Phenotypic screening identifies HDAC6 inhibitors as cardioprotective agents in BAG3 cardiac-knockout mouse model of dilated cardiomyopathy

Jin Yang[1], Francis Grafton[1], Anastasiia Budan[1], Emma Xu[1], Marie Cho[1], Farshad Farshidfar[1], Jaclyn Ho[1], Snahel Patel[1], Mahnaz Maddah[2], Kevin E. Loewke[2], Julio Medina[3], David Sperandio[3], Tim Hoey[1], Mohammad A. Mandegar[1]

1. Tenaya Therapeutics, 171 Oyster Point Blvd, Suite 500, South San Francisco, CA, USA
2. Dana Solutions LLC, 3505 El Camino Real, Palo Alto, CA, USA
3. R2M Pharma, South San Francisco, CA, USA

Use of iPSC-CM as Drug Discovery Platform For Dilated Cardiomyopathy

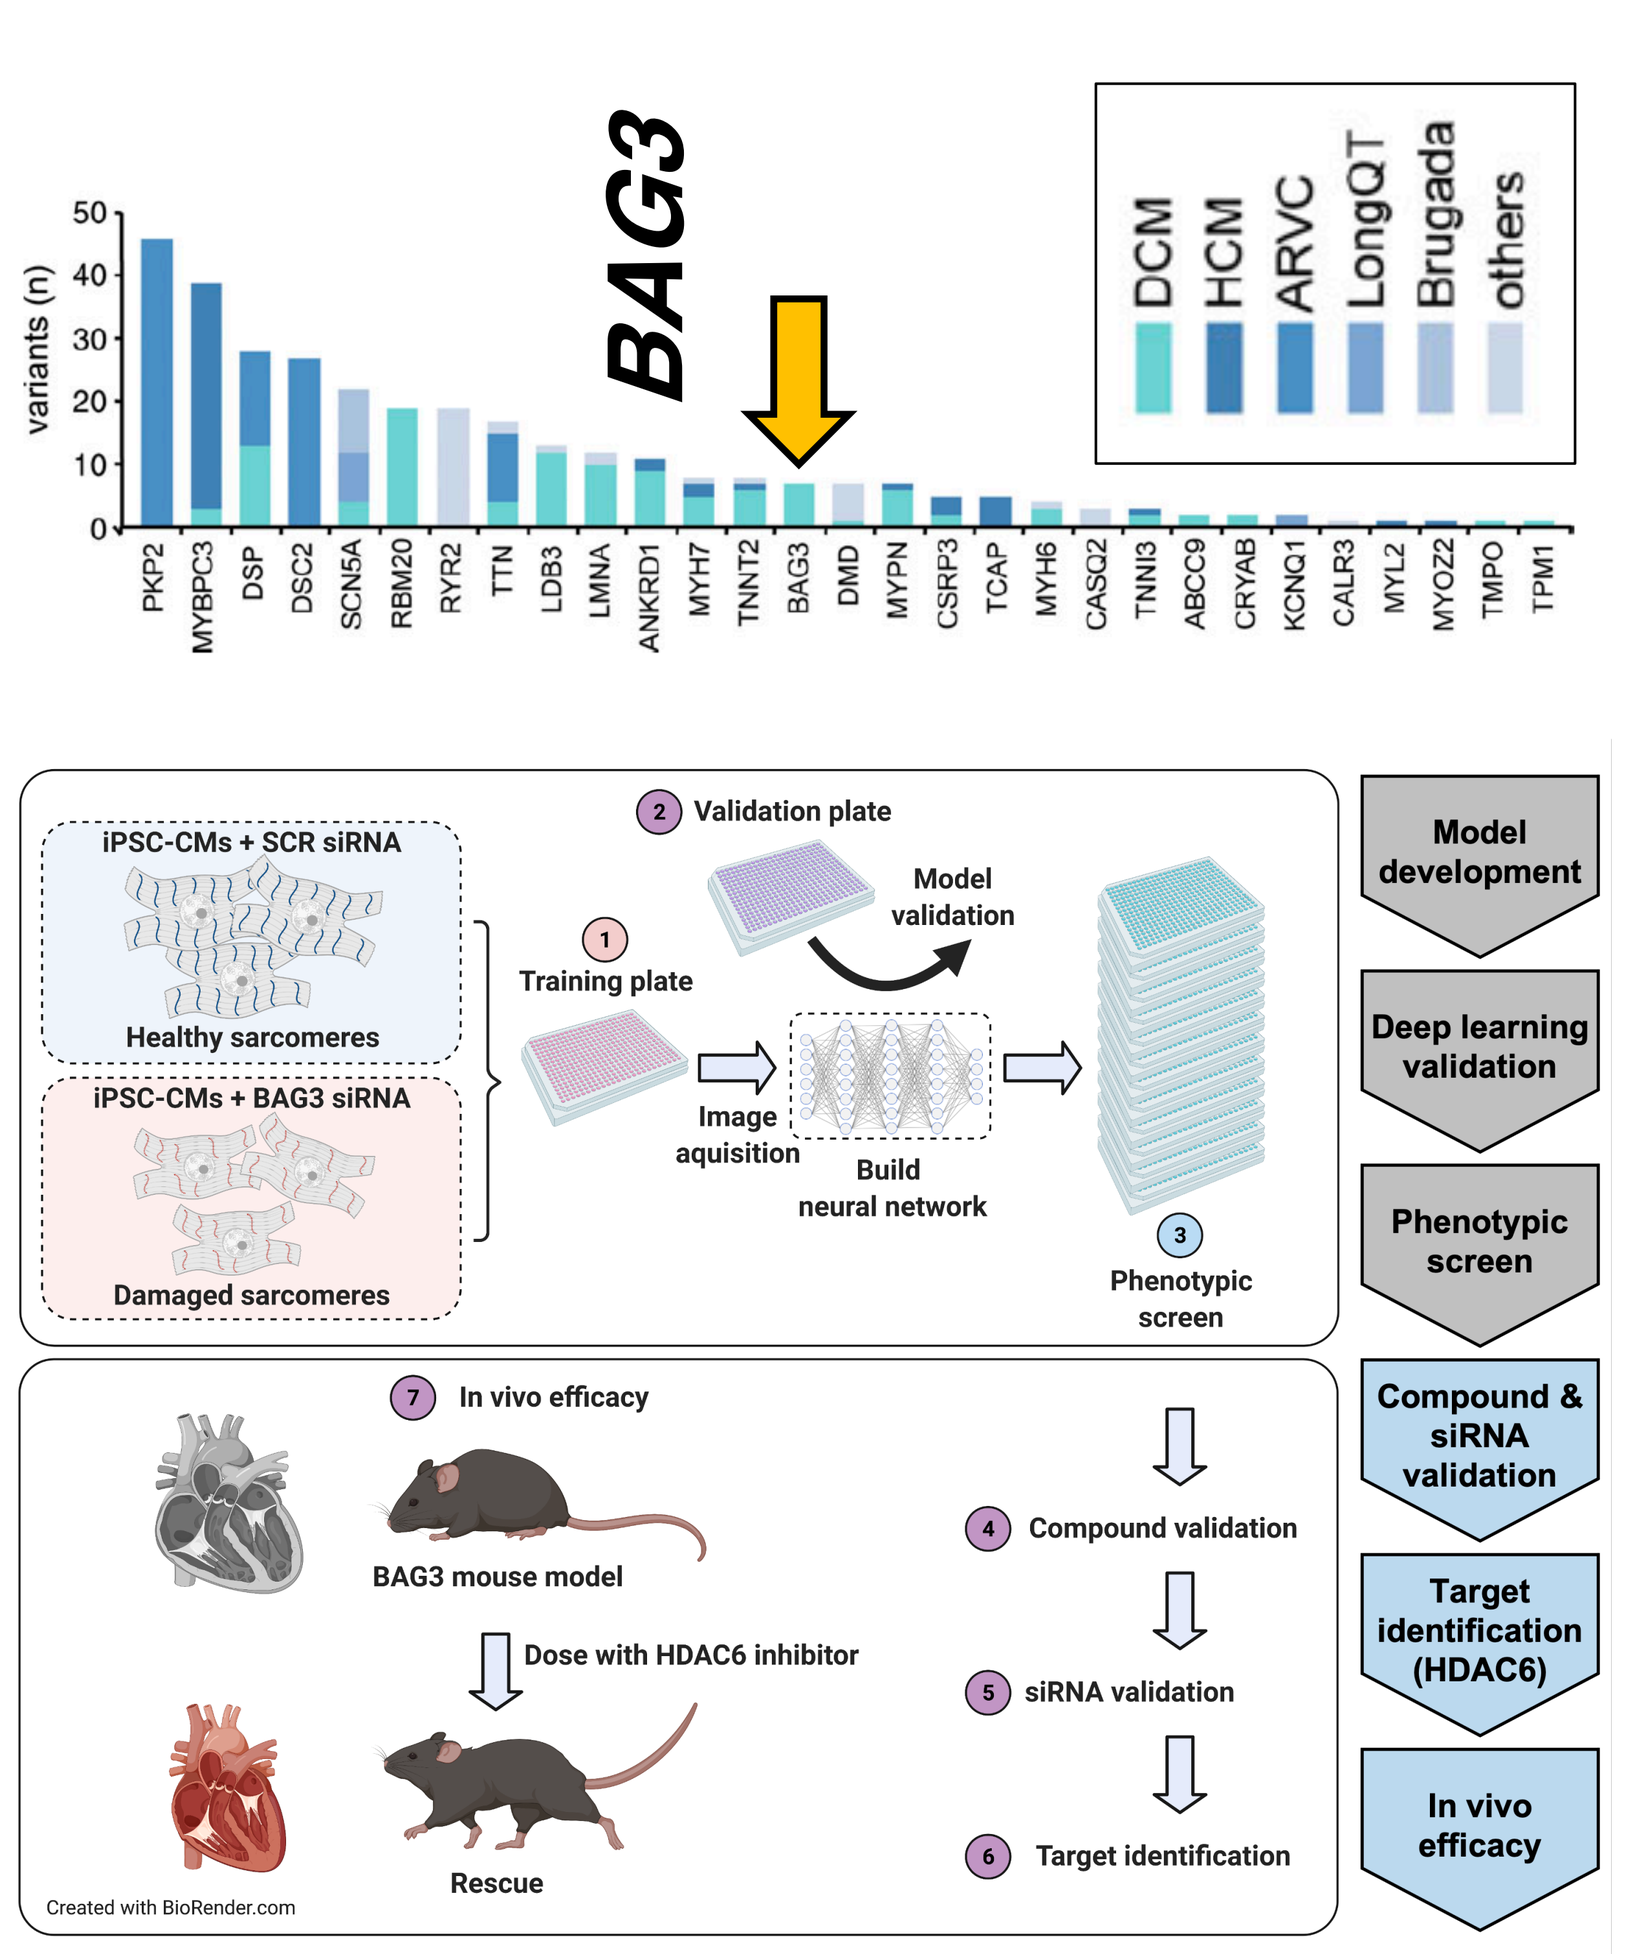

BAG3
variants (n)
DCM
HCM
ARVC
LongQT
Brugada
others
iPSC-CMs + SCR siRNA
Healthy sarcomeres
iPSC-CMs + BAG3 siRNA
Damaged sarcomeres
Validation plate
Training plate
Model validation
Image aquisition
Build neural network
Phenotypic screen
In vivo efficacy
BAG3 mouse model
Dose with HDAC6 inhibitor
Rescue
Compound validation
siRNA validation
Target identification
Model development
Deep learning validation
Phenotypic screen
Compound & siRNA validation
Target identification (HDAC6)
In vivo efficacy
Created with BioRender.com

Background: Familial dilated cardiomyopathy (DCM) is characterized by reduced cardiac output, as well as thinning and enlargement of left ventricular chambers. These characteristics eventually lead to heart failure. Current standards of care do not target the underlying molecular mechanisms associated with genetic forms of heart failure, driving a need to develop novel therapeutics that target genetic DCM.
Purpose: To identify candidate therapeutics, we developed an in vitro DCM model using induced pluripotent stem cell–derived cardiomyocytes (iPSC-CMs) deficient in BAG3.
Methods and Results: Using BAG3-deficient iPSC-CMs, we identified cardioprotective drugs with a phenotypic screen and deep learning. Using a library of 5500 bioactive compounds and siRNA validation, we identified that inhibiting HDAC6 was cardioprotective at the sarcomere level. We translated this finding to a BAG3 cardiac-knockout (BAG3^{cKO}) mouse model of DCM, showing that inhibiting HDAC6 with two isoform-selective inhibitors (tubastatin A and a novel inhibitor TYA-018) protected heart function. HDAC6 inhibitors also protected the microtubule network from mechanical damage, increased autophagic flux, decreased apoptosis, and reduced inflammation in the heart.
Conclusion: HDAC6 inhibitors improved left ventricular ejection fraction and extended lifespan in a BAG3^{cKO} mouse model of DCM. Our results demonstrate the power of combining iPSC-CMs with phenotypic screening and deep learning to identify therapeutic targets for DCM.

BAG3 Deficiency Leads to Sarcomere Damage

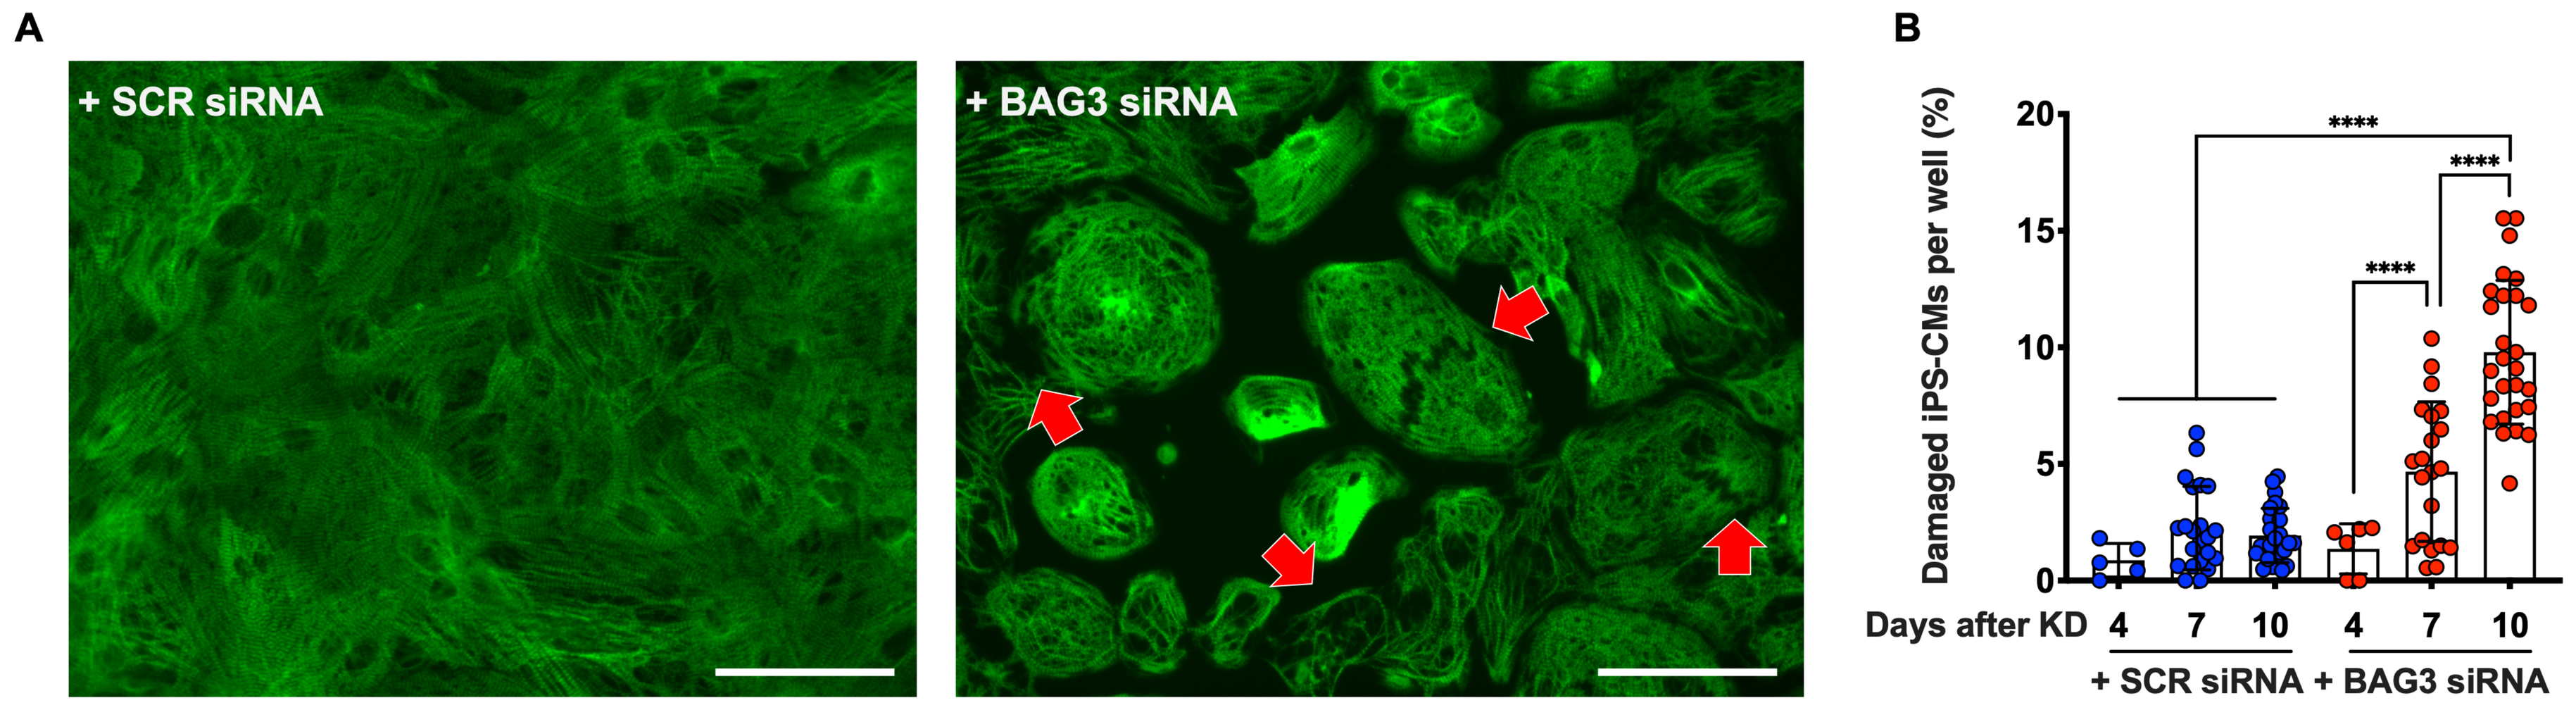

A
+ SCR siRNA
+ BAG3 siRNA
B
Damaged iPS-CMs per well (%)
Days after KD 4 7 10 4 7 10
+ SCR siRNA + BAG3 siRNA

(A) Representative immunostaining of iPSC-CMs treated with SCR or BAG3 siRNA, and then stained with an antibody against MYBPC3. Arrows indicate breaks and reduced sarcomere content in BAG3 knockdown cells. Scale bars = 50 μm. (B) Quantification of sarcomere damage in iPSC-CMs treated with SCR or BAG3 siRNA. The number of damaged iPSC-CMs increased as a function of time in BAG3 knockdown (KD) cells. Error bars = SD. ****P < 0.0001.

High throughput Screening Identifies HDAC and Microtubule Inhibitors a Cardioprotective Agents

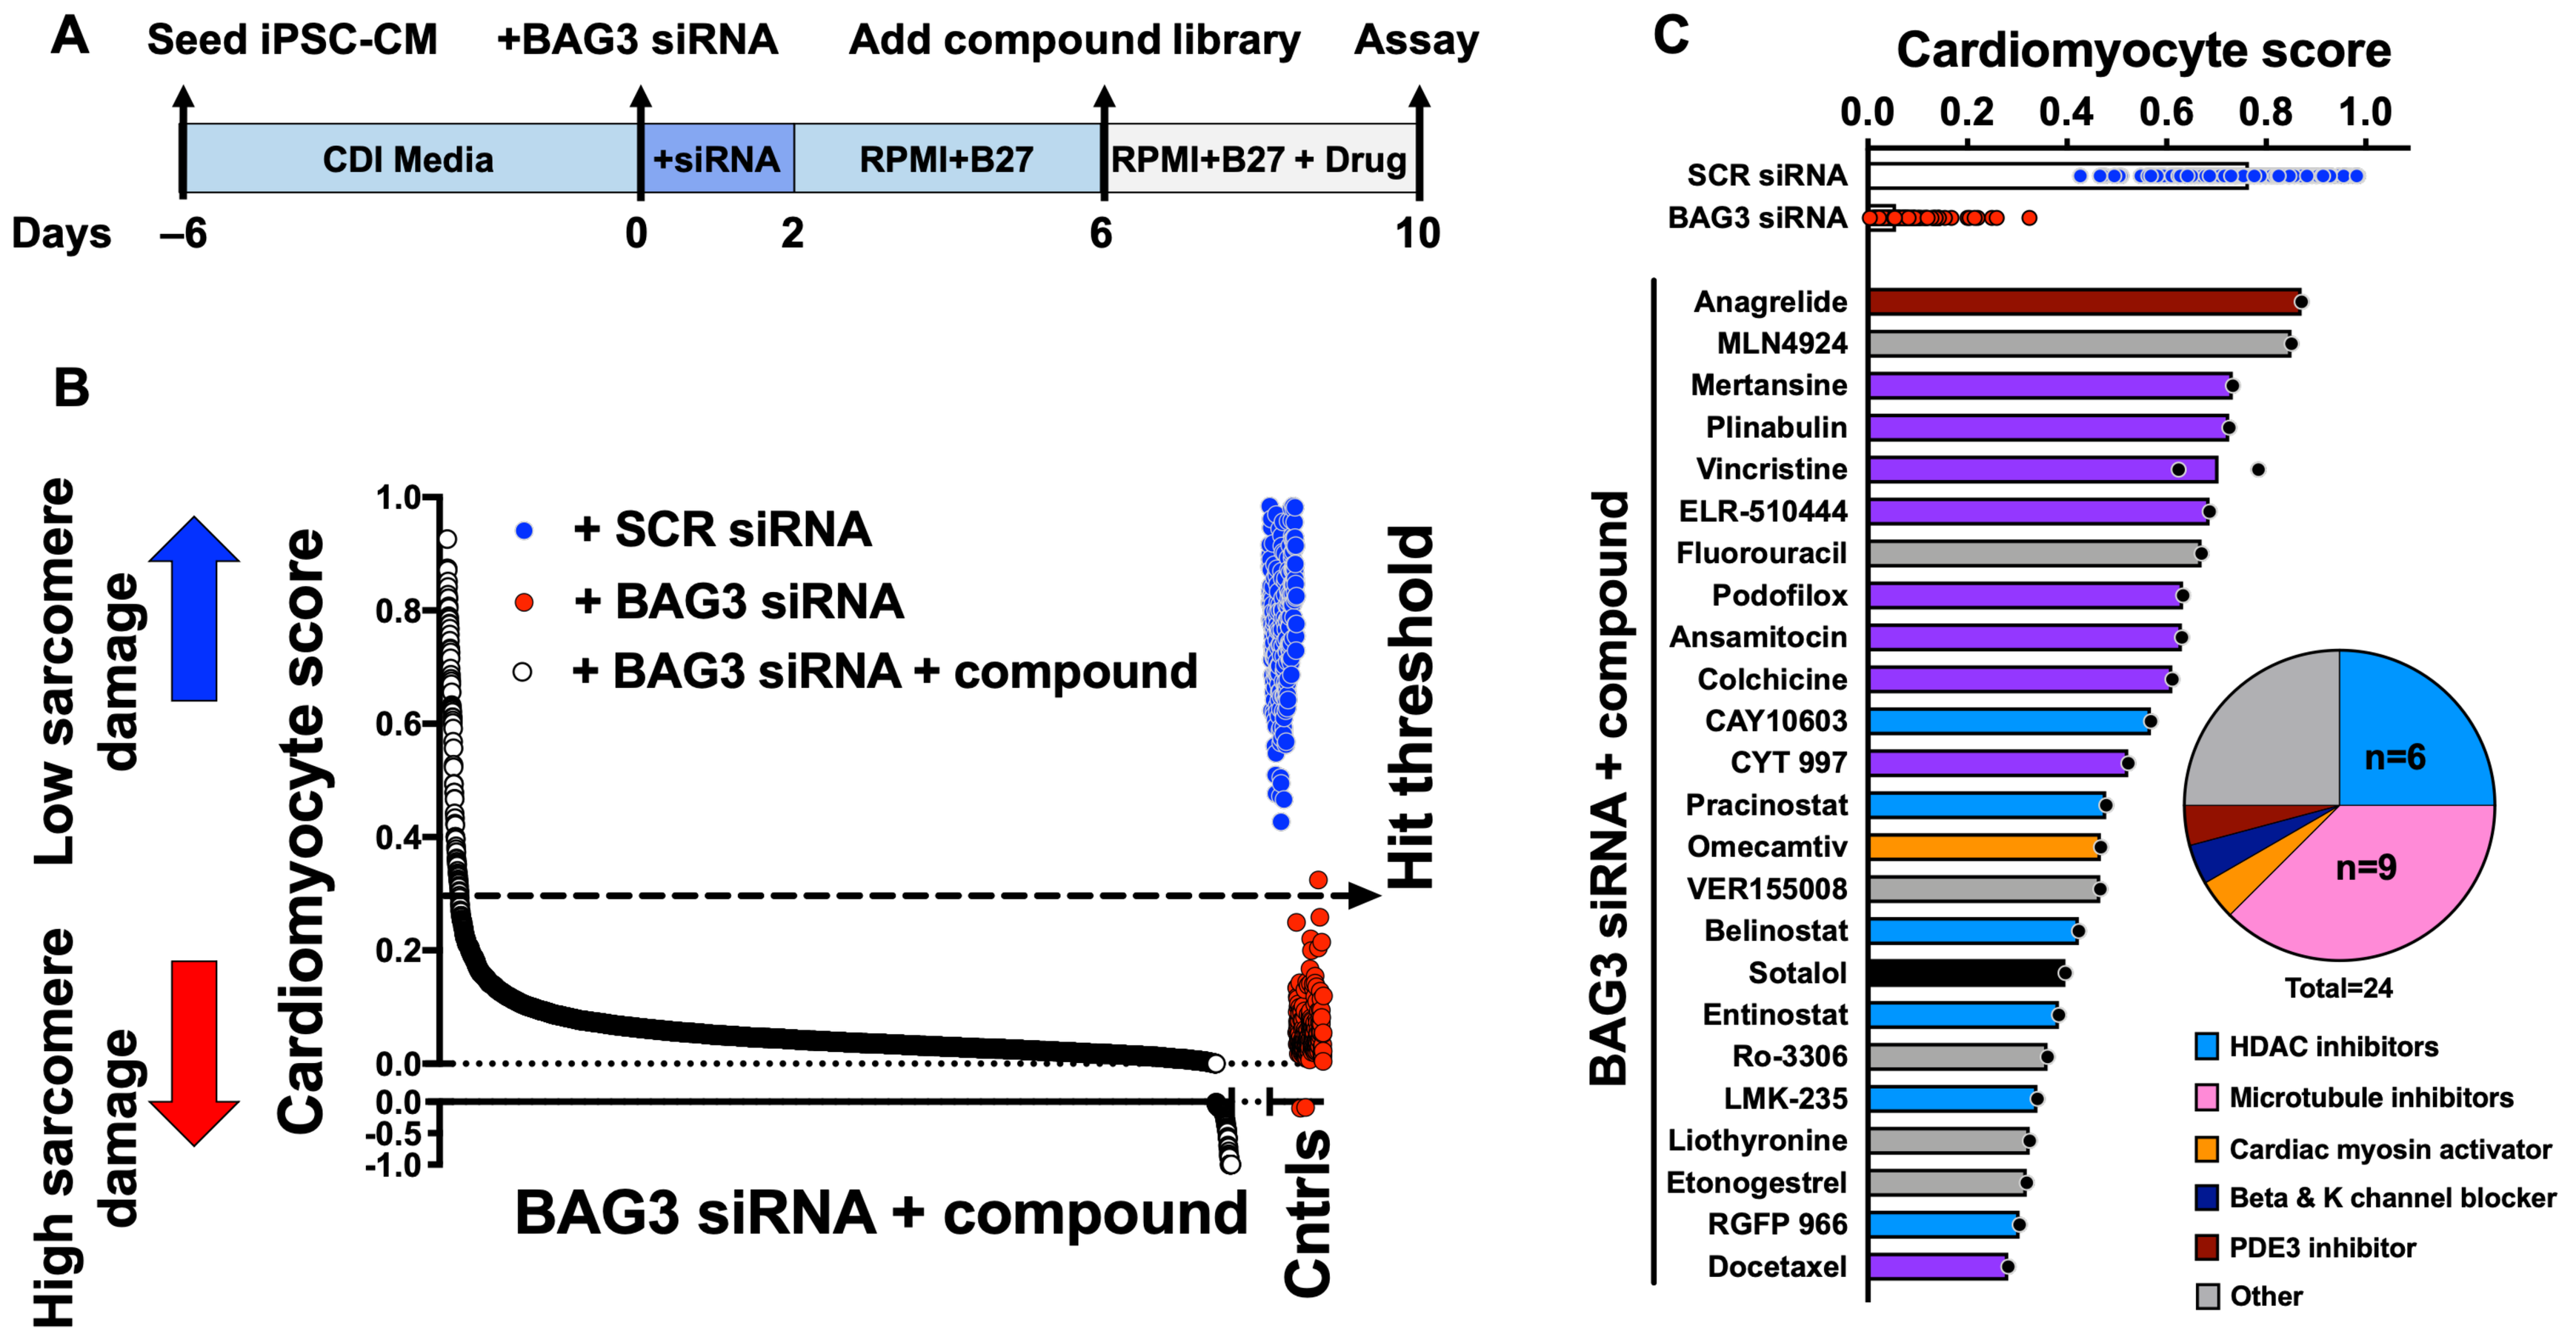

A
Seed iPSC-CM
+BAG3 siRNA
Add compound library
Assay
CDI Media
+siRNA
RPMI+B27
RPMI+B27 + Drug
Days –6 0 2 6 10
B
Low sarcomere damage
High sarcomere damage
Cardiomyocyte score
+ SCR siRNA
+ BAG3 siRNA
+ BAG3 siRNA + compound
Hit threshold
BAG3 siRNA + compound
Cntrls
C
Cardiomyocyte score
SCR siRNA
BAG3 siRNA
BAG3 siRNA + compound
Anagrelide
MLN4924
Mertansine
Plinabulin
Vincristine
ELR-510444
Fluorouracil
Podofilox
Ansamitocin
Colchicine
CAY10603
CYT 997
Pracinostat
Omecamtiv
VER155008
Belinostat
Sotalol
Entinostat
Ro-3306
LMK-235
Liothyronine
Etonogestrel
RGFP 966
Docetaxel
n=6
n=9
Total=24
HDAC inhibitors
Microtubule inhibitors
Cardiac myosin activator
Beta & K channel blocker
PDE3 inhibitor
Other

(A) An unbiased screen was performed using a library of 5500 bioactive compounds. iPSC-CMs were treated with BAG3 siRNA and compounds at a concentration of 1μM. Hits were first identified using deep learning on control iPSC-CMs treated with either SCR or BAG3 siRNA. The hit threshold was set at a cardiomyocyte score of 0.3. (C) The top 24 compounds consisted of histone deacetylase (HDAC) and microtubule inhibitors. In addition, three known heart failure agents were identified: sotalol (beta-blocker and K-channel blocker), omecamtiv mecarbil (cardiac myosin activator), and anagrelide (PDE3 inhibitor).

Target Validation Shows Inhibiting HDAC6 is Sufficient to Protect Against Sarcomere Damage

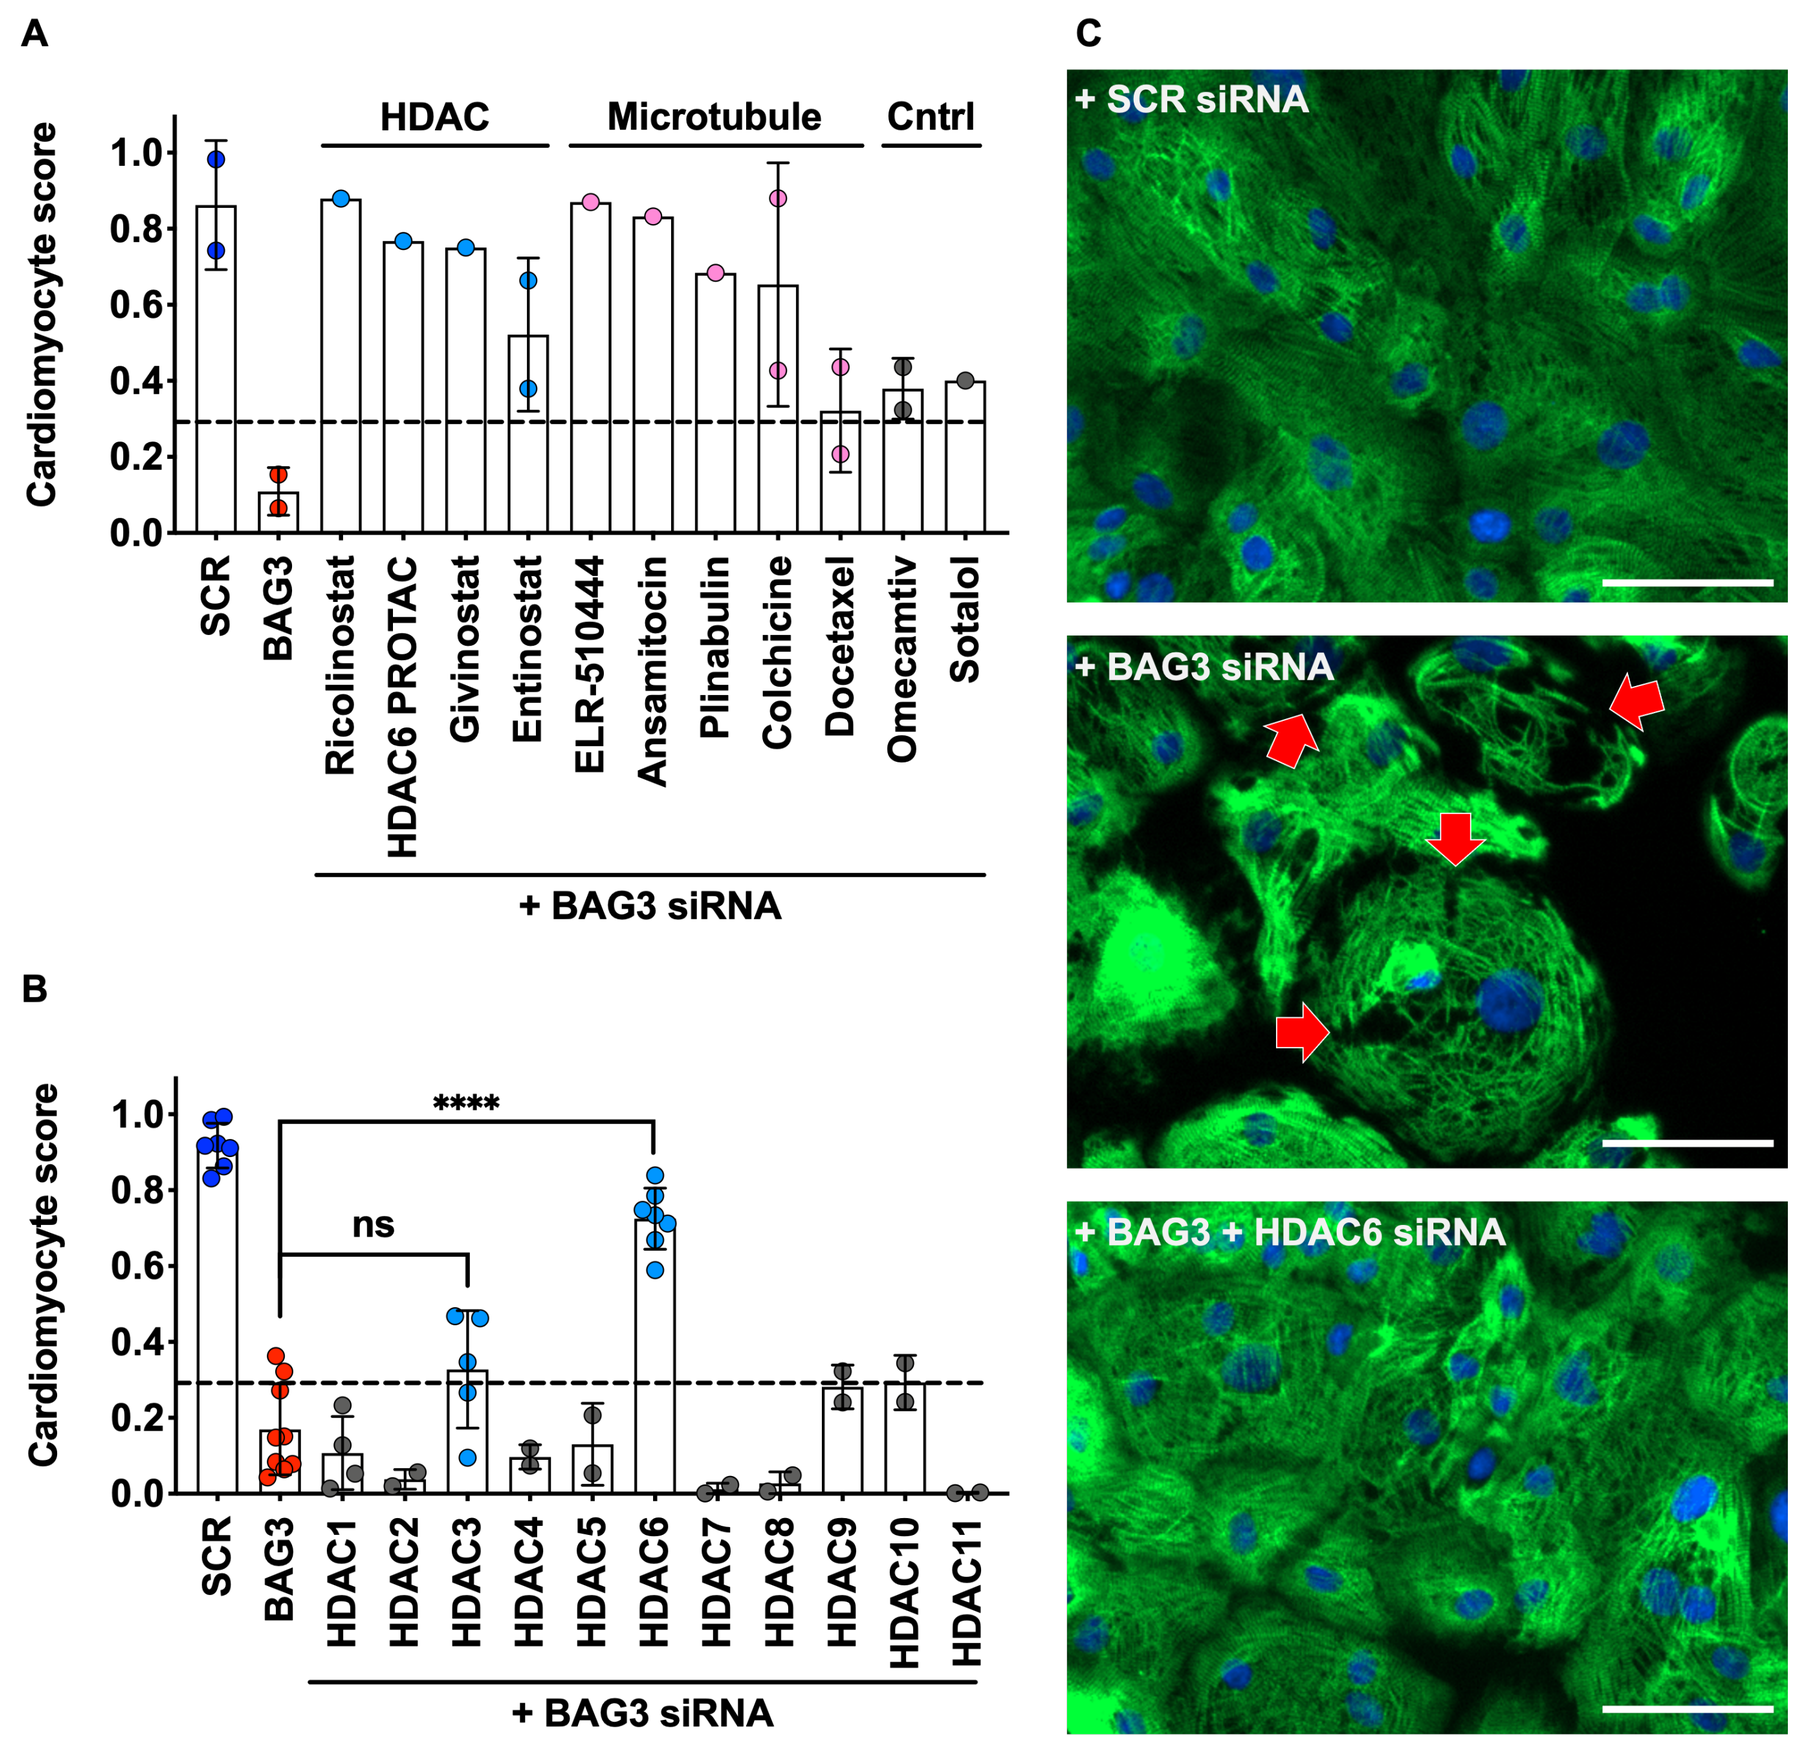

A
Cardiomyocyte score
HDAC
Microtubule
Cntrl
SCR
BAG3
Ricolinostat
HDAC6 PROTAC
Givinostat
Entinostat
ELR-510444
Ansamitocin
Plinabulin
Colchicine
Docetaxel
Omecamtiv
Sotalol
+ BAG3 siRNA
B
ns

SCR BAG3 HDAC1 HDAC2 HDAC3 HDAC4 HDAC5 HDAC6 HDAC7 HDAC8 HDAC9 HDAC10 HDAC11
+ BAG3 siRNA
C
+ SCR siRNA
+ BAG3 siRNA
+ BAG3 + HDAC6 siRNA

(A) Top compound classes (HDAC inhibitors, microtubule inhibitors) from the library screen and two cardiovascular standard-of-care agents [omecamtiv mecarbil (Omecamtiv) and sotalol] identified from the screen were validated at a 1μM dose using the cardiomyocyte score. Data from 1-2 independent biological replicates. n = 4 – 16 technical replicates per condition. Error bars = SD.
(B) Further validation using siRNAs showed that knockdown of HDAC6 protected against sarcomere damage in BAG3^{KD} iPSC-CMs using the cardiomyocyte score with the deep learning algorithm. Data from 2-7 independent biological replicates. n = 4 – 16 technical replicates per condition. Error bars = SD. ****P < 0.0001.
(C) Representative immunostaining of anti-MYBPC3 in iPSC-CMs treated with scramble (SCR), BAG3, or BAG3+HDAC6 siRNA. Arrows indicate sarcomere damage. Scale bar = 50 μm.

Inhibiting HDAC6 with TYA-018 Protects Heart Function in BAG3^{cKO} Mice

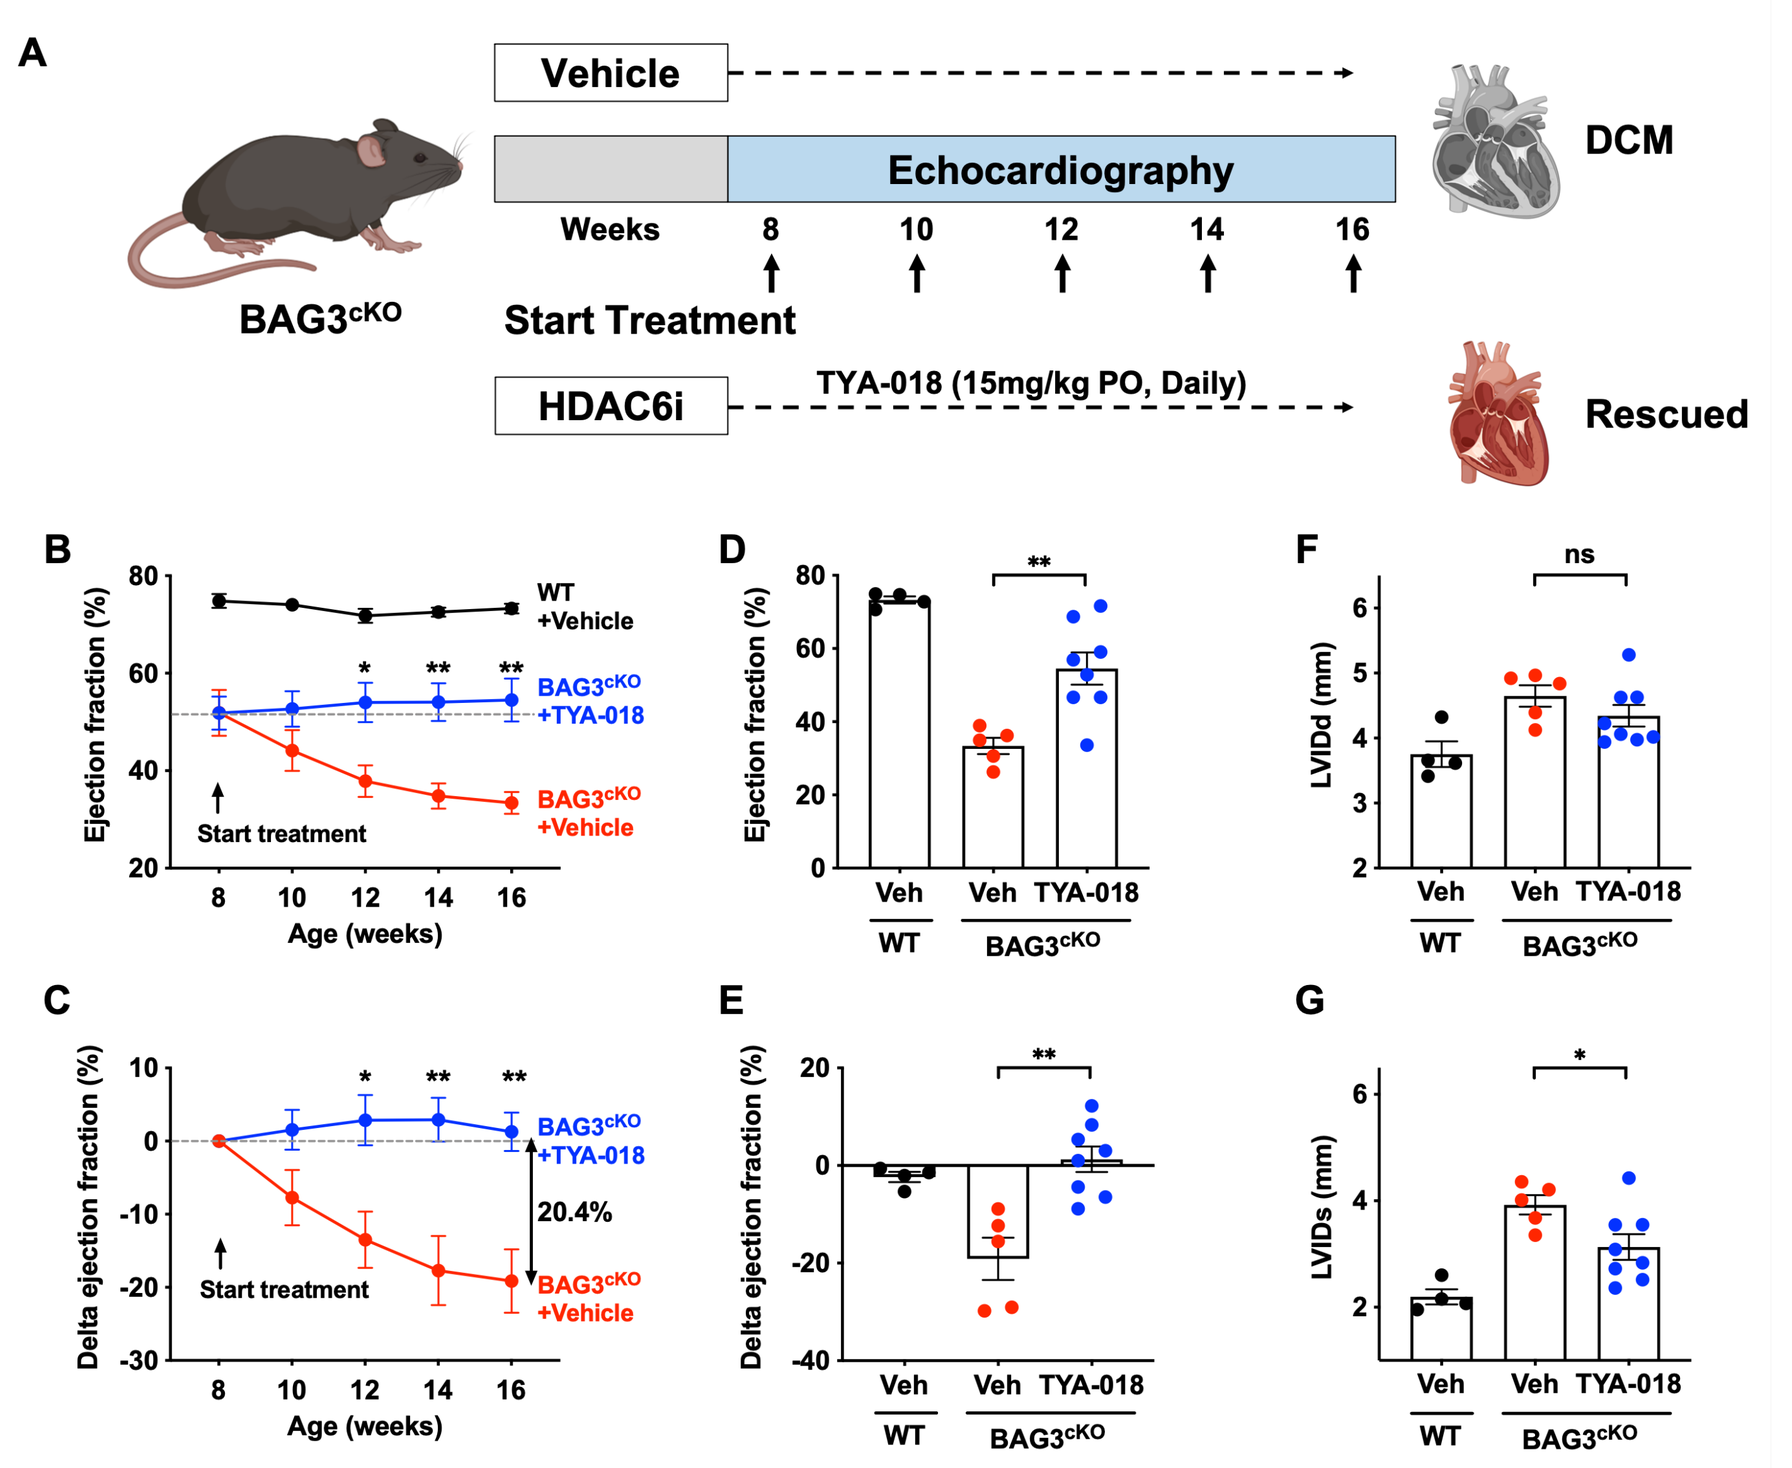

A
Vehicle
Echocardiography
Weeks 8 10 12 14 16
Start Treatment
HDAC6i
TYA-018 (15mg/kg PO, Daily)
BAG3cKO
DCM
Rescued
B
Ejection fraction (%)
WT +Vehicle
BAG3cKO +TYA-018
BAG3cKO +Vehicle
Start treatment
Age (weeks)
C
Delta ejection fraction (%)
20.4%
D
E
F
LVIDd (mm)
G
LVIDs (mm)
Veh TYA-018
WT
BAG3cKO

(A) Schematic of drug treatment in BAG3^{cKO} mouse model. TYA-018 (highly selective HDAC6 inhibitor) was administered daily by oral gavage at 15 mg/kg starting when mice were 2 months of age.
(B) Daily dosing of TYA-018 protected heart function during the 8-week dosing period as measured by ejection faction. Error bars = SEM. **P < 0.01.
(C) Ejection fraction was tracked from the first day of dosing, and delta ejection fraction was measured. During the 8-week period, heart function did not decline in the TYA-018-treated arm, whereas it dropped by 19.1% in the vehicle-treated arm. Error bars = SEM. **P < 0.01.
(D & E) Ejection fraction (D) and delta ejection fraction (E) (compared to the pre-dose baseline) at 4 months of age and 8 weeks of dosing shows TYA-018 protects against declining heart function in BAG3^{cKO} mice. Error bars = SEM. **P < 0.01. Veh, vehicle.
(F & G) Left ventricular internal diameter at diastole (LVIDd) (F) and systole (LVIDs) (G) were reduced by TYA-018 in BAG3^{cKO} mice, bringing the levels closer to that of their wild-type (WT) littermates. Error bars = SEM. *P < 0.05.

TYA-018 Restores Dysregulated Cardiac and Metabolic Transcripts in BAG3^{cKO} Hearts

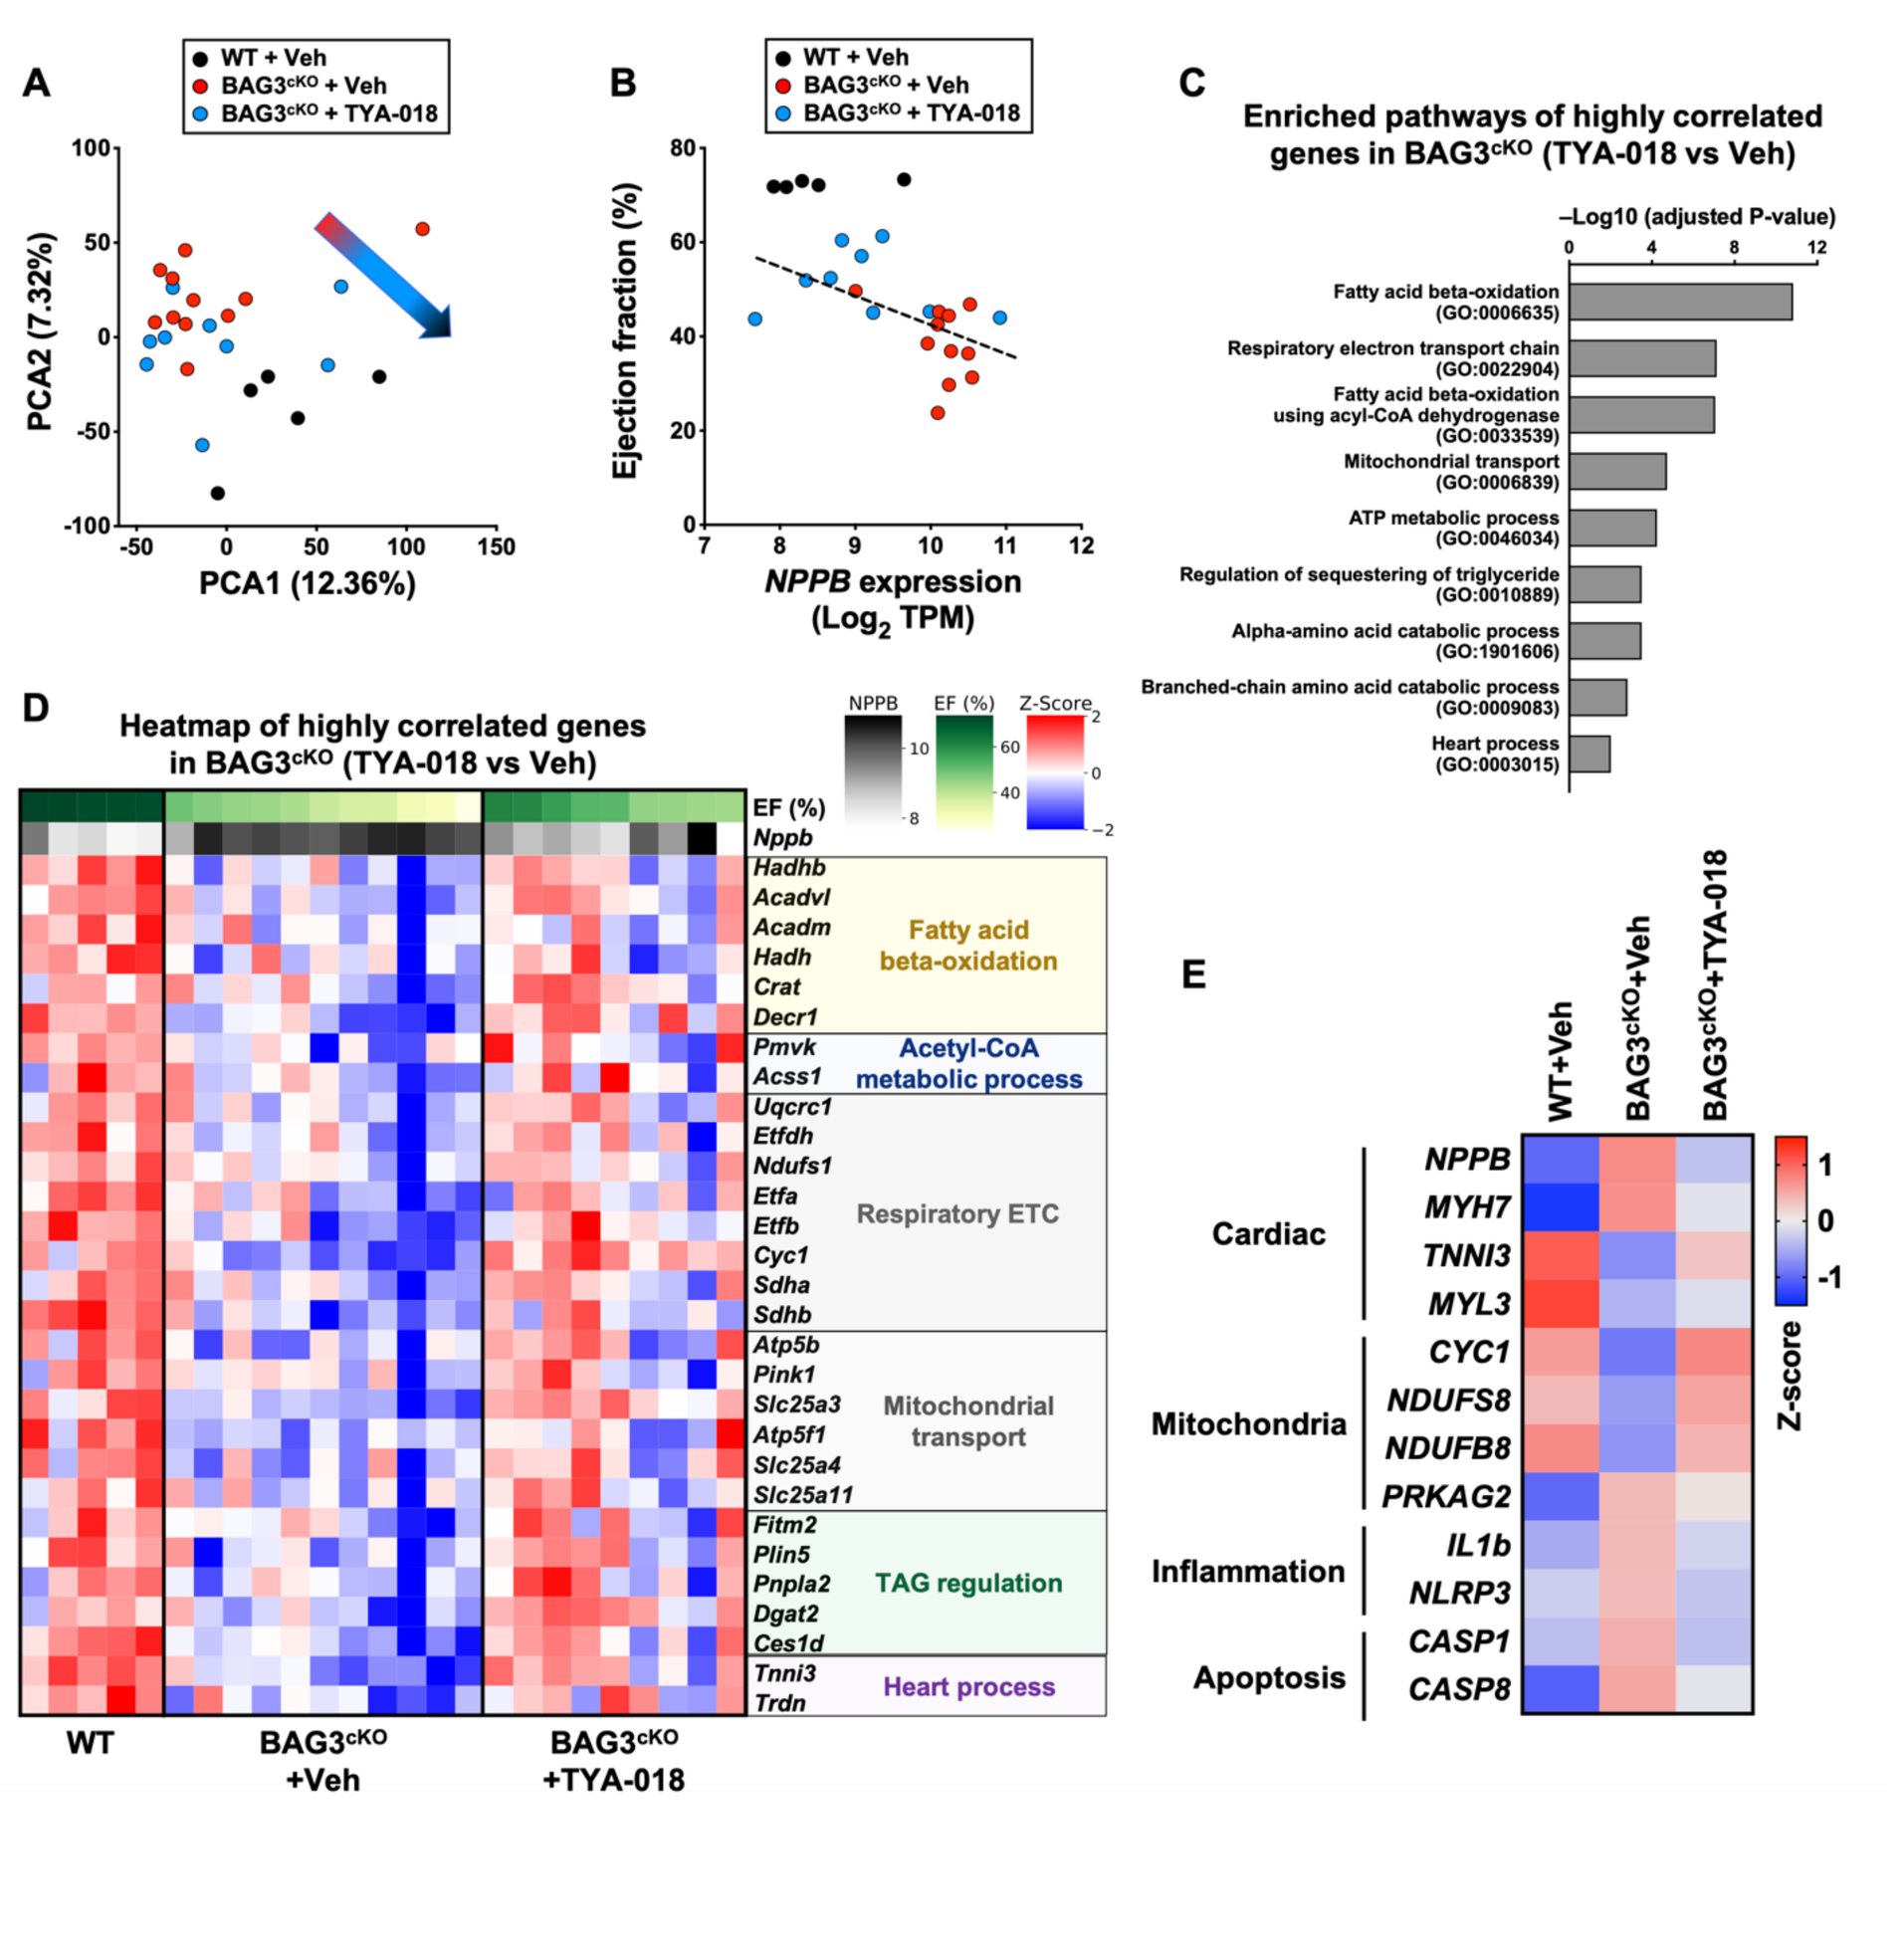

A
WT + Veh
BAG3cKO + Veh
BAG3cKO + TYA-018
PCA2 (7.32%)
PCA1 (12.36%)
B
Ejection fraction (%)
NPPB expression (Log2 TPM)
C
Enriched pathways of highly correlated genes in BAG3cKO (TYA-018 vs Veh)
–Log10 (adjusted P-value)
Fatty acid beta-oxidation (GO:0006635)
Respiratory electron transport chain (GO:0022904)
Fatty acid beta-oxidation using acyl-CoA dehydrogenase (GO:0033539)
Mitochondrial transport (GO:0006839)
ATP metabolic process (GO:0046034)
Regulation of sequestering of triglyceride (GO:0010889)
Alpha-amino acid catabolic process (GO:1901606)
Branched-chain amino acid catabolic process (GO:0009083)
Heart process (GO:0003015)
D
Heatmap of highly correlated genes in BAG3cKO (TYA-018 vs Veh)
Fatty acid beta-oxidation
Acetyl-CoA metabolic process
Respiratory ETC
Mitochondrial transport
TAG regulation
Heart process
WT
BAG3cKO +Veh
BAG3cKO +TYA-018
E
WT+Veh
BAG3cKO+Veh
BAG3cKO+TYA-018
Cardiac
NPPB
MYH7
TNNI3
MYL3
Mitochondria
CYC1
NDUFS8
NDUFB8
PRKAG2
Inflammation
IL1b
NLRP3
Apoptosis
CASP1
CASP8
Z-score

(A) Hearts from all three arms of the study were analyzed using RNA-Seq. Principal component analysis of all coding genes (with Log_2 TPM >1) showed a global correction of BAG3^{cKO}+TYA-018 coding genes toward WT mice.
(B) RNA-Seq analysis shows *NPPB* expression increased by approximately fourfold in BAG3^{cKO} mice compared to WT mice at 4 months of age. TYA-018 treatment reduced *NPPB* levels by twofold in BAG3^{cKO} mice. The level of *NPPB* in BAG3^{cKO}+TYA-018 mice was anticorrelated with heart function.
(C) The top terms in the biological process category enriched in BAG3^{cKO} mice treated with TYA-018. These genes were selected based on positive correlation (R>0.5) with cardiac function (EF%) and negative correlation (R<-0.5) with *NPPB* expression.
(D) Heatmap shows genes from the selected GO terms.
(E) Heatmap of RNA-Seq analysis from a selected number of genes. The data shows correction of key sarcomere genes (*MYH7*, *TNNI3*, and *MYL3*) and genes regulating mitochondrial function and metabolism (*CYC1*, *NDUFS8*, *NDUFB8*, *PPKARG2*) in BAG3^{cKO}+TYA-018 mice. Inflammatory (*IL-1β*, *NLRP3*) and apoptosis (*CASP1*, *CAPS8*) markers were also reduced.