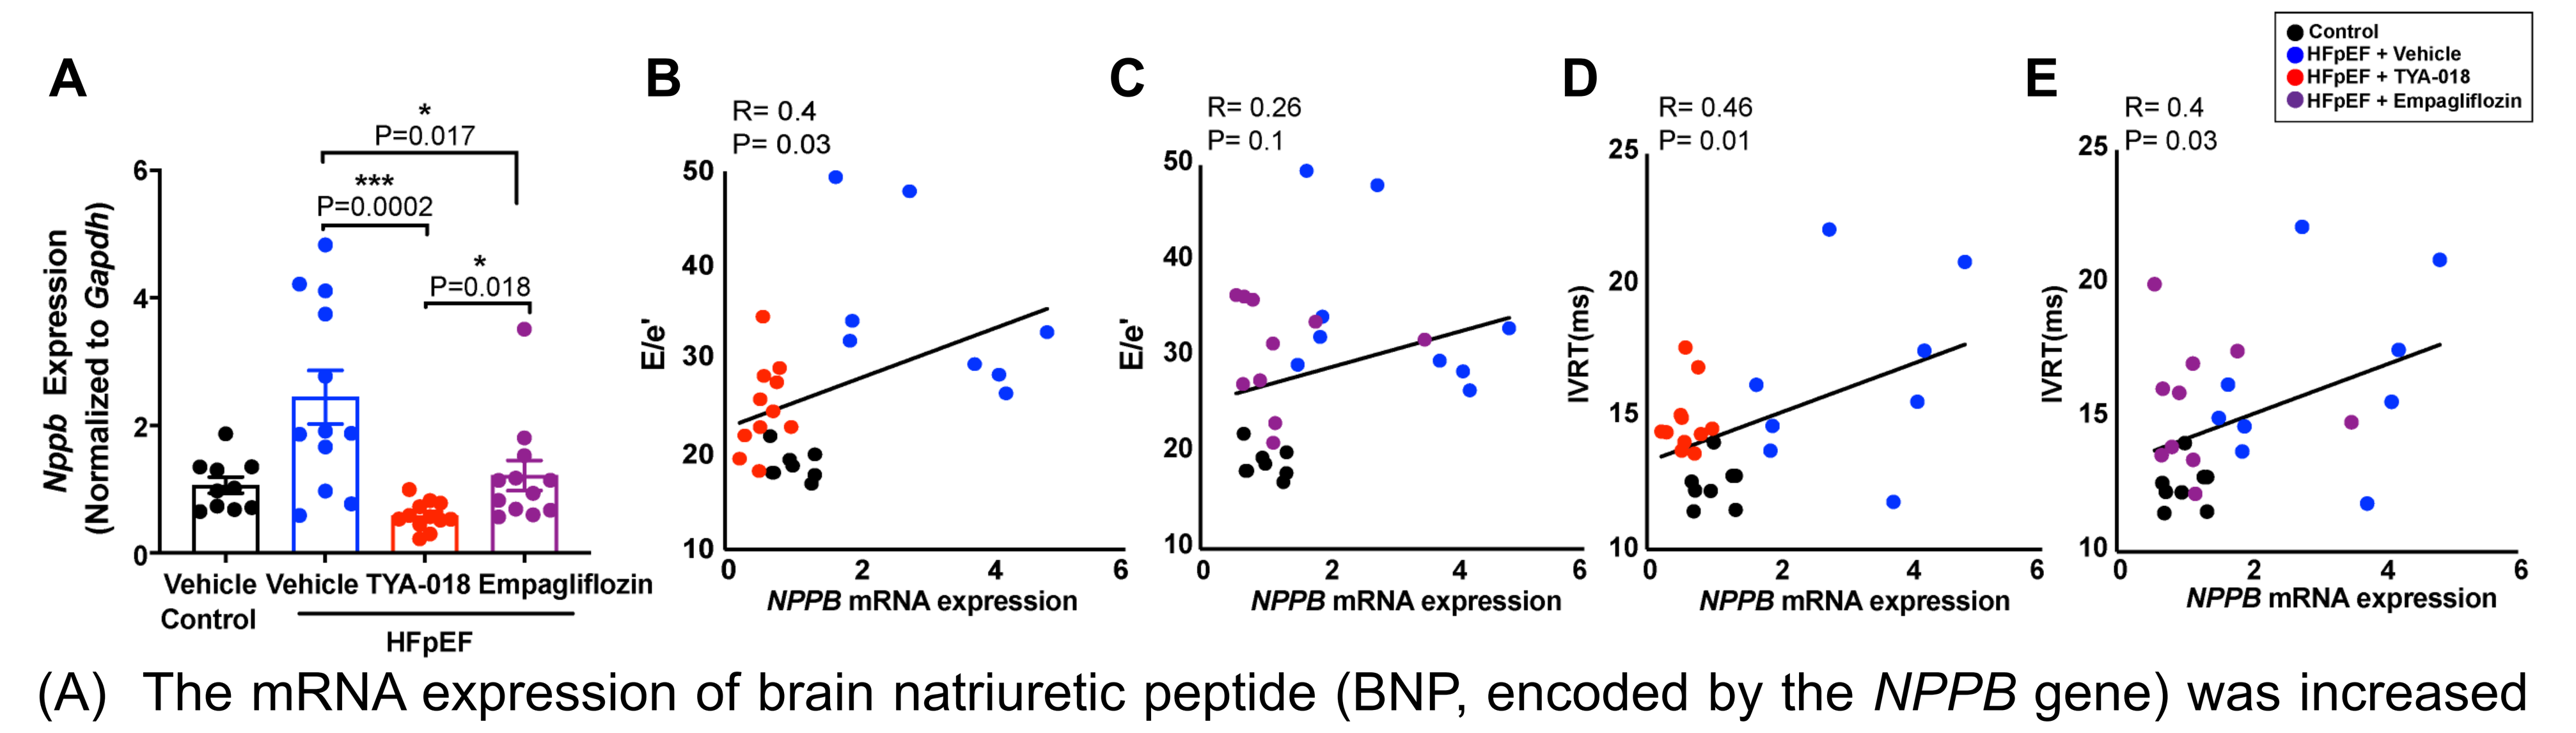
A
Nppb Expression (Normalized to Gapdh)
P=0.017
P=0.0002
P=0.018
Vehicle Control
Vehicle TYA-018 Empagliflozin
HFpEF
B
R= 0.4
P= 0.03
E/e'
C
R= 0.26
P= 0.1
E/e'
D
R= 0.46
P= 0.01
IVRT(ms)
E
R= 0.4
P= 0.03
IVRT(ms)
NPPB mRNA expression
Control
HFpEF + Vehicle
HFpEF + TYA-018
HFpEF + Empagliflozin

(A) The mRNA expression of brain natriuretic peptide (BNP, encoded by the *NPPB* gene) was increased in mouse HFpEF hearts and was significantly decreased by both TYA-018 and empagliflozin treatment. Compared to empagliflozin, TYA-018 reduced *NPPB* to a greater extent. (B-E) Reduced expression of *NPPB* in response to both treatments was correlated with improved diastolic function in HFpEF mice as shown by decreased E/e′ ratio and isovolumic relaxation time (IVRT).

Data in A panel is presented as the mean ± SEM. *P < 0.05 and ***P < 0.001 by unpaired t-test. R values are Pearson correlation coefficients. N=8-11 mice per group.

TYA-018 Improved Fasting Glucose and Glucose Tolerance to Levels Similar to Empagliflozin

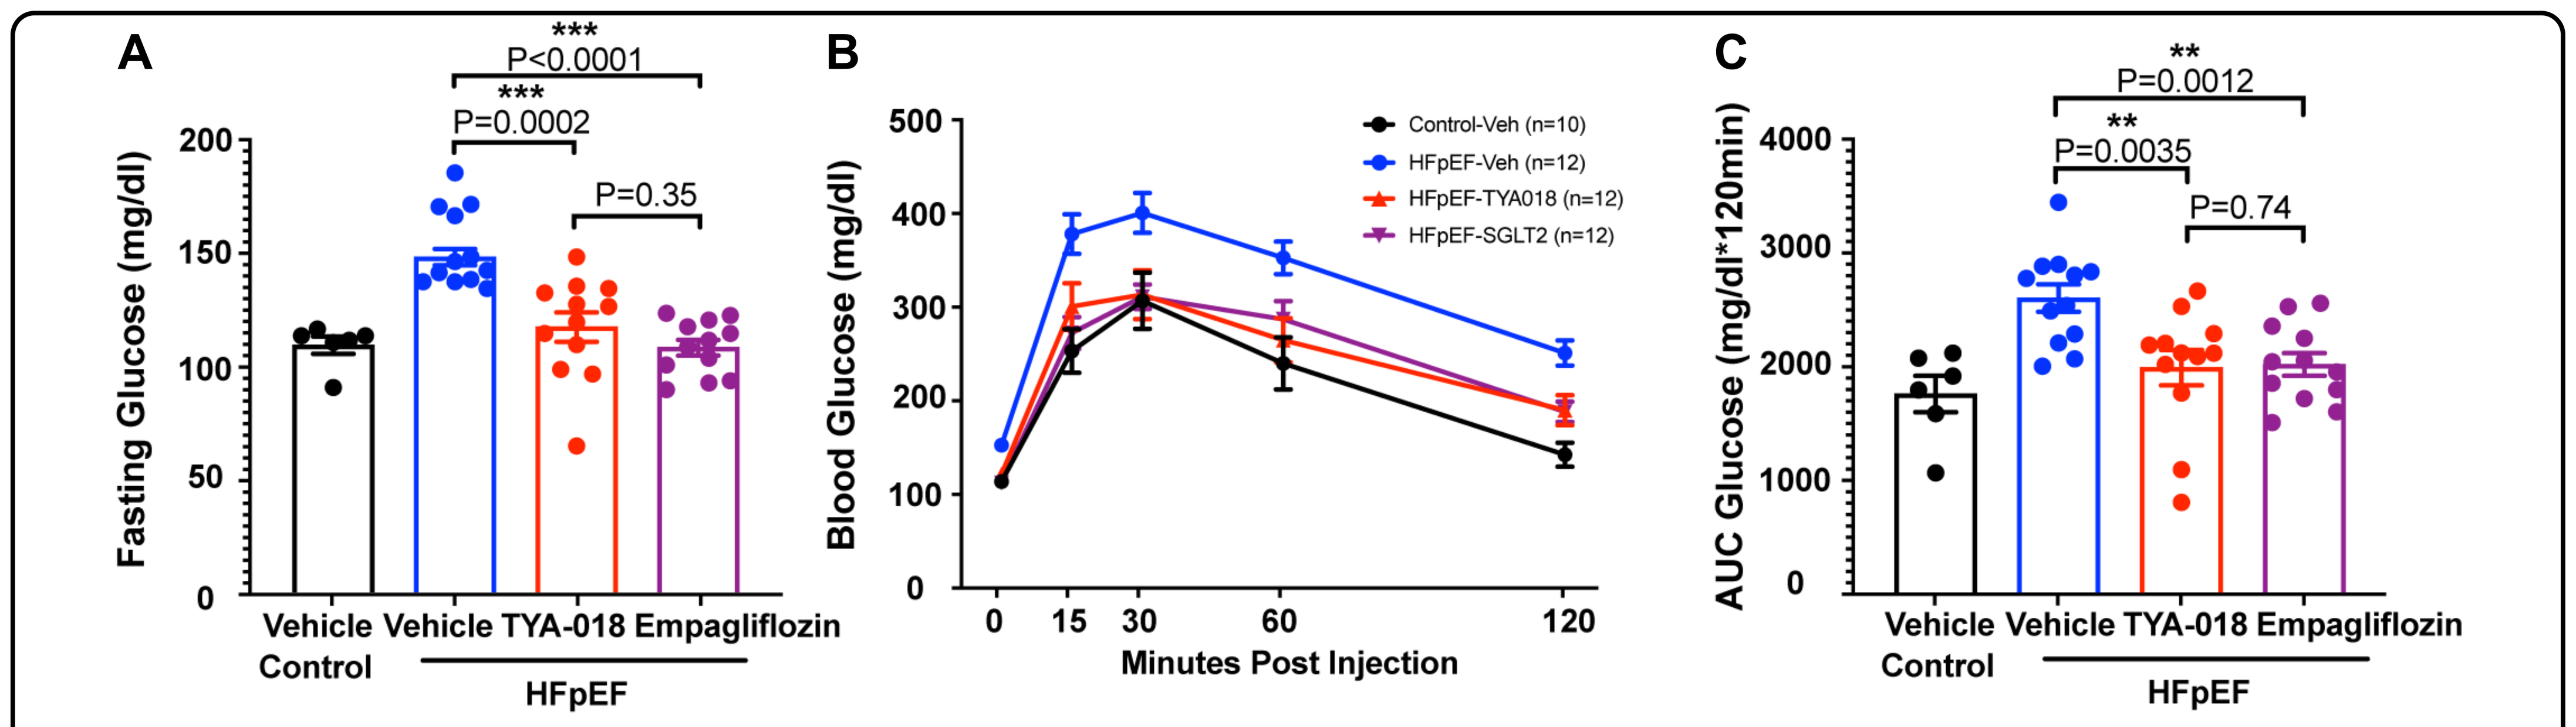
A
Fasting Glucose (mg/dl)
P<0.0001
P=0.0002
P=0.35
B
Blood Glucose (mg/dl)
Control-Veh (n=10)
HFpEF-Veh (n=12)
HFpEF-TYA018 (n=12)
HFpEF-SGLT2 (n=12)
Minutes Post Injection
C
AUC Glucose (mg/dl*120min)
P=0.0012
P=0.0035
P=0.74
Vehicle Control
Vehicle TYA-018 Empagliflozin
HFpEF

(A) Single oral dose of TYA-018 significantly reduced fasting glucose and improved glucose tolerance (B and C) in HFpEF mice, to levels similar to empagliflozin.

All data are presented as the mean ± SEM. N=6-12 mice per group. **P < 0.01 and ***P < 0.001 by unpaired t-test. Historical data was used as vehicle control group.

TYA-018 Corrects Dysregulated Fibrosis and Oxidative Phosphorylation Gene Expression Better than Empagliflozin

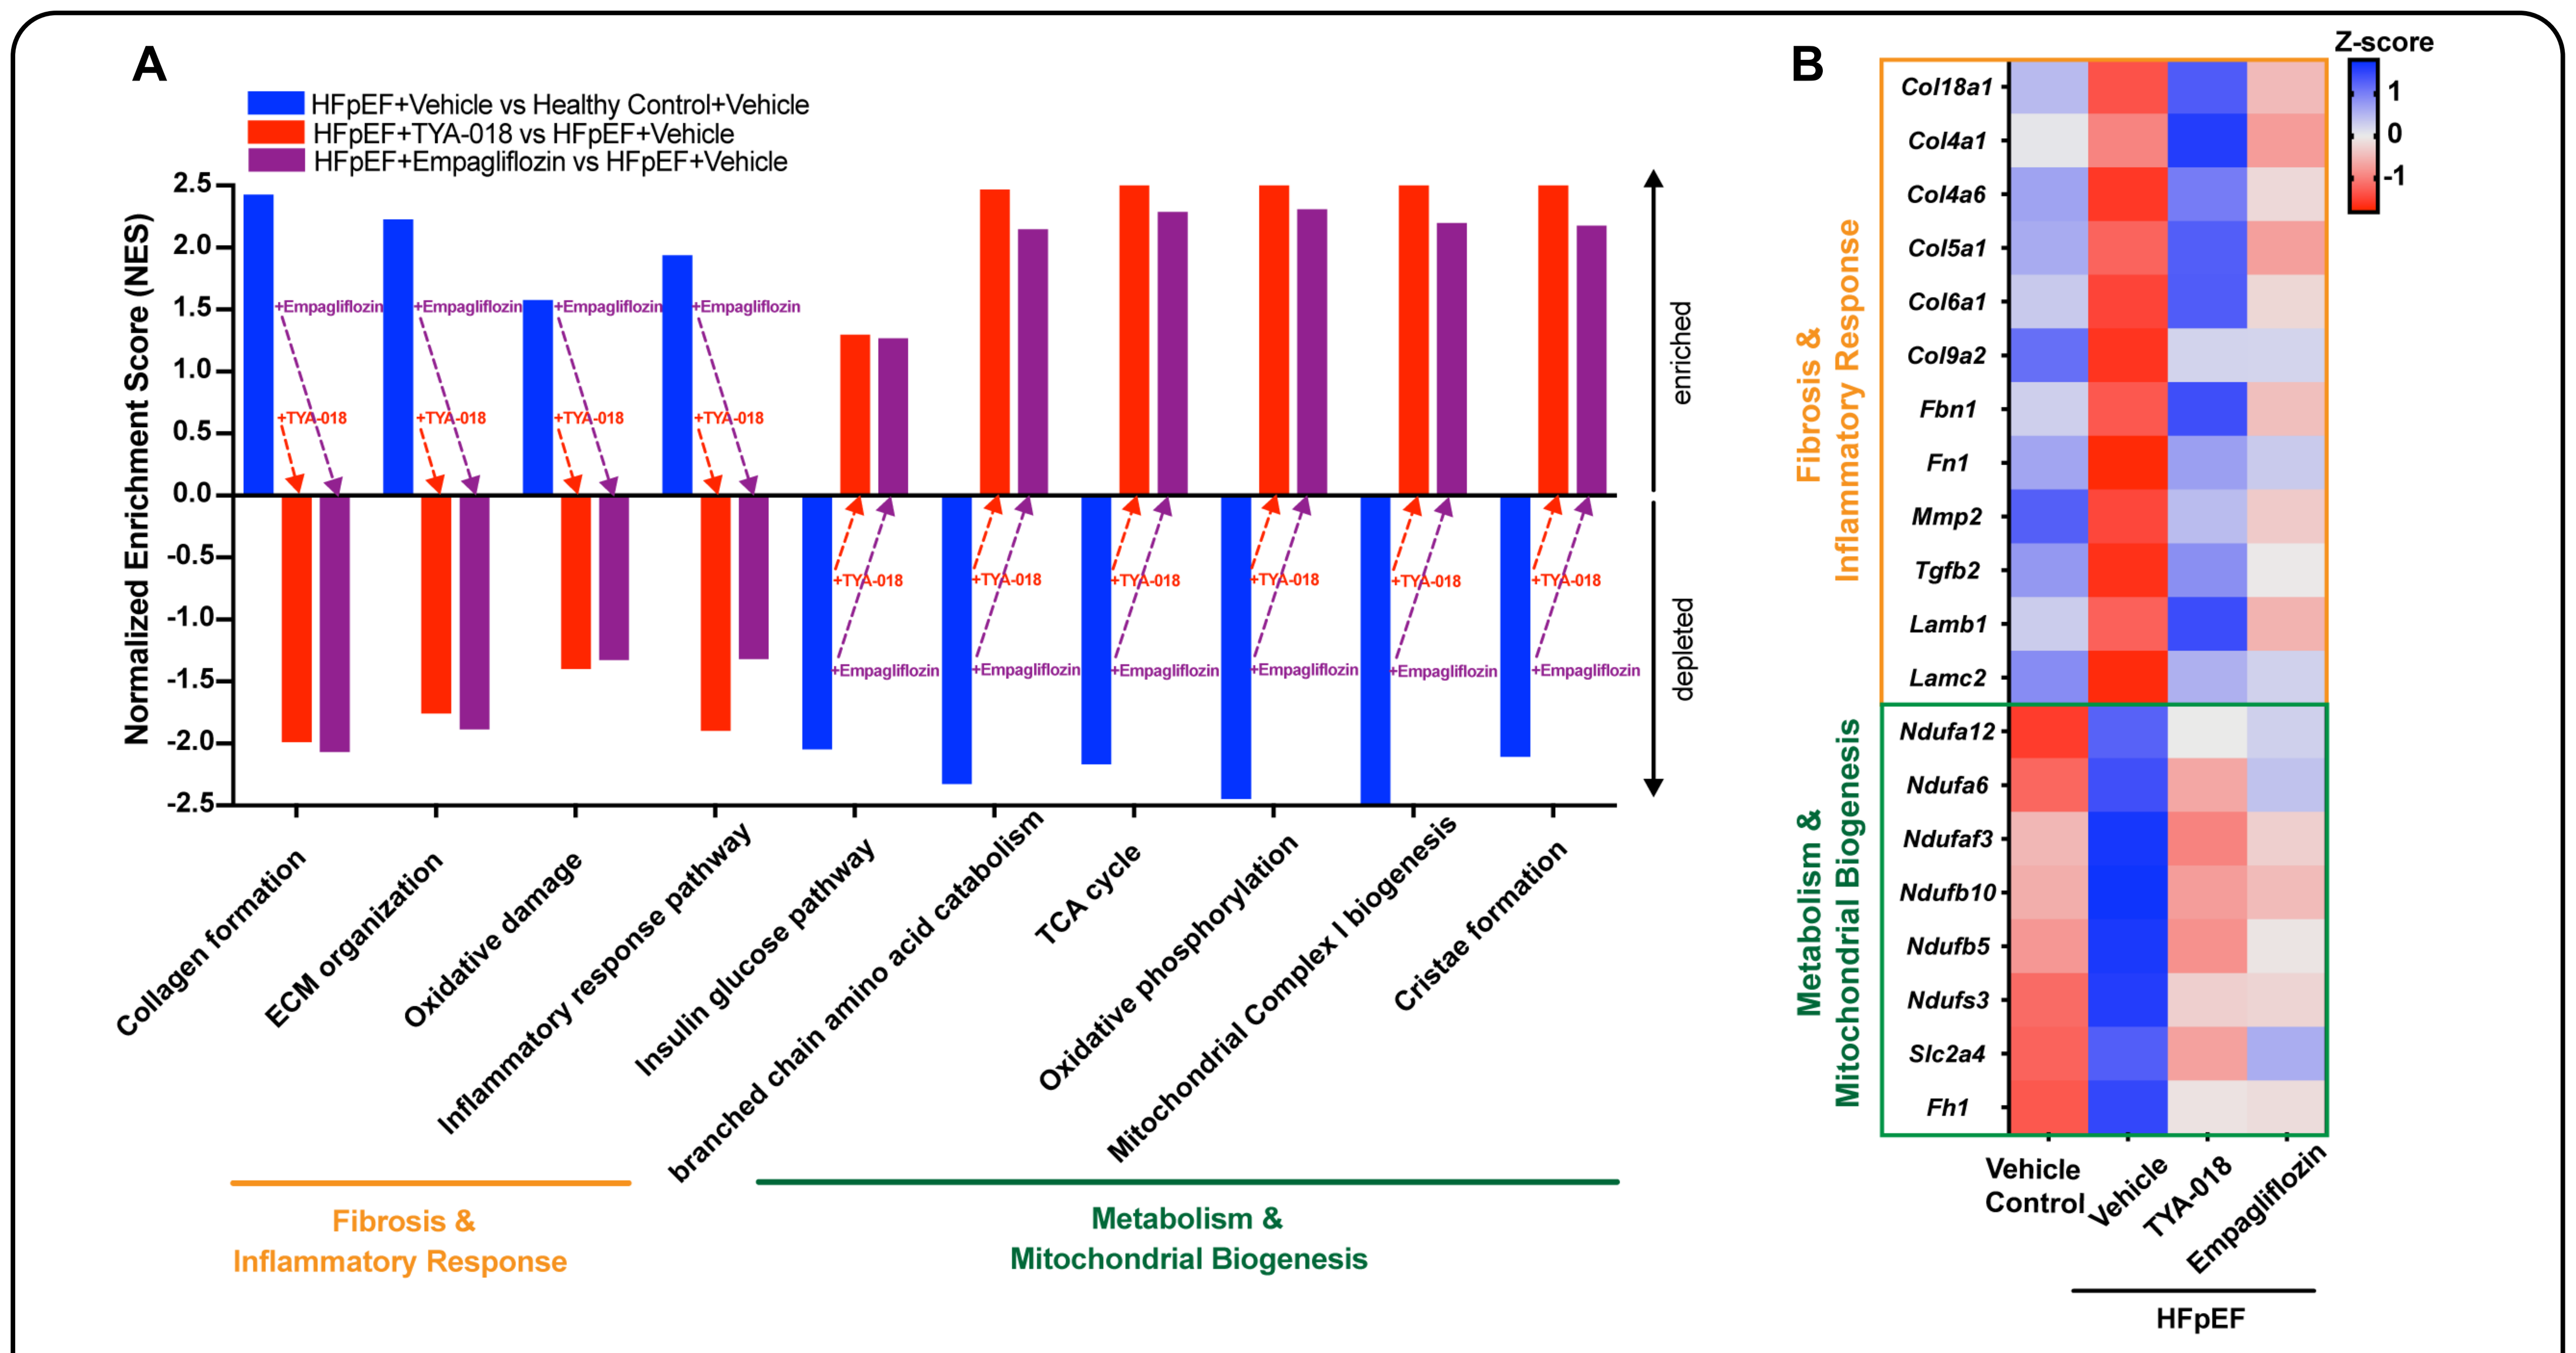
A
HFpEF+Vehicle vs Healthy Control+Vehicle
HFpEF+TYA-018 vs HFpEF+Vehicle
HFpEF+Empagliflozin vs HFpEF+Vehicle
Normalized Enrichment Score (NES)
enriched
depleted
+Empagliflozin
+TYA-018
Collagen formation
ECM organization
Oxidative damage
Inflammatory response pathway
Insulin glucose pathway
branched chain amino acid catabolism
TCA cycle
Oxidative phosphorylation
Mitochondrial Complex I biogenesis
Cristae formation
Fibrosis & Inflammatory Response
Metabolism & Mitochondrial Biogenesis
B
Z-score
Col18a1
Col4a1
Col4a6
Col5a1
Col6a1
Col9a2
Fbn1
Fn1
Mmp2
Tgfb2
Lamb1
Lamc2
Ndufa12
Ndufa6
Ndufaf3
Ndufb10
Ndufb5
Ndufs3
Slc2a4
Fh1
Vehicle Control
Vehicle
TYA-018
Empagliflozin
HFpEF

(A) RNA sequencing analysis shows enrichment of gene sets associated with inflammation, fibrosis, and oxidative stress in vehicle-treated HFpEF animals relative to control (shown in blue bars) which were all reversed with TYA-018 (red bars) or empagliflozin (bars in purple) treatments. Gene sets associated with mitochondrial function including oxidative phosphorylation, mitochondrial biogenesis, and different metabolic pathways were significantly depleted in HFpEF mice treated with vehicle relative to healthy control animals. These mitochondrial and metabolic gene sets were significantly enriched in both treatment groups. (B) Heatmap shows TYA-018 reduced the expression of inflammatory and fibrotic genes in vehicle treated HFpEF hearts while mitochondrial energy production genes were upregulated in response to TYA-018 treatment, superior to empagliflozin.

N=8-11 mice per group.

Conclusion

Our results show that TYA-018-mediated HDAC6 inhibition reverses preexisting hypertrophy, diastolic dysfunction, and glucose tolerance in a HFpEF mouse model, with similar efficacy to that of empagliflozin when each was administered at maximally efficacious doses. TYA-018 treatment significantly reduced the expression of *NPPB*, a well-established cardiac stress biomarker, to near healthy levels, which was correlated with improved diastolic function. Notably, TYA-018 showed greater effects on gene expression of multiple pathways associated with fibrosis and mitochondrial dysfunction, both of which contribute to HFpEF pathogenesis[(2)]. The comparable efficacy seen in this HFpEF model with TYA-018 and empagliflozin provides early but encouraging evidence of the potential translatability of these findings to clinical development. We are developing TN-301 for treating HFpEF in humans and will explore its utility in individuals both with and without diabetes and metabolic syndrome.

Disclosures: All authors are shareholders and/or employees of Tenaya Therapeutics, Inc.

References

(1) Schiattarella et al., Nature. 2019

(2) Lam et al., Eur Heart J. 2018